Checkpoint inhibitor administered either systemically or subcutaneously enhances the anti-tumor immunity of SD-101 delivered by Pressure-enabled Drug Delivery (PEDD) device in treating Liver Metastasis (LM)

TriSalus LIFE SCIENCES®

Chandra C. Ghosh[1], Lauren Counoyer[2], Jennie Yujia Liu[1], Alizee Ballarin[1], Prajna Guha[1], Bryan F. Cox[1], Steven C. Katz[1,2]

[1]TriSalus Life Sciences, Westminster, CO-80031; [2]Department of Surgery, Brown University School of Medicine, Providence-02912

Abstract #1123

Abstract

Background: The success of checkpoint inhibitors (CPIs) in treating LM is limited due to liver's inherent immunosuppressive nature, critically contributed by Myeloid-derived suppressive cells (MDSCs). SD-101, a TLR9 receptor agonist delivered by using PEDD device promotes an immune-active anti-tumor microenvironment. Use of CPI in combination with the regionally delivered SD-101 enhances anti-tumor efficacy without a complete mechanistic understanding. Currently, CPIs are delivered intravenously which is inconvenient, costly and time consuming. Thus, there is a growing interest in developing alternate routes of administration. In this study we investigated whether anti-PD-1 delivered via subcutaneous (SQ) route is as effective as the systemic delivery and provide equivalent survival benefit to LM bearing mice.

Methods*:* LM model was developed by inoculating MC38-Luc cells via the spleen of 8-12 weeks old male C57/BL6 mice followed by splenectomy. After a week, fluorescently labelled SD-101 (10µg/mouse) was delivered by using PEDD with anti-PD-1 delivered either via SQ or Intraperitoneally (Sys). Anti-PD-1 was delivered, and tumor burden was monitored by In Vivo Imaging System. Circulatory levels of pro-inflammatory cytokines were analyzed by using Luminex. Tissues were harvested on D3 or D10 to isolate CD45+ cells. RNA isolated from these cells on D3 was used for NanoString analysis and cells isolated from D10 were used for flow cytometry (FC). For Nanostring analysis, the innate immune panels and for FC, MDSCs ($CD11b^+Gr1^+$), B cells ($B220^+$), T ($CD3^+$) cells and M1 ($F4/80^+CD38^+Egr2^-$) macrophages were quantified.

Results: SD-101 delivered via PEDD in combination with anti-PD-1 antibody delivered SQ or Sys significantly reduced LM progression (Figure 1). Moreover, reduction of MDSCs with increase in B, T, and M1 macrophages within the LM were observed, irrespective of the routes of delivery. The pro-inflammatory cytokines such as IFNγ and IP10 significantly increased in the circulation of mice that received SD-101 as compared to the vehicle control. Nanostring analysis revealed that mono- and combination therapies inhibited myeloid cell differentiation and maintenance, angiogenesis, and increased cytokine, lymphocyte activation and TLR signaling pathways. Interestingly, combination of SD-101 and anti-PD1 irrespective of the routes of delivery enhanced the survival of mice as compared to monotherapy and Veh control.

Conclusion: SD-101 administered regionally via PEDD as monotherapy resolved the tumor progression in mice with LM which was potentiated by combining anti-PD-1 administered via SQ or Sys and enhanced the survival of LM bearing mice, irrespective of the route of delivery.

Introduction and Methods

- ***SD-101, a class C TLR9 receptor agonist administration by PEDD promotes MDSC depletion and broad immune stimulation, in association with encouraging clinical outcomes in combination with CPI.***
- ***Currently, CPIs are delivered intravenously and there is a growing interest in subcutaneous (SQ) administration.***
- ***We compared PEDD-SD-101 in combination with Sys or SQ CPI in a murine LM model.***

Figure 1: PEDD-SD-101 effect on LM growth was enhanced by CPI irrespective of the delivery route

