Regional administration of Class C CpG Oligodeoxynucleotides results in superior intrahepatic TLR9 activation and immunomodulation compared to systemic infusion

Abstract #1793

Chandra C. Ghosh[1], Kara Heatherton[1], Kyle O'Connell[1], Jason LaPorte[1], Prajna Guha[1], Bryan F. Cox[2], David Jaroch[2], Steven C. Katz[1,2]

[1.] Immuno-oncology Institute (Ix2), Roger Williams Medical Center, Providence, RI-02908; [2.] TriSalus Life Sciences, Westminster, CO-80031

Abstract

Rationale: CpG oligodeoxynucleotides (ODNs) are TLR9 agonists (TLR9As) that stimulate pDC and B cells. Yet, multiple other cell types in the liver express TLR9. The effect of TLR9A in reshaping the tumor microenvironment (TME), particularly in the myeloid-derived suppressor cells (MDSC) compartment, is yet to be determined. Clinical success of TLR9As has been limited by toxicity when given systemically and delivery limitations when administered by direct needle injection. Here, we hypothesized that regional intra-vascular infusion of TRL9As would reprogram the liver MDSC compartment to enable immune control of liver metastases (LM). In this study, we evaluated the effect of class C ODN-2395 as TLR9A in controlling LM progression and its impact on the liver MDSC population.

Methods: C57/BL6 mice were challenged with 2.5e6 MC38-CEA-Luc cells via intra-splenic route. After a week, mice were treated with 1, 3, 10 or 30µg ODN2395 via Portal Vein (PV) or 30 µg Intravenously (IV). Bioluminescence was measured at 24 and 48 hrs post-ODN administration (**Schema**). CD45+ cells were isolated, and flow cytometry (FC) analysis was performed to quantify MDSCs, monocytic MDSCs (M-MDSC), and M1-macrophage subsets, along with downstream signaling.

Results: We found that 30µg ODN2395 delivered via PV was more efficient compared to TV in reducing tumor burden at 24 hrs and persisted up to 48 hrs. Regional delivery of ODN2395 also reduced the frequency of MDSCs, predominantly the more immunosuppressive M-MDSCs subpopulation in LM. Furthermore, enhanced pro-inflammatory/anti-tumorigenic M1 macrophages. Using an NFκB-dependent soluble alkaline phosphatase assay (SEAP), we determined that ODN2395 dose-dependently enhanced NFκB transcription factor activity ($p<0.001$). Western Blot analysis of tumor lysates show that ODN delivery by PV significantly increased NFκB (pP65) activity and production of IL-6 as well as reduced STAT3 activity relative to IV.

Summary: Overall, our data suggest that the regional delivery of a TLR9A has the potential to improve control of liver tumor growth in conjunction with the elimination of immunosuppressive myeloid cells.

Schema

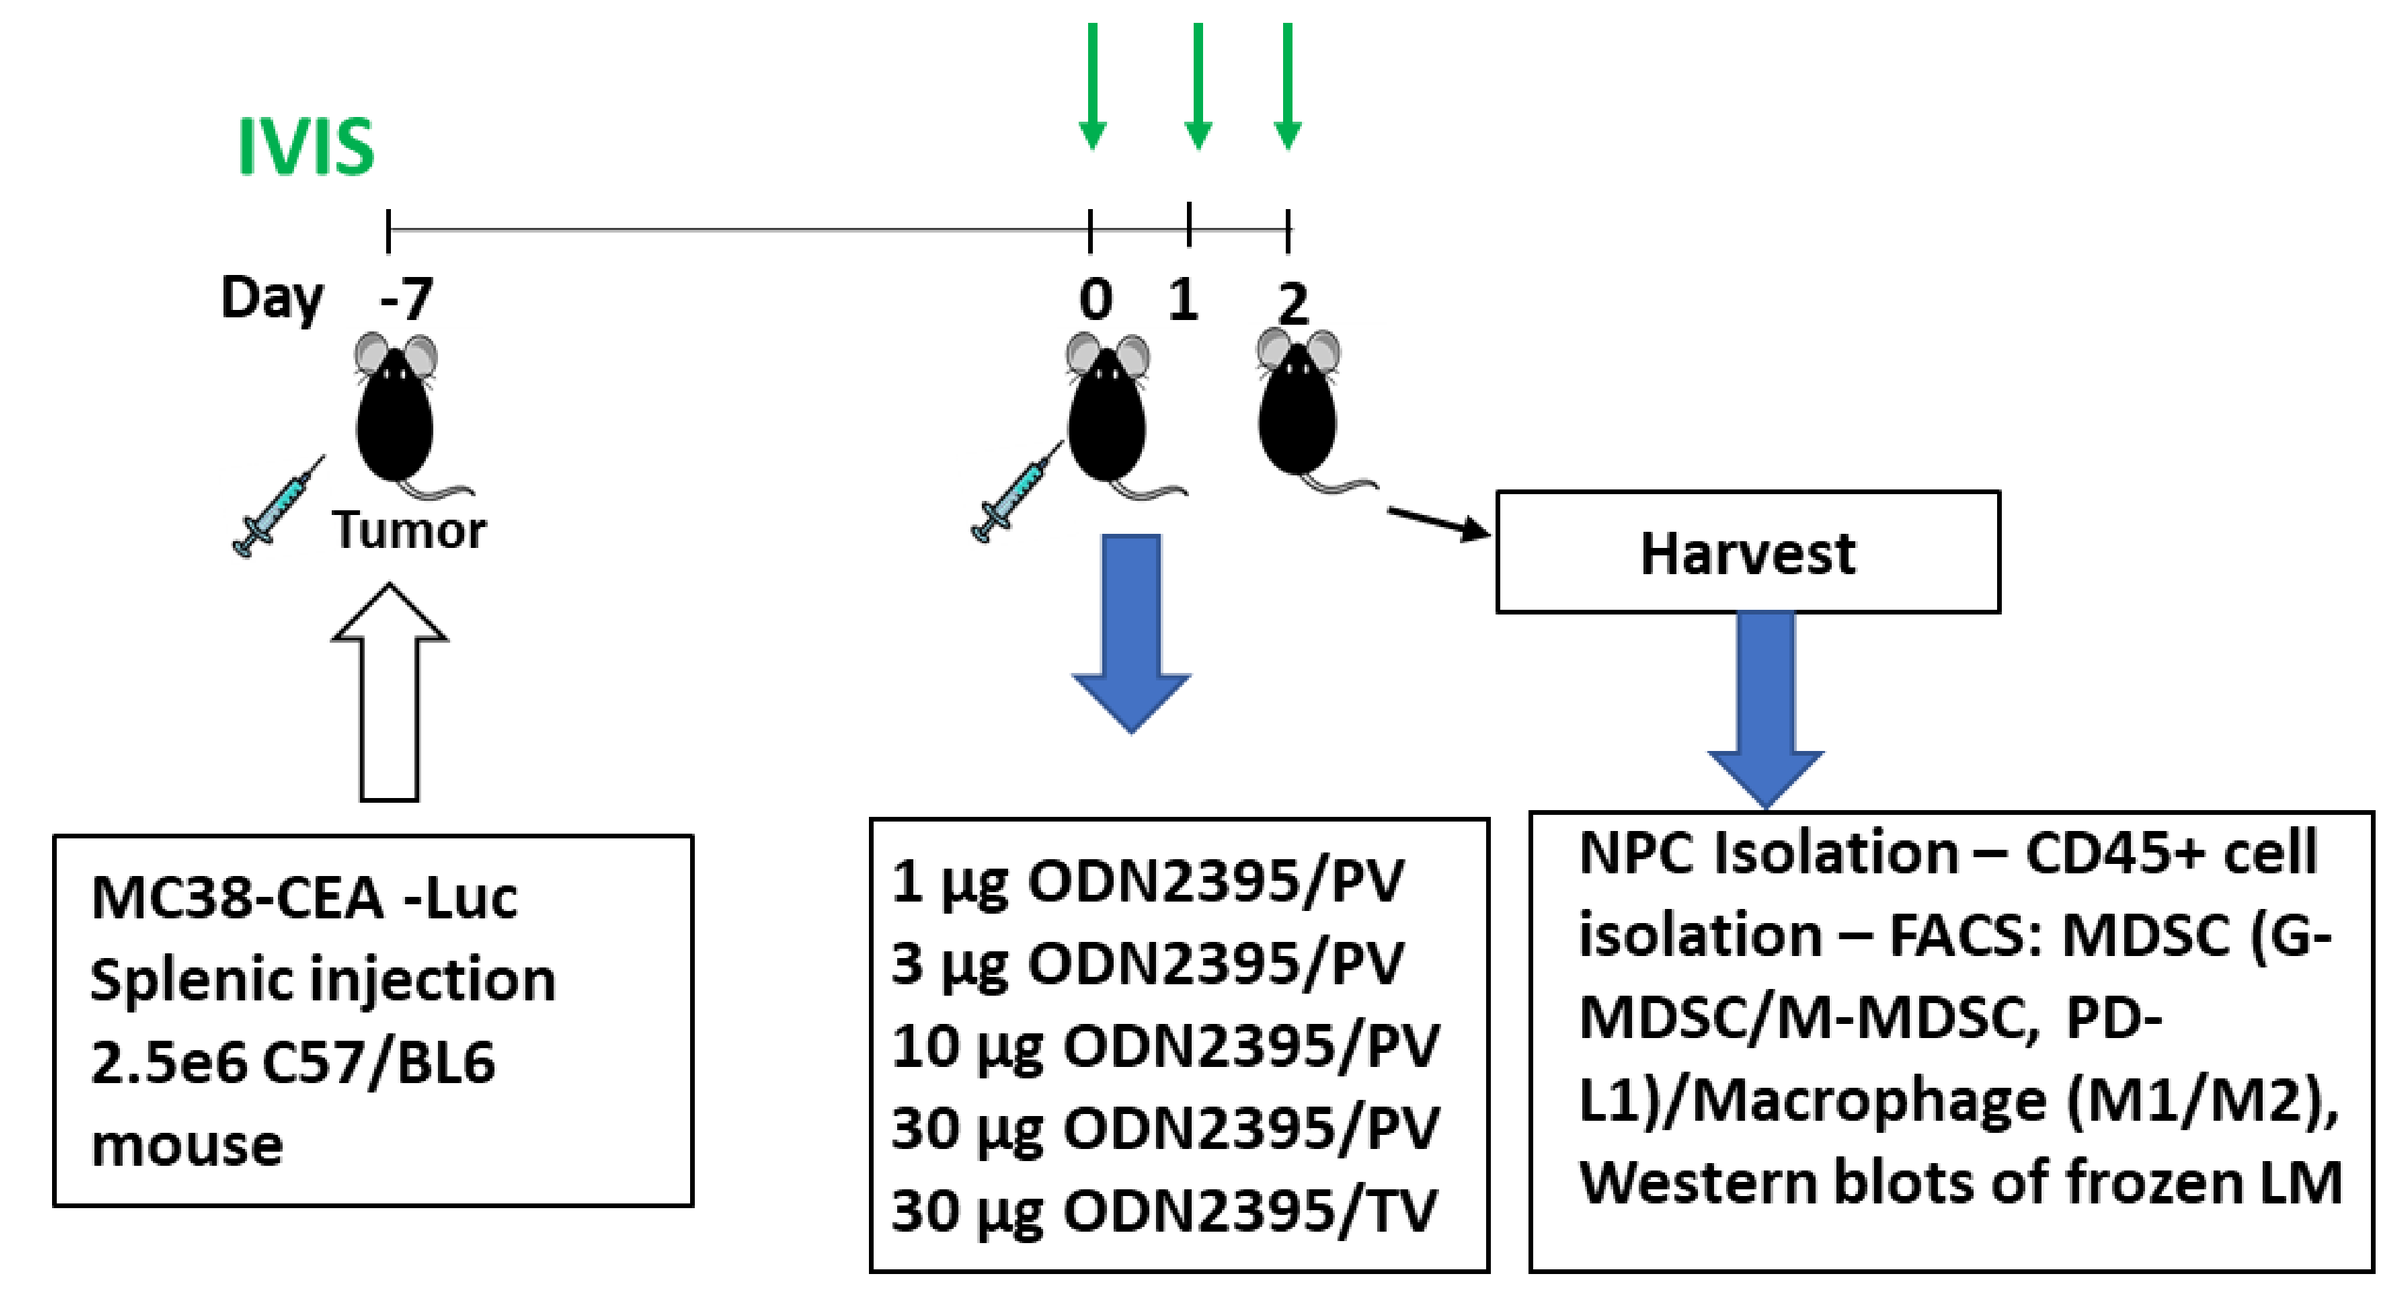
IVIS
Day -7
0 1 2
Tumor
Harvest
MC38-CEA -Luc Splenic injection 2.5e6 C57/BL6 mouse
1 µg ODN2395/PV
3 µg ODN2395/PV
10 µg ODN2395/PV
30 µg ODN2395/PV
30 µg ODN2395/TV
NPC Isolation – CD45+ cell isolation – FACS: MDSC (G-MDSC/M-MDSC, PD-L1)/Macrophage (M1/M2), Western blots of frozen LM