A. Schema

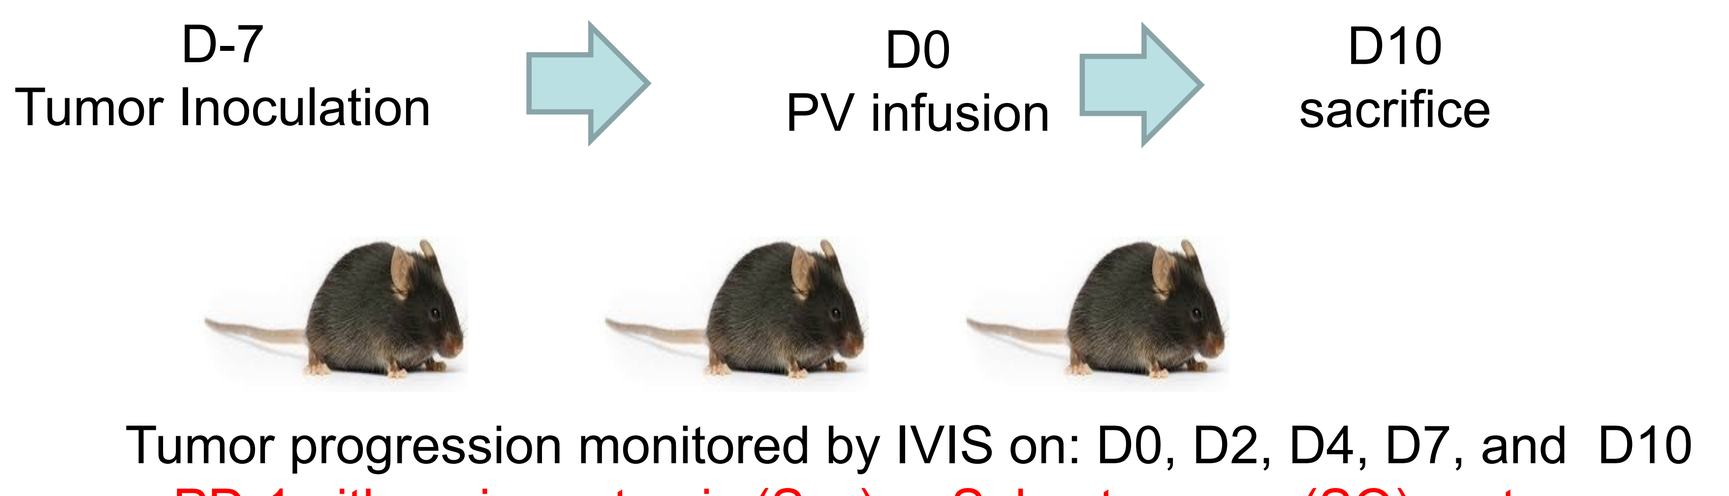

D-7
Tumor Inoculation
D0
PV infusion
D10
sacrifice

Tumor progression monitored by IVIS on: D0, D2, D4, D7, and D10
α-PD-1 either via systemic (Sys) or Subcutaneous (SQ) routes