Schema: Illustrates the steps to develop LM and the treatment protocol. Eight to twelve weeks male C57/BL6 mice were challenged intra-splenic with $2.5e^6$ MC38-CEA-Luc cells for a week. Bioluminescence value was determined by IVIS, and mice were randomized accordingly and treated with 1, 3, 10, 30µg/mouse ODN 2395 via PV and 30µg/mouse ODN 2395 via TV. For the subsequent study on D+2 post-treatment, mice were sacrificed, and liver was harvested to isolate CD45⁺ cells. Isolated CD45⁺ NPCs were evaluated for MDSCs and macrophages.

Figure 1. ODN2395 administered via PV are more effective in inhibiting tumor progression.

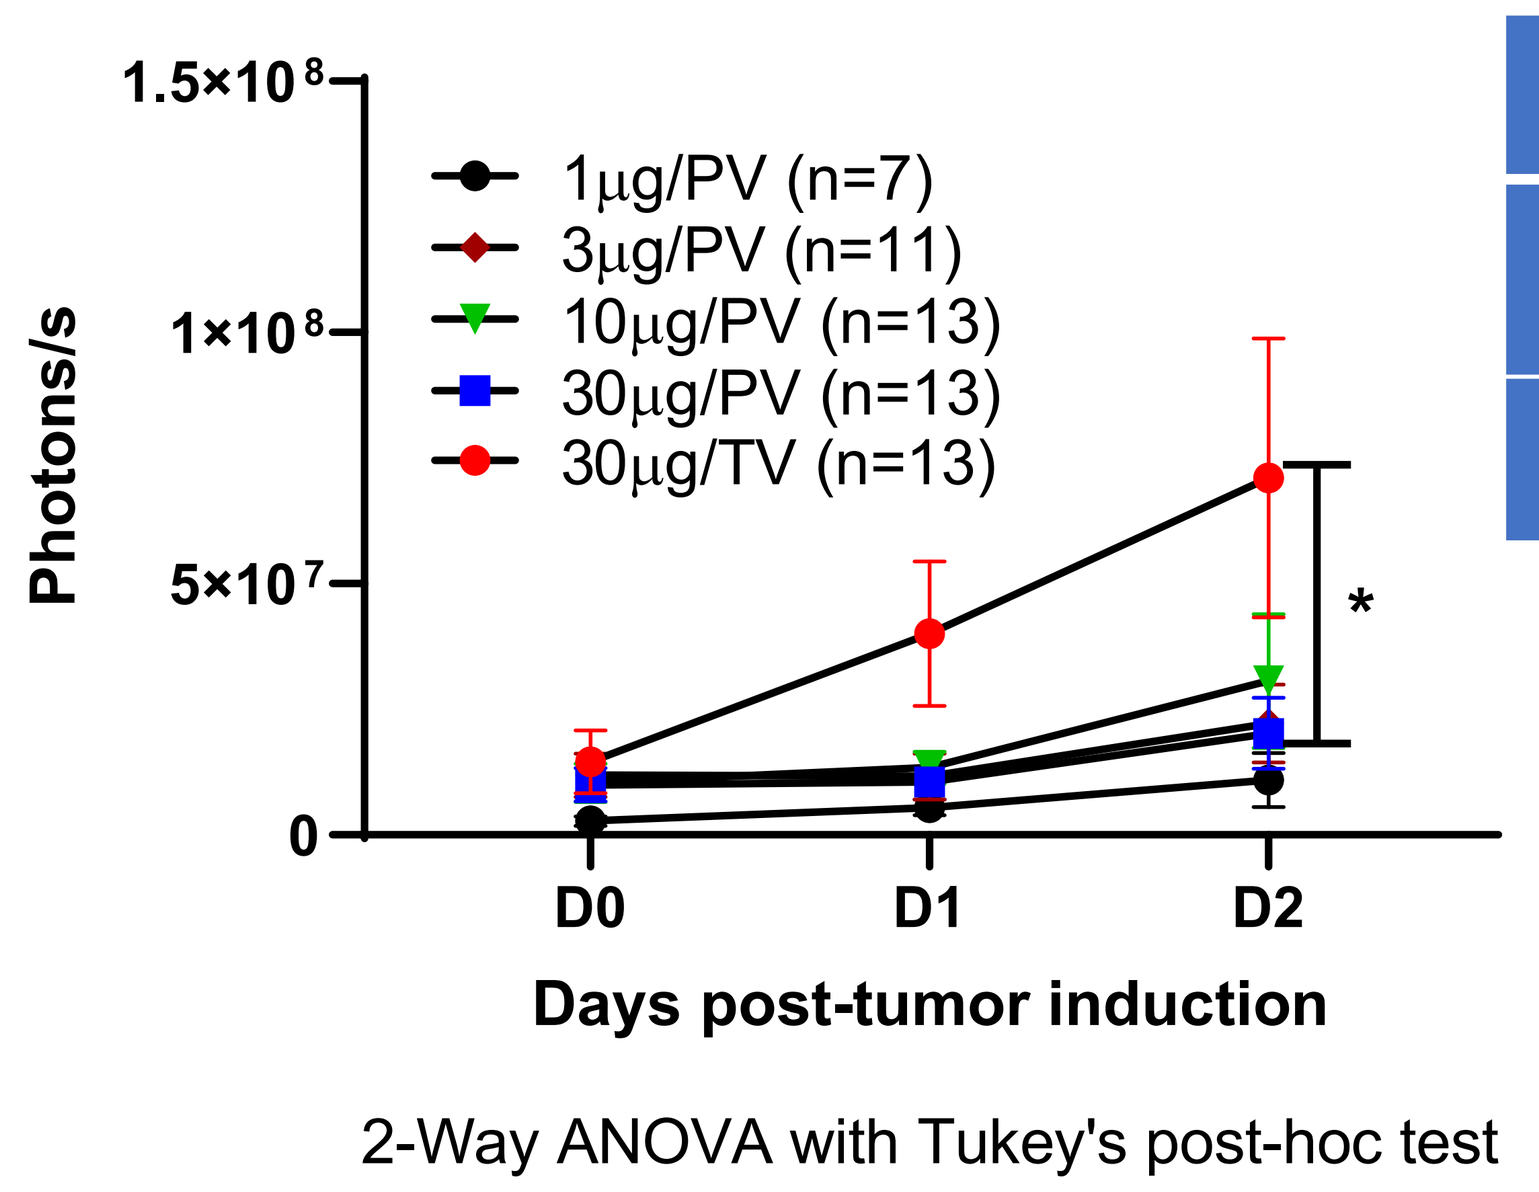
Photons/s
1.5×10⁸
1×10⁸
5×10⁷
0
1µg/PV (n=7)
3µg/PV (n=11)
10µg/PV (n=13)
30µg/PV (n=13)
30µg/TV (n=13)
*
D0
D1
D2
Days post-tumor induction
2-Way ANOVA with Tukey's post-hoc test

	µg TV 13	µg PV 13	µg PV 7
D2 (Photons/s)	e7	e7	e7
value 30µg TV		0.05	0.05

Figure 1: A. Tumor growth was monitored by IVIS imaging at the day of treatment (D0), D1 and D2.Tumor progression was analyzed by 2-Way ANOVA followed by Tukey's post-hoc test (*p<0.05). B. Tabular representation of tumor burden that were treated by 30µg ODN2395 either by PV or TV.