B. Tumor Burden

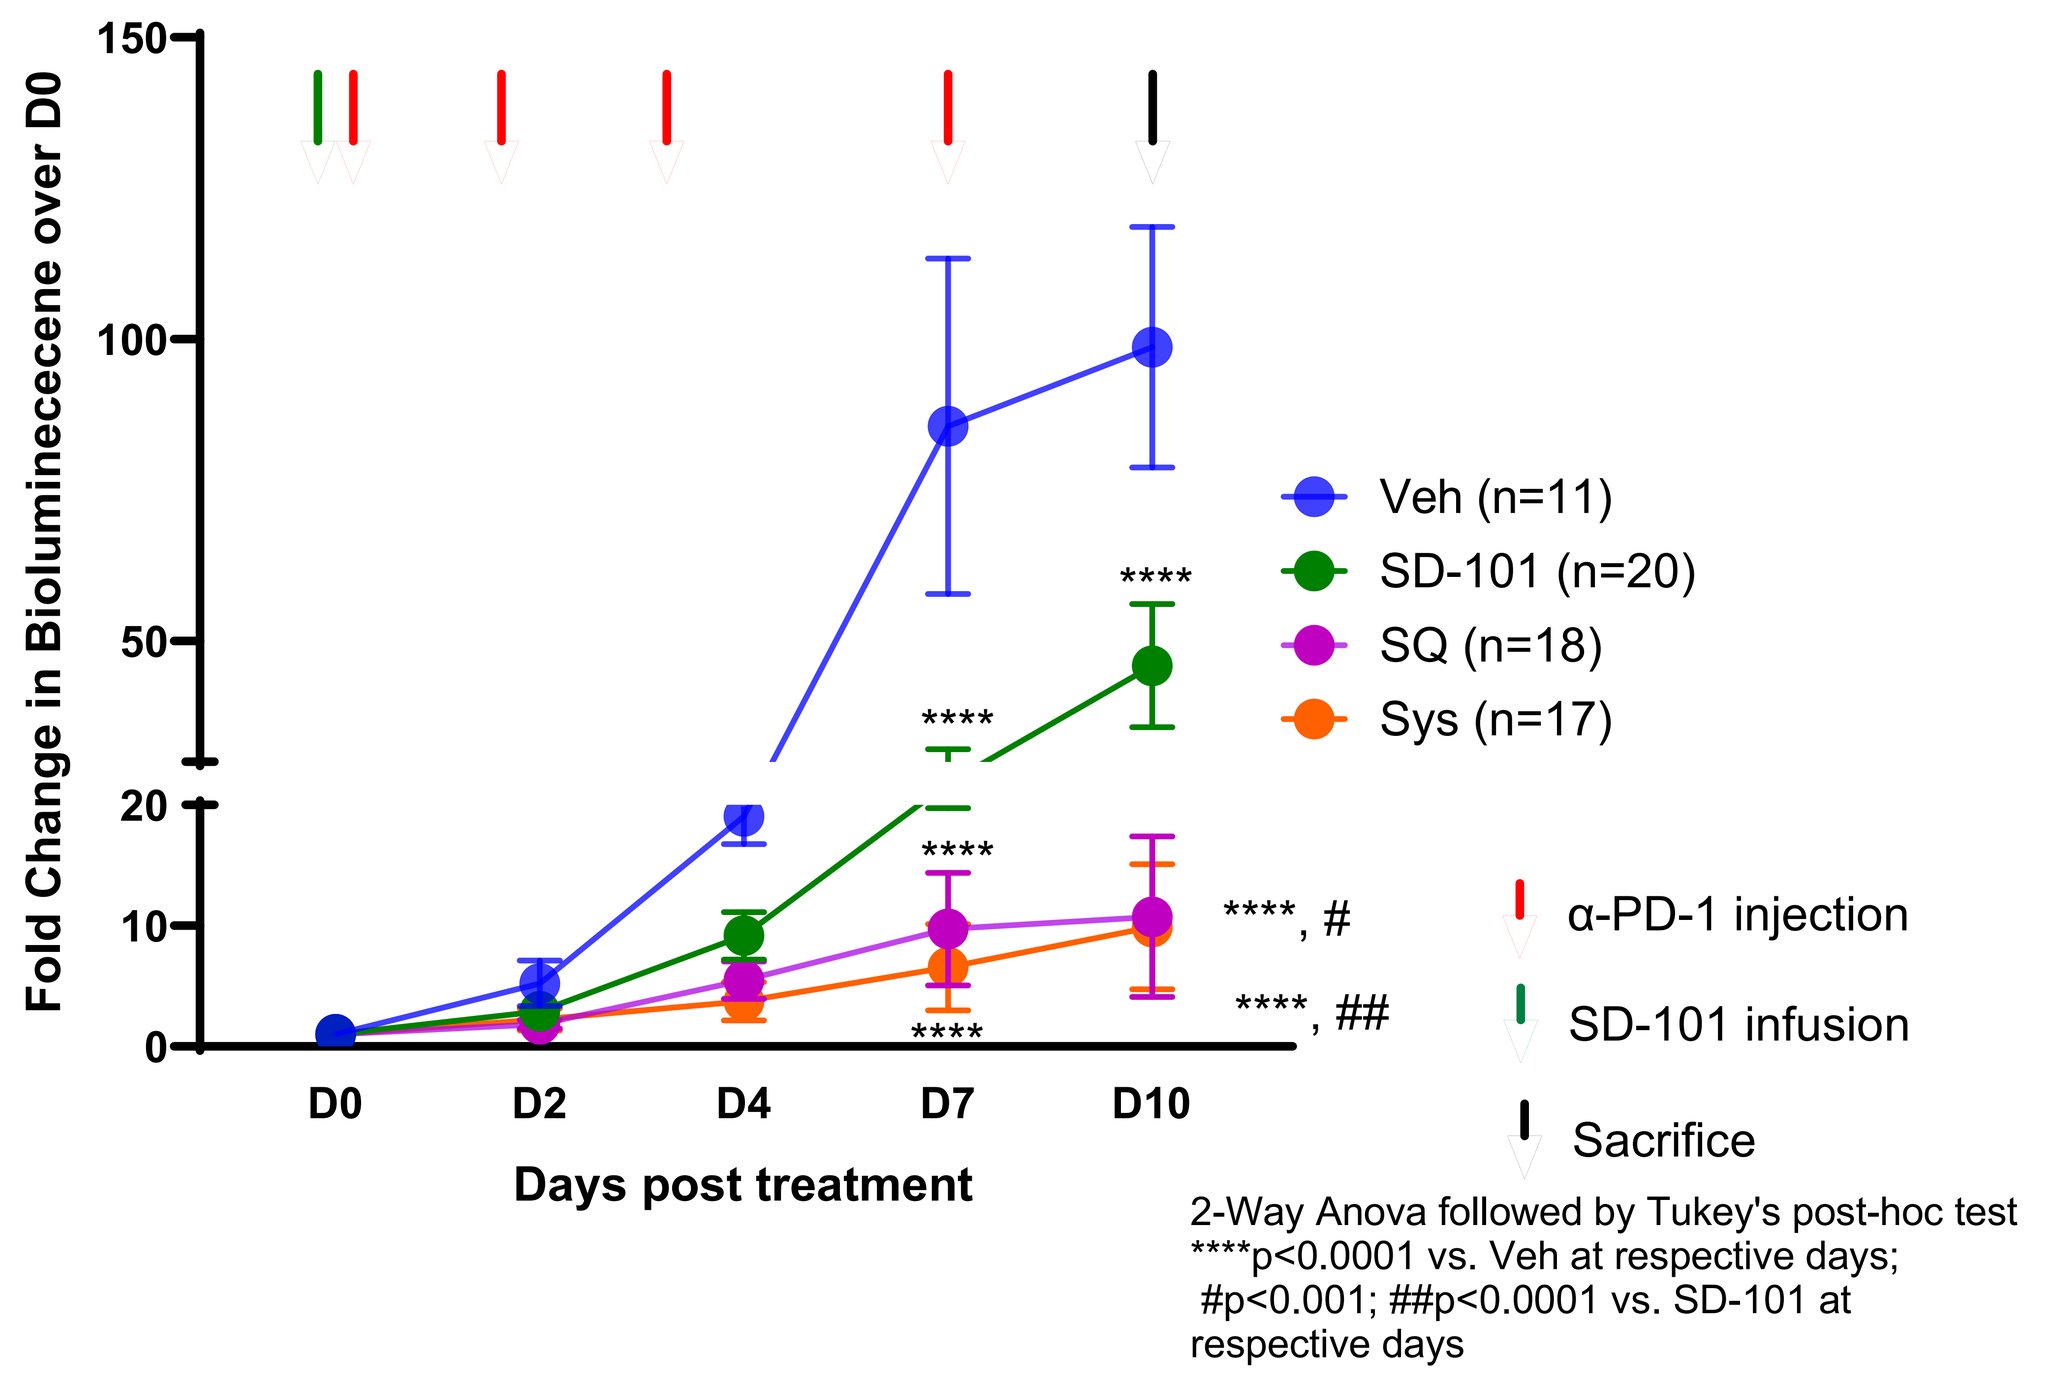

Fold Change in Bioluminececene over D0
Days post treatment
Veh (n=11)
SD-101 (n=20)
SQ (n=18)
Sys (n=17)
****, #
****, ##
α-PD-1 injection
SD-101 infusion
Sacrifice

2-Way Anova followed by Tukey's post-hoc test ****p<0.0001 vs. Veh at respective days; #p<0.001; ##p<0.0001 vs. SD-101 at respective days