Results

Figure 2. ODN2395 administered via PV reduced MDSC population in LM

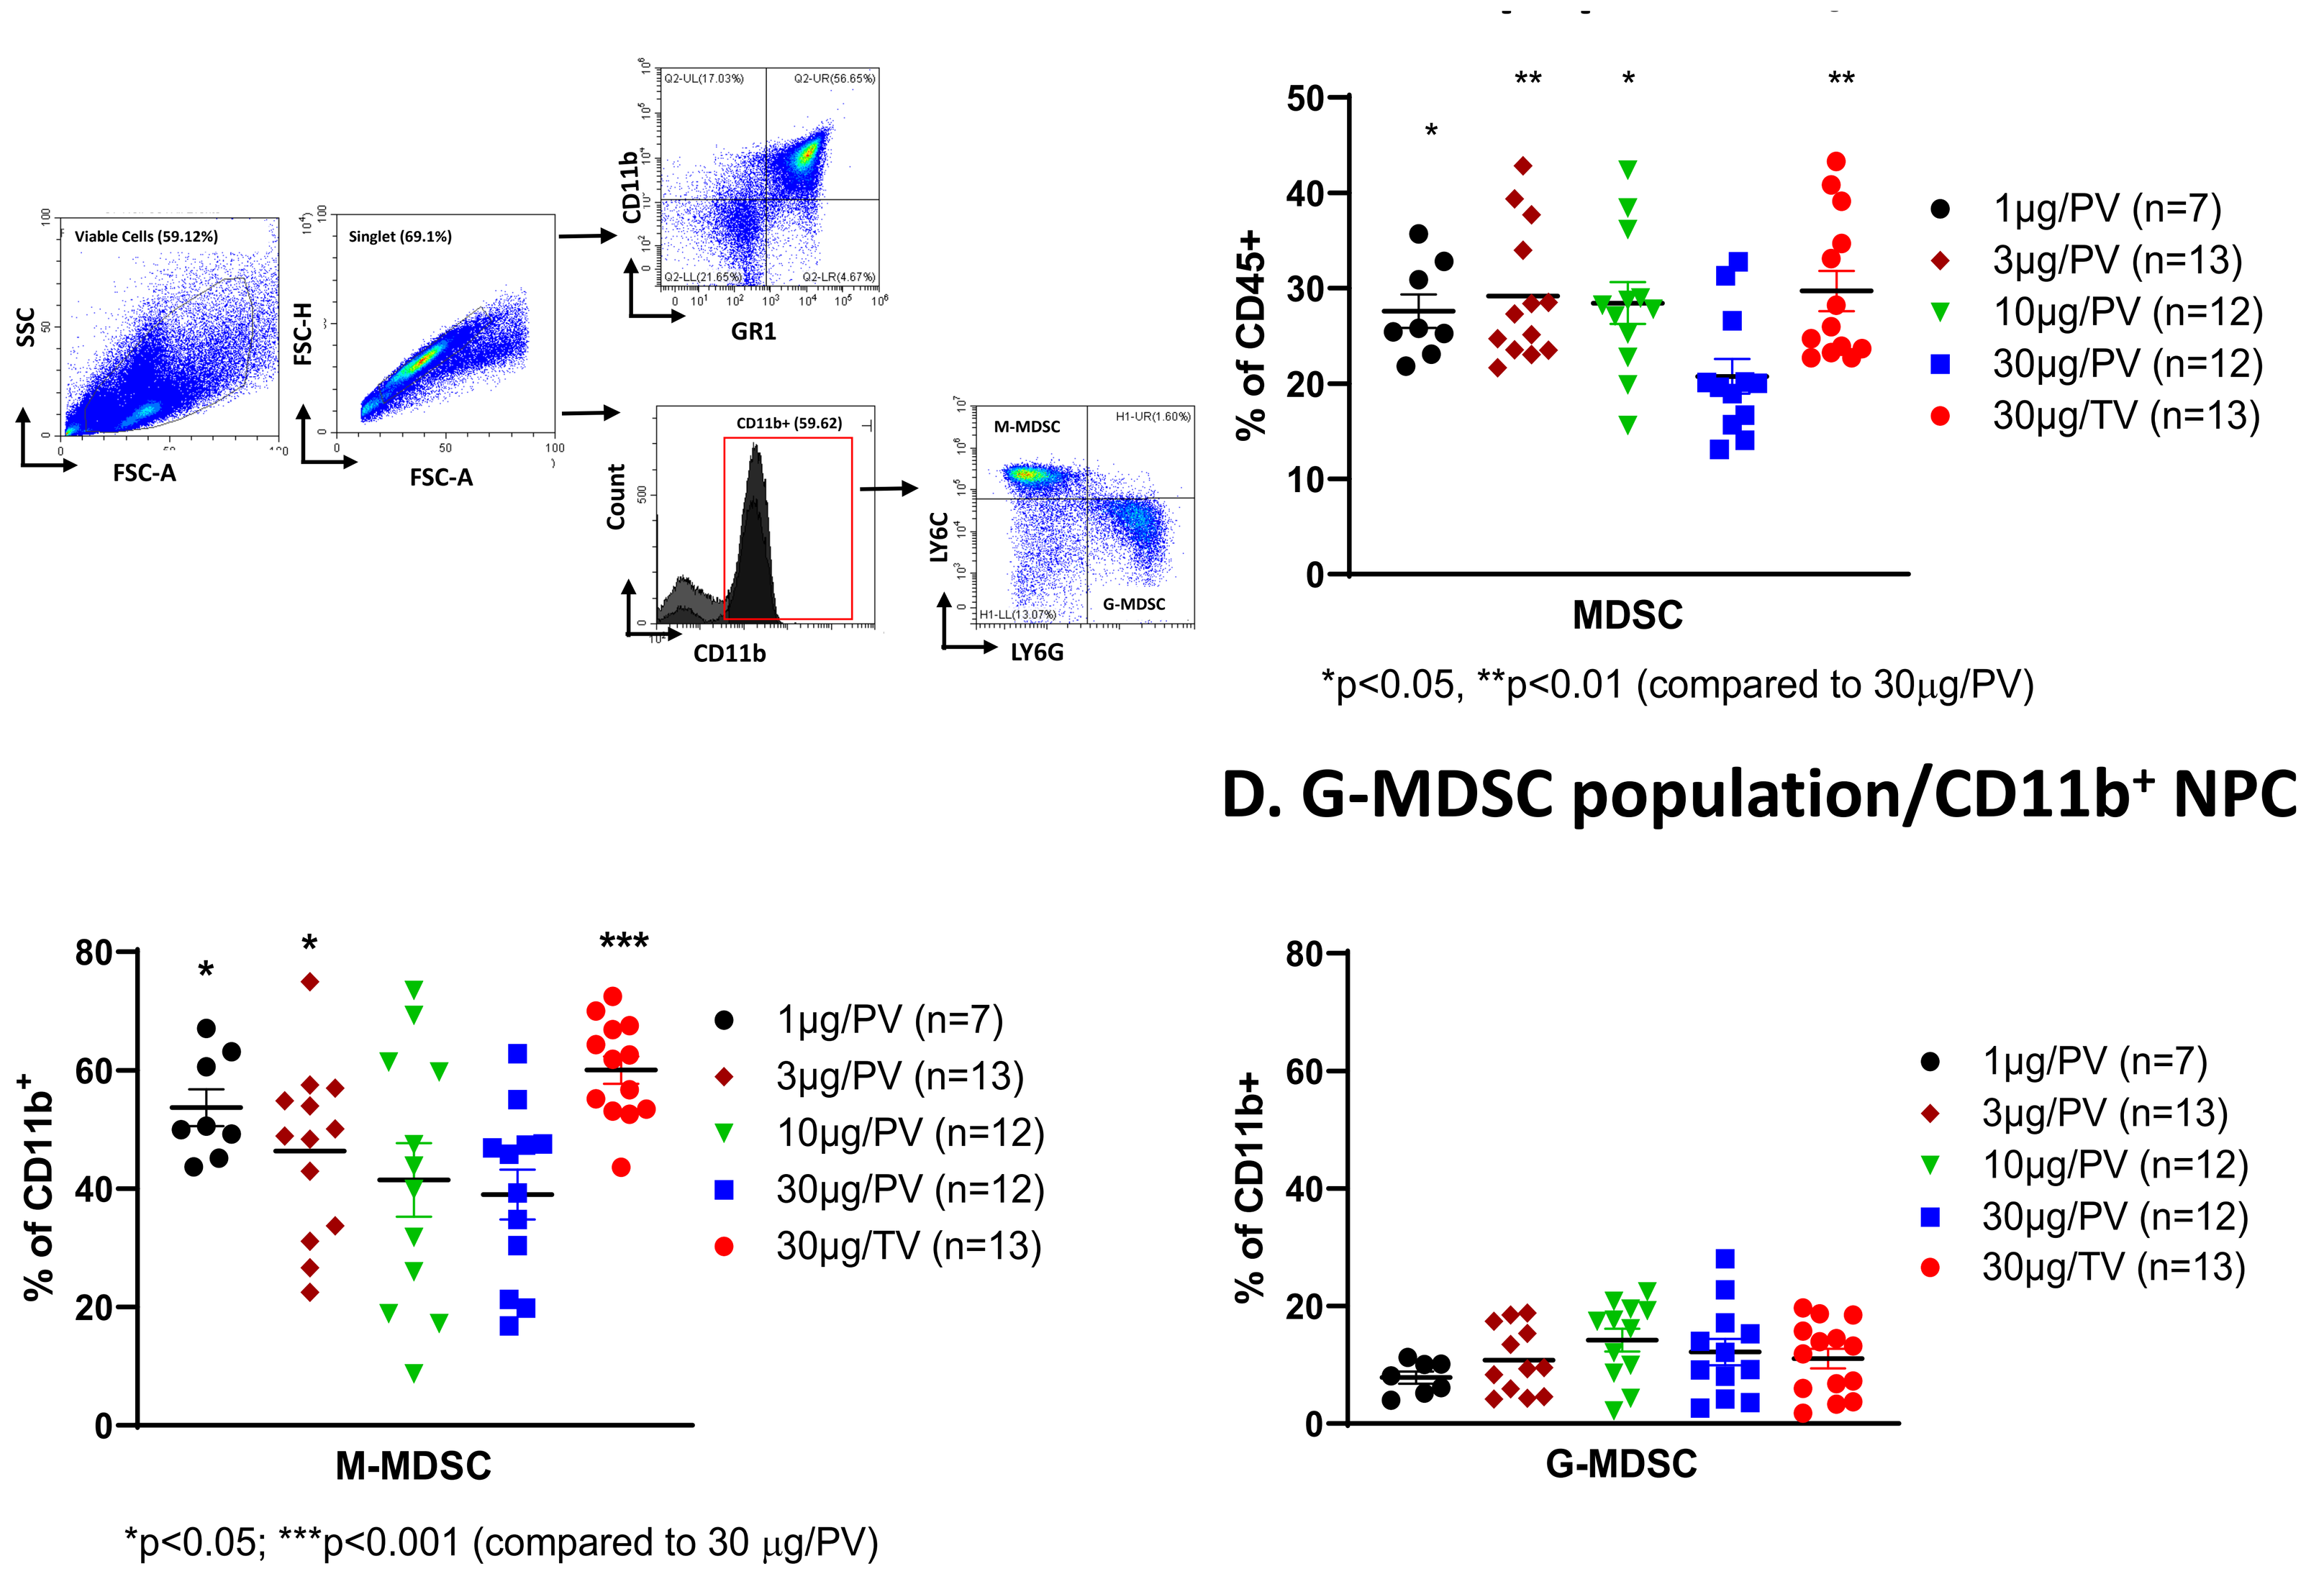
Viable Cells (59.12%)
Singlet (69.1%)
SSC
FSC-H
FSC-A
CD11b
GR1
Count
CD11b+ (59.62)
LY6C
LY6G
M-MDSC
G-MDSC
% of CD45+
MDSC
1µg/PV (n=7)
3µg/PV (n=13)
10µg/PV (n=12)
30µg/PV (n=12)
30µg/TV (n=13)
*p<0.05, **p<0.01 (compared to 30µg/PV)
D. G-MDSC population/CD11b⁺ NPC
% of CD11b⁺
M-MDSC
*p<0.05; ***p<0.001 (compared to 30 µg/PV)
% of CD11b+
G-MDSC

Figure 2. As described in schema, mice were sacrificed after two days post-treatment. CD45⁺ cells were isolated from NPCs. **A.** Gating strategy to analyze CD45⁺ cells isolated from the LM by FC. **B, C, D**. MDSC cell population (CD11b⁺Gr1⁺), monocytic MDSC (M-MDSC; CD11b⁺Ly6C⁺) cells and granulocytic MDSC (G-MDSC; CD11b⁺LY6G⁺) were measured. Data of each animal is represented on a scattered plot and presented as mean±SEM from at least 3 different experiments. Student's t test was performed for groupwise comparison and are described in each graph .

Figure 3. ODN2395 administered via PV increased M1- and reduced M2-macrophage populations in LM

A. Gating Strategy

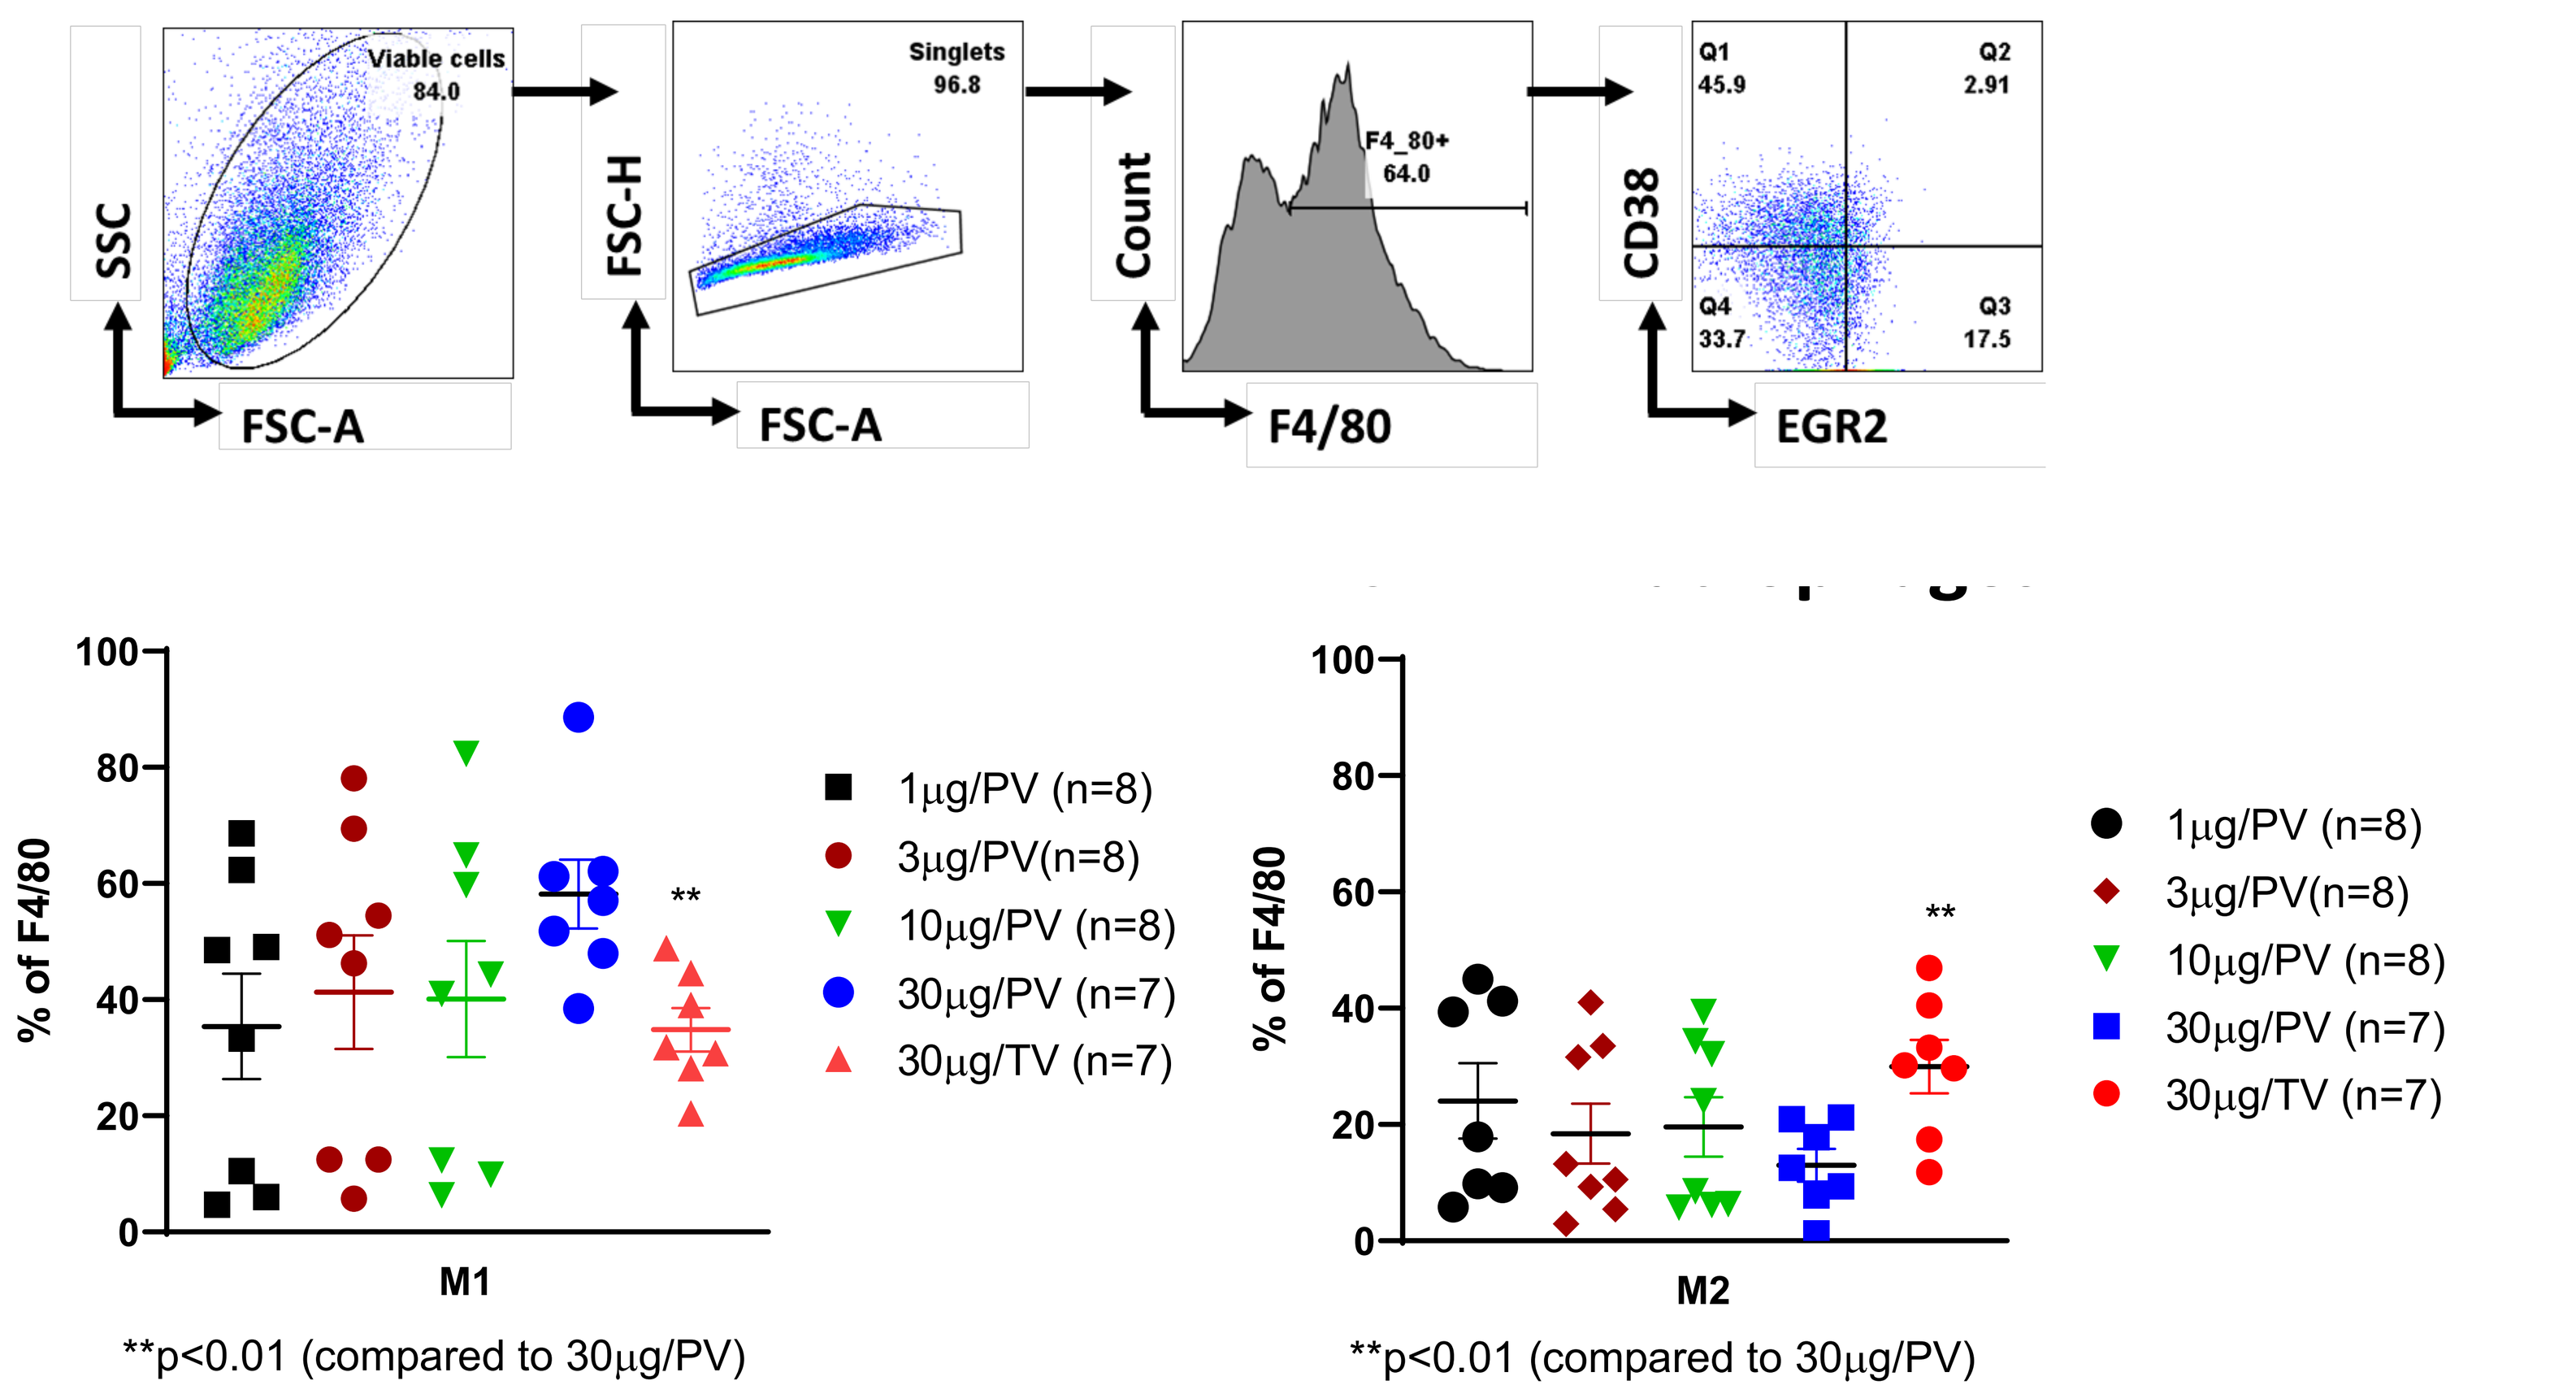
Viable cells 84.0
Singlets 96.8
F4_80+ 64.0
Q1 45.9
Q2 2.91
Q4 33.7
Q3 17.5
SSC
FSC-H
Count
CD38
FSC-A
F4/80
EGR2
% of F4/80
M1
1µg/PV (n=8)
3µg/PV(n=8)
10µg/PV (n=8)
30µg/PV (n=7)
30µg/TV (n=7)
**p<0.01 (compared to 30µg/PV)
M2