C. Bioluminescence image

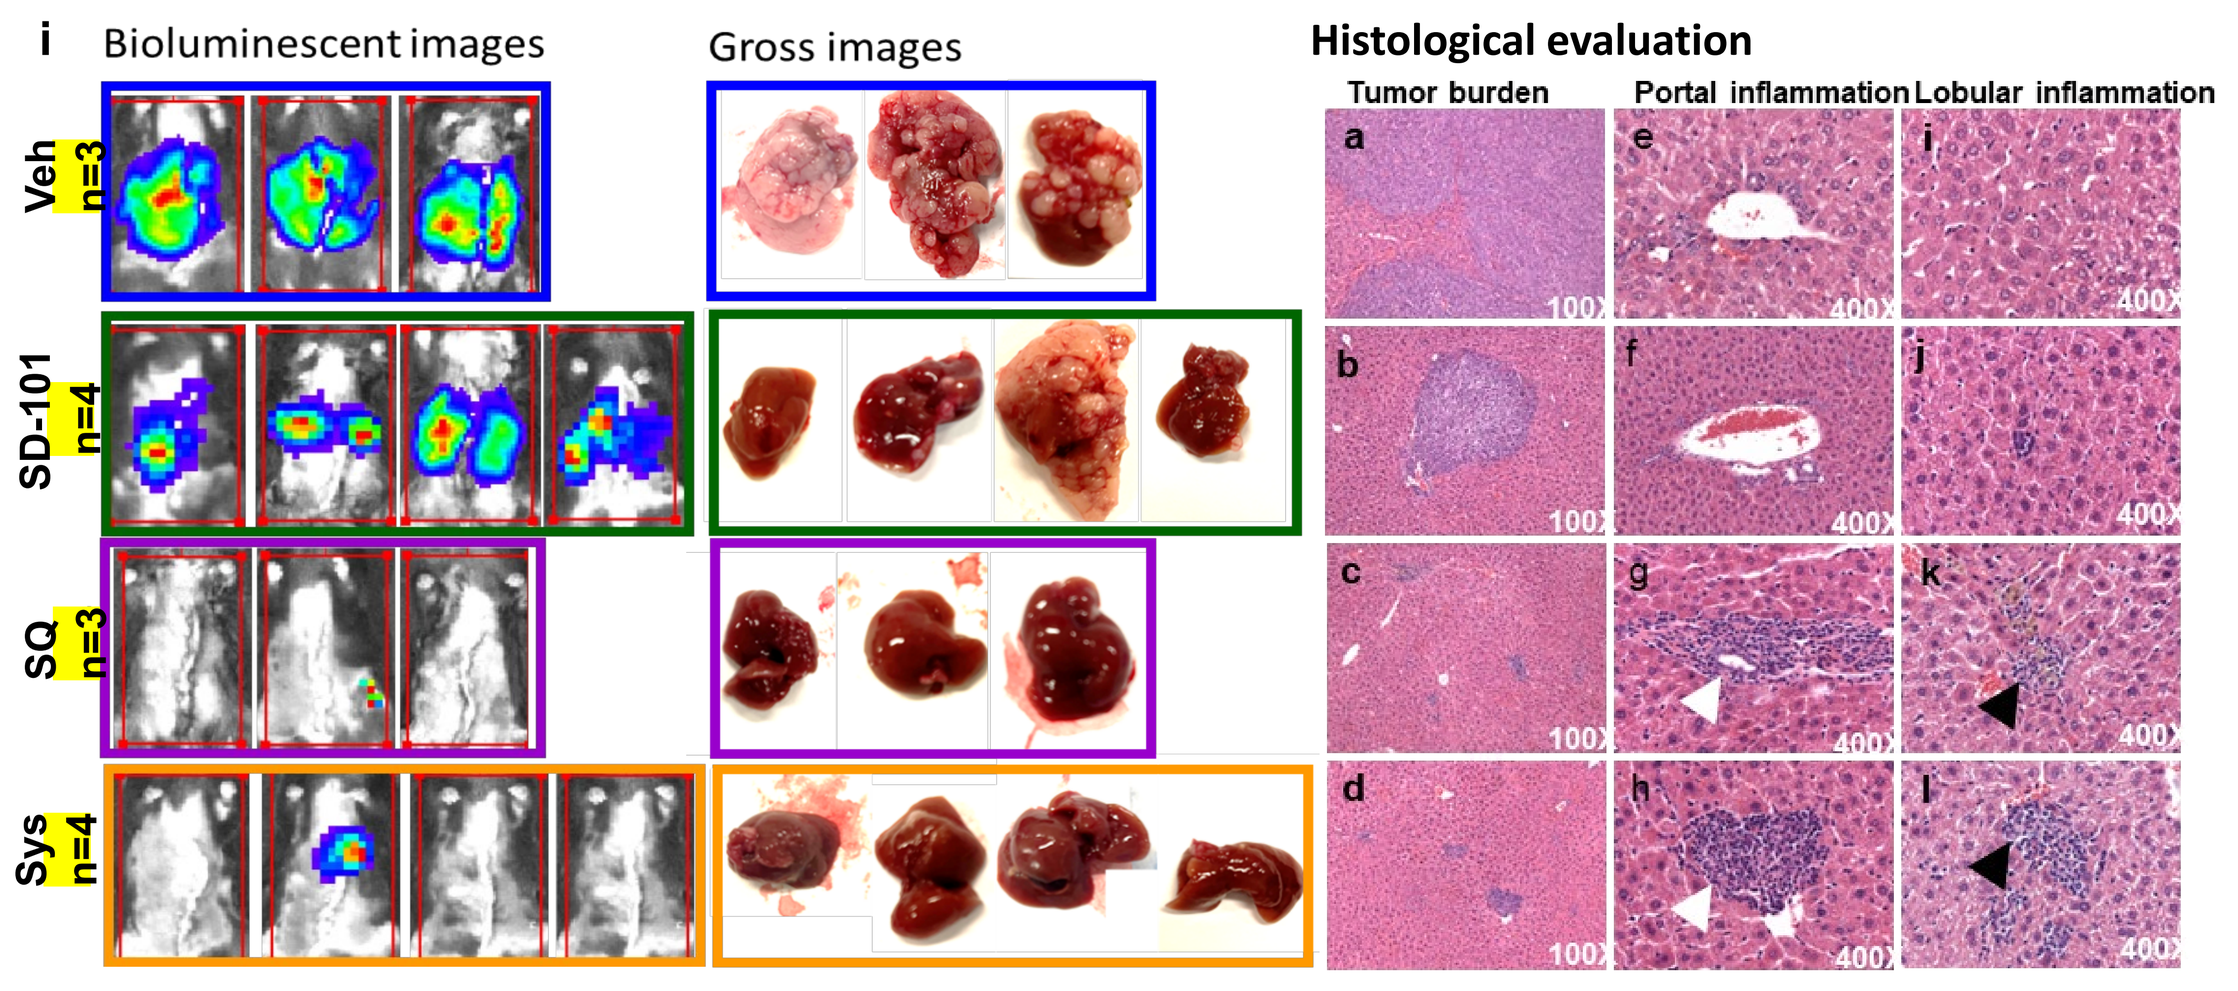

i
Bioluminescent images
Gross images
Histological evaluation
Tumor burden
Portal inflammation
Lobular inflammation
Veh n=3
SD-101 n=4
SQ n=3
Sys n=4

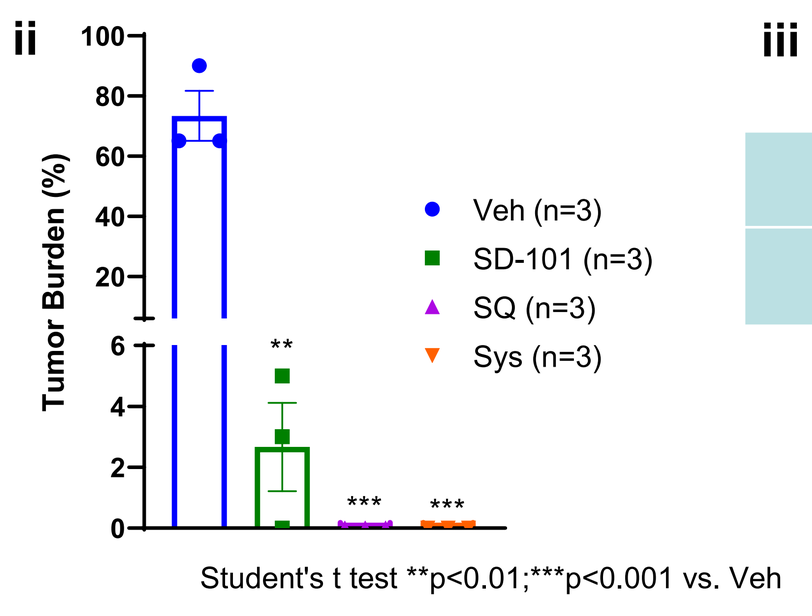

ii
Tumor Burden (%)
Veh (n=3)
SD-101 (n=3)
SQ (n=3)
Sys (n=3)
Student's t test **p<0.01;***p<0.001 vs. Veh

iii

	Veh (n=3)	SD-101 (n=3)	SQ (n=3)	Sys (n=3)
Portal tract inflammation	0	0	3	3
Lobular inflammation	0	2	3	3

Figure 1: A. Schema: Eight- to twelve-week-old male C57/BL6 mice were inoculated intra-splenic with 1.0e⁶ MC38-Luc cells for a week. **B.** Bioluminescence value was determined by IVIS on D0, and mice were randomized accordingly and treated with 10 µg/mouse IRD labelled SD-101 via Portal Vein (PV) along with 25 mg/Kg/mouse administered via SQ or Sys. Tumor burden by bioluminescence was measured on D0, D4, D7 and D10. PBS delivered via PV served as Vehicle control (Veh). Fold change of the tumor burden was calculated based on D0 baseline bioluminescence. **C. i.** Mice were sacrificed on D10 and representative images depicting the bioluminescence on D10, gross and representative H&E images of the harvested livers. (**a-d**) the morphology of tumor bearing liver tissue followed by treatment; (**e-h**) depicting portal tract inflammation (white arrow heads); (**i-l**) demonstrating lobular inflammation (black arrow heads). **ii.** Graphical representation of tumor burden data, **iii.** Table showing the presence of portal tract or lobular inflammations within tumor-bearing liver treated with SD-101 ± α-PD-1, respectively. 2-Way ANOVA with Tukey's multiple comparison test was performed to compare the tumor progression among the groups. Data presented as mean±SEM; n≥3 per group and p value is mentioned in the graph.

Contact: Chandra.ghosh@trisaluslifesci.com

Figure 2: Modulation of liver myeloid and lymphoid compartments by PEDD-SD-101 was preserved in combination with Sys or SQ CPI

A. Gating Strategy

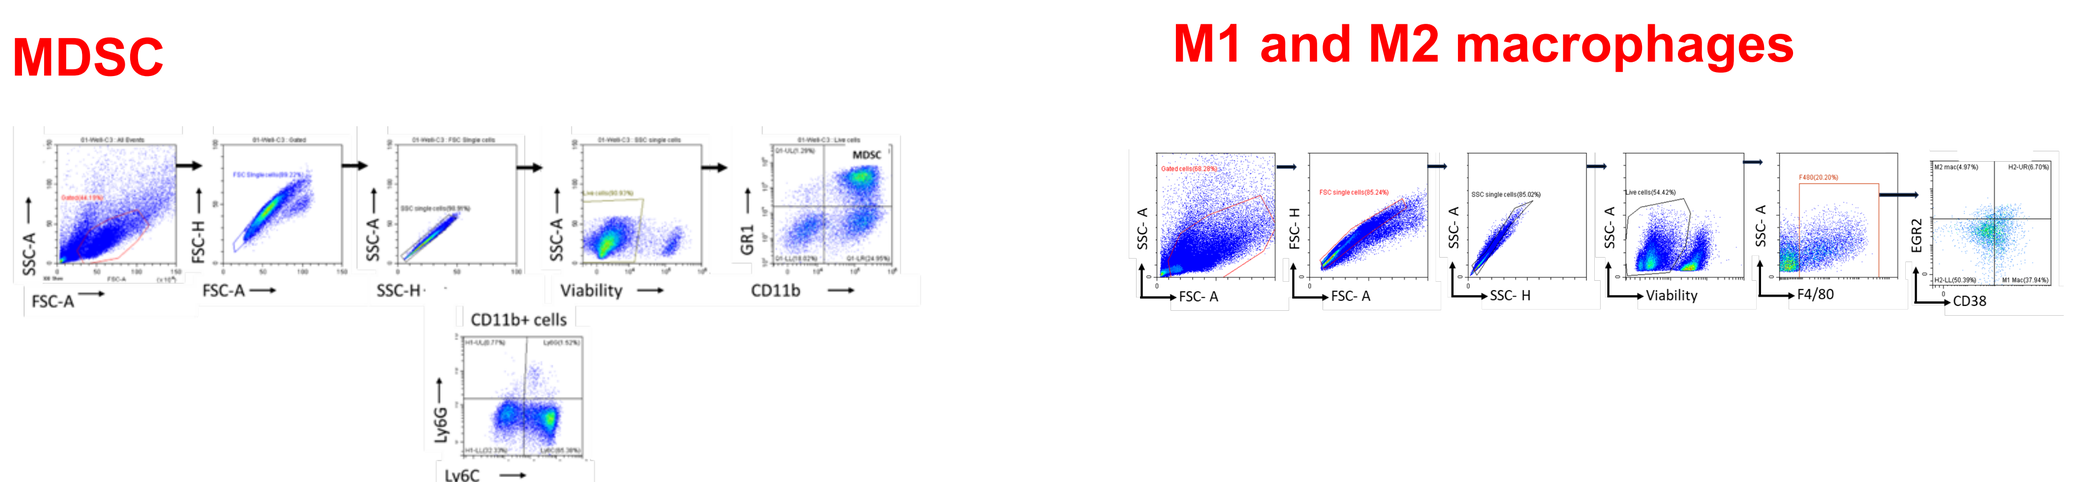

MDSC
M1 and M2 macrophages

B. MDSC

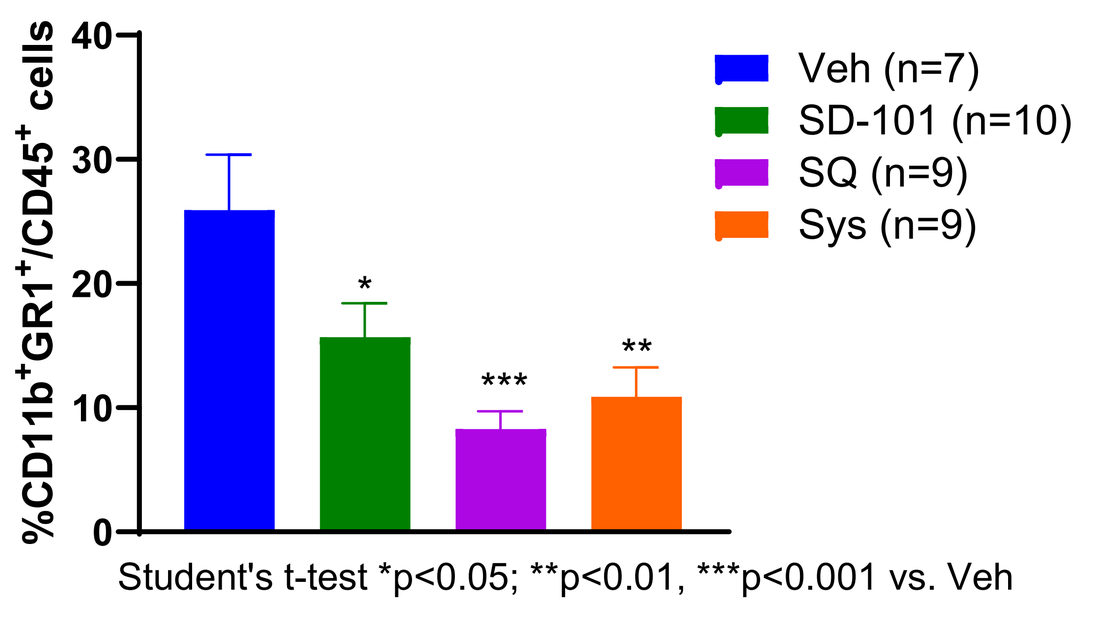

%CD11b⁺GR1⁺/CD45⁺ cells
Veh (n=7)
SD-101 (n=10)
SQ (n=9)
Sys (n=9)
Student's t-test *p<0.05; **p<0.01, ***p<0.001 vs. Veh

C. M-/G-MDSC Ratio

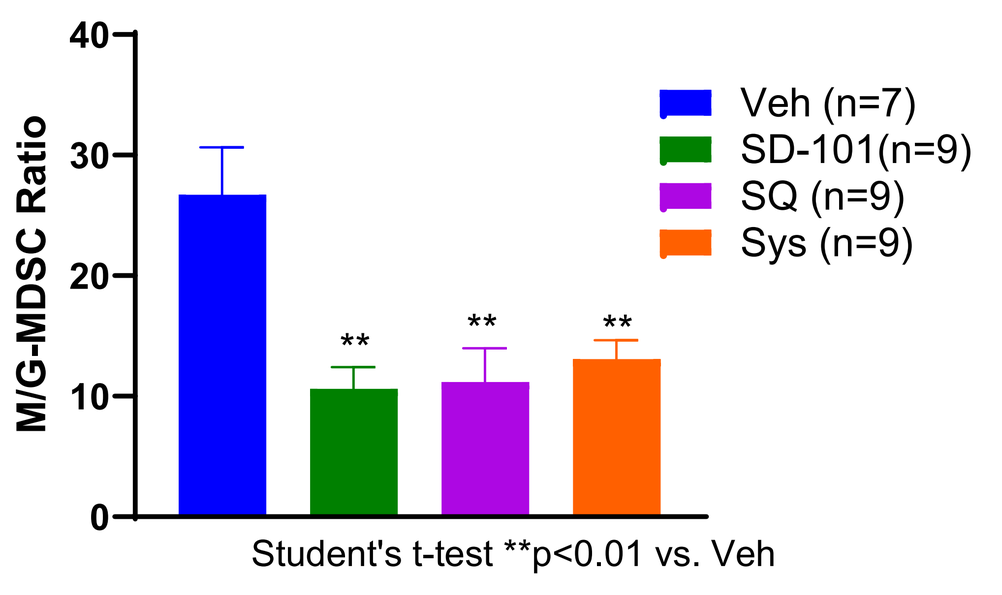

M/G-MDSC Ratio