Figure 3: Isolated CD45⁺ cells from LM, were analyzed for M1- and M2-macrophages. **A.** Gating strategy of the phenotypic analysis of macrophages isolated from the LM is shown. **B, C.** M1-macrophage cell population (F4/80⁺CD38⁺EGR2⁻) and M2-macrophage cell population (F4/80⁺CD38⁻EGR2⁺) were measured. Student's t-test was performed for group-wise comparison.

Figure 4. ODN2395 administered via PV enhanced tumor NFκB signaling compared to systemic delivery

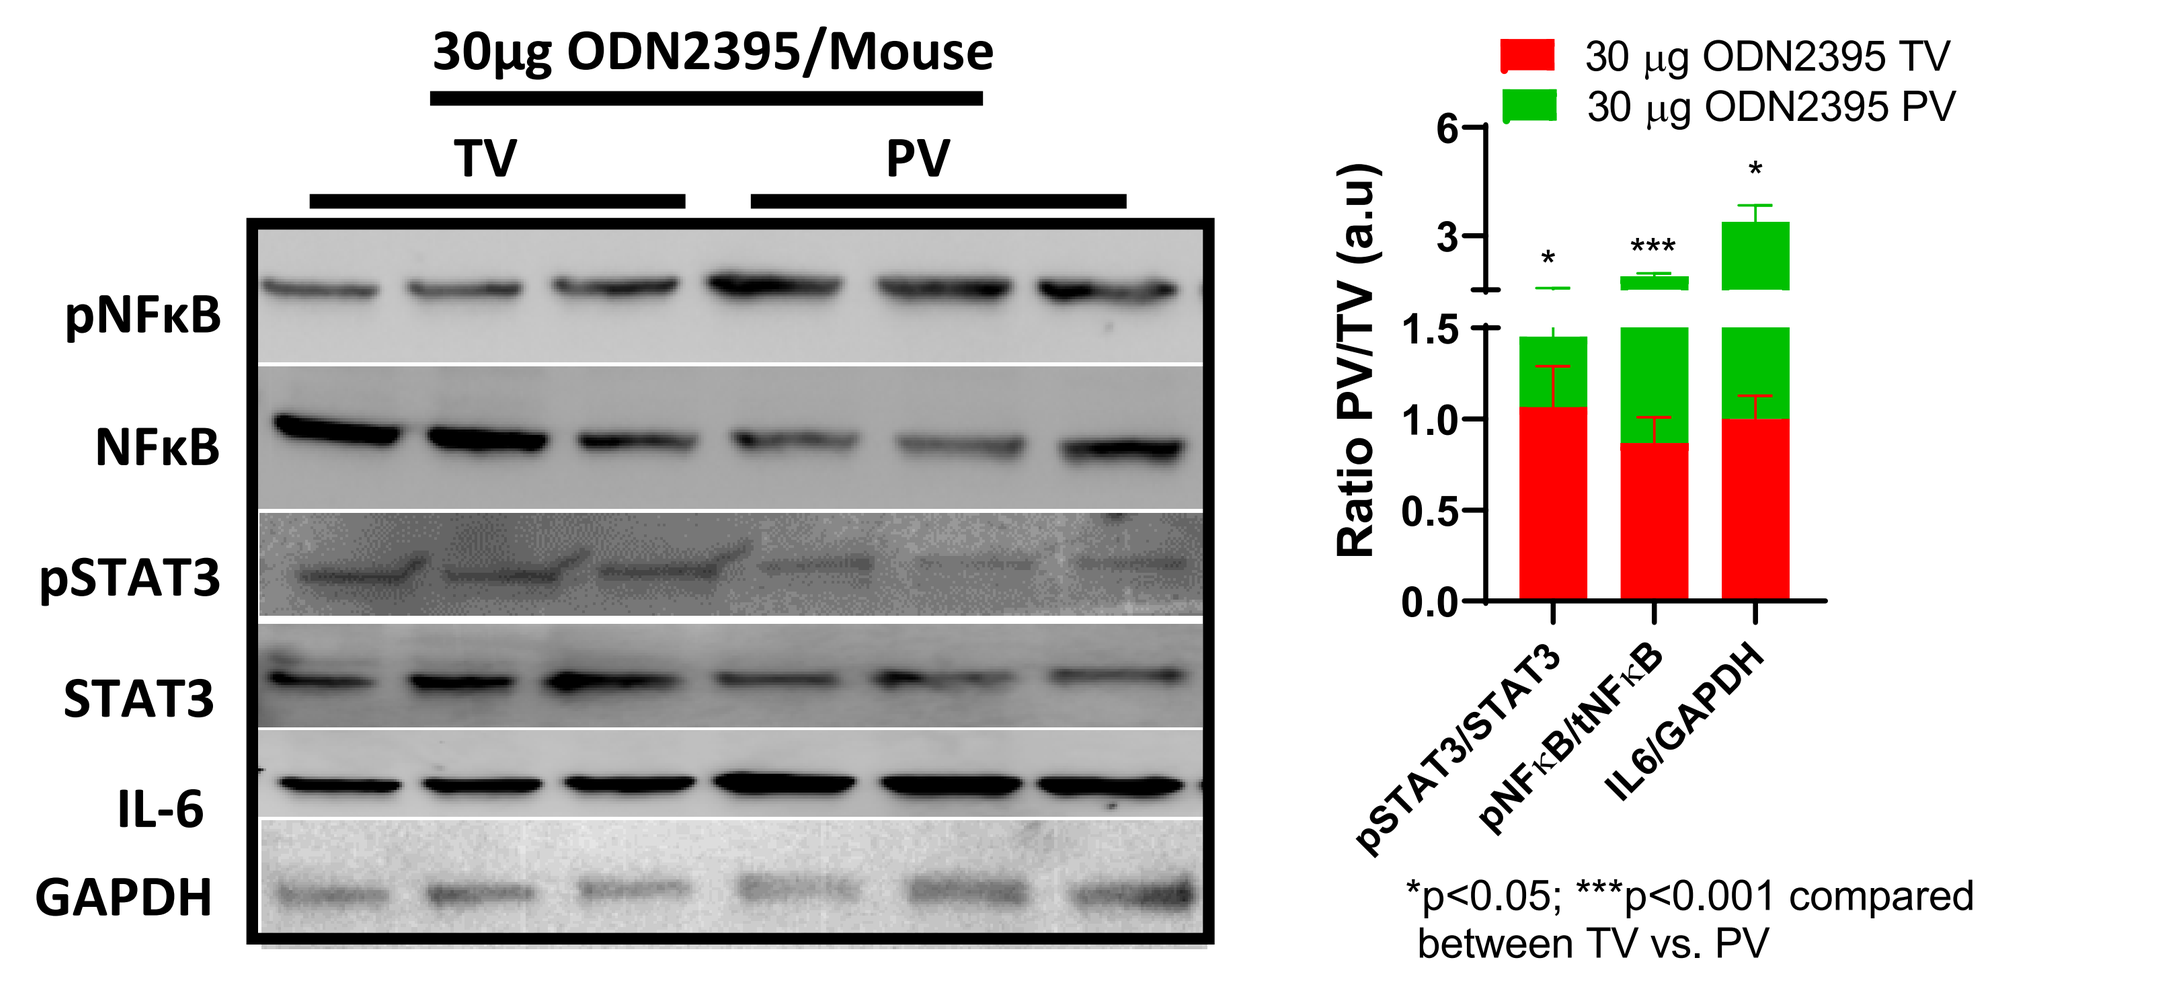
30µg ODN2395/Mouse
TV
PV
pNFκB
NFκB
pSTAT3
STAT3
IL-6
GAPDH
30 µg ODN2395 TV
30 µg ODN2395 PV
Ratio PV/TV (a.u)
pSTAT3/STAT3
pNFκB/tNFκB
IL6/GAPDH
*p<0.05; ***p<0.001 compared between TV vs. PV

Figure 4: As per schema, mice (n=3/group) were sacrificed after two days post-treatment. **A.** The LM tissues were evaluated for pNFκB ($p65^{S536}$), $pSTAT3^{Y705}$, total NFκB, STAT3, and IL-6 by western blotting. GAPDH was used as a housekeeping protein control. **B.** Densitometric calculation. Student's t-test was performed (*p<0.05, ***p<0.001, n=6).

Figure 5. ODN2395 activates NFκB signaling dependent on TLR9-mediated activation

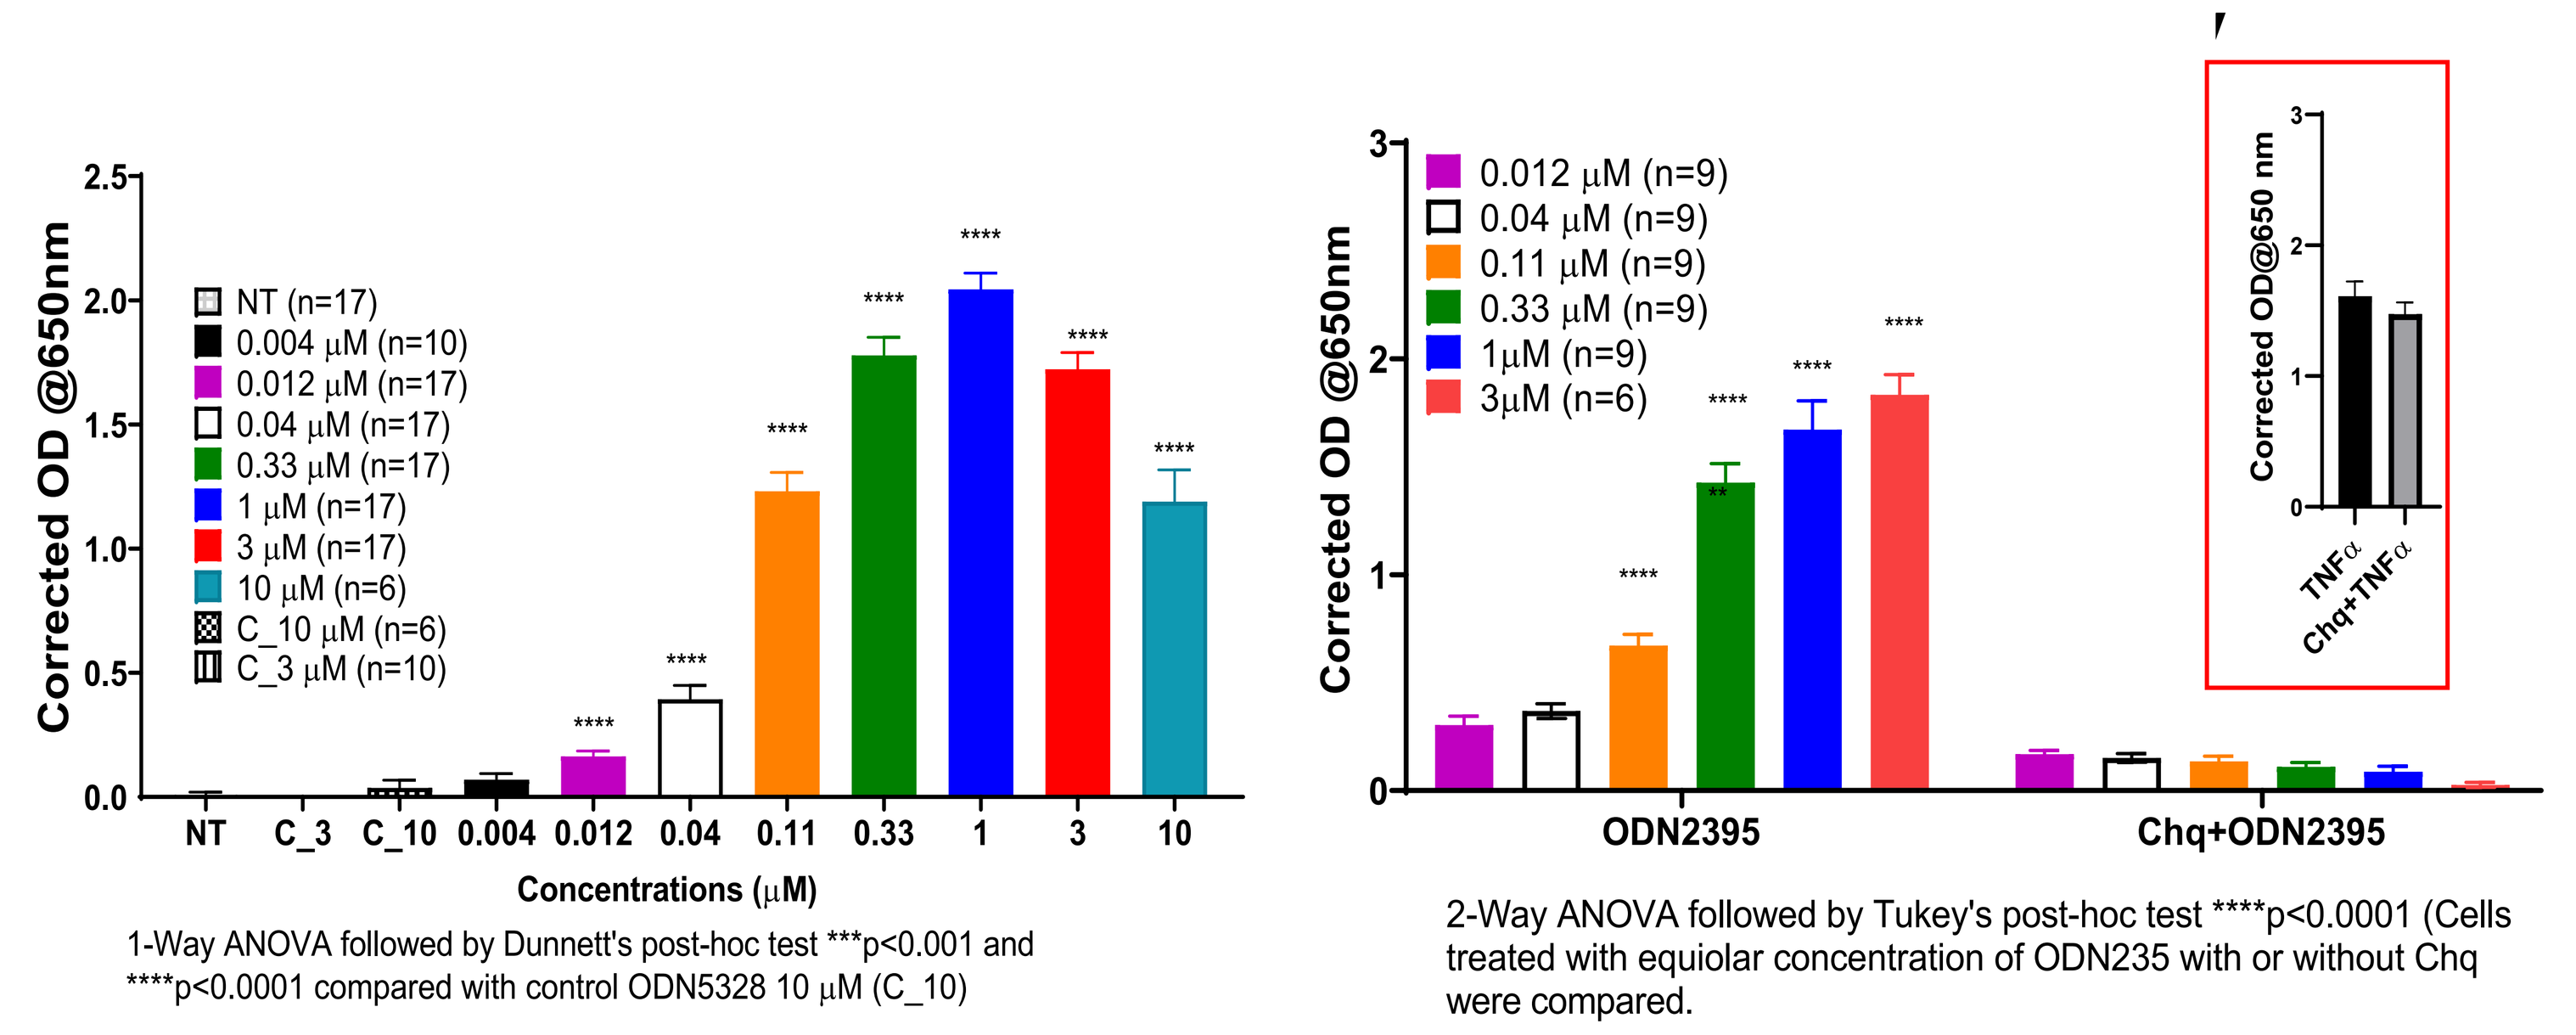
Corrected OD @650nm
NT (n=17)
0.004 µM (n=10)
0.012 µM (n=17)
0.04 µM (n=17)
0.33 µM (n=17)
1 µM (n=17)
3 µM (n=17)
10 µM (n=6)
C_10 µM (n=6)
C_3 µM (n=10)
NT C_3 C_10 0.004 0.012 0.04 0.11 0.33 1 3 10
Concentrations (µM)
1-Way ANOVA followed by Dunnett's post-hoc test ***p<0.001 and ****p<0.0001 compared with control ODN5328 10 µM (C_10)
0.012 µM (n=9)
0.04 µM (n=9)
0.11 µM (n=9)
0.33 µM (n=9)
1µM (n=9)
3µM (n=6)
ODN2395
Chq+ODN2395
2-Way ANOVA followed by Tukey's post-hoc test ****p<0.0001 (Cells treated with equiolar concentration of ODN235 with or without Chq were compared.
Corrected OD@650 nm
TNFα
Chq+TNFα

Figure 5: In this reporter-based assay, HEK293-Blue cells were treated with ODN2395 and SD101 at increasing doses (0.004-10 µM) for 21 hours. **A.** The released SEAP (secreted embryonic alkaline phosphatase) was determined by measuring the absorbance at 650 nm. **B.** Cells were pre-treated with chloroquine (1µg/ml) for 45 minutes before the addition of ODN2395 at increasing concentrations (0.012-3 µM) for 21 hours and absorbance at 650 nm was measured. Absorbance at 650 nm of cells treated with TNFα with/without Chq pretreatment is shown in the **Inset.**

Conclusion

- *In vivo* PV delivery of 30µg of ODN2395 caused **decreased** tumor burden, **decreased** the frequency of MDSCs (predominantly the more immunosuppressive M-MDSCs subpopulation) and **enhanced** pro-inflammatory/anti-tumorigenic M1 macrophages and concomitant decrease in immunosuppressive M2 macrophage.
- At the molecular level in the tissue, ODN2395 **increased** the phosphorylation of NFκB along with IL6 expression and **decreased** phosphorylation of STAT3. *In vitro* SEAP assay confirmed that ODN2395-mediated NFκB activation is TLR9 dependent.

Overall, our data suggest that the regional delivery of a TLR9A has the potential to provide superior modulation of the TME within the liver.

Contact: chandra.ghosh@chartercare.org

MIO-0004 R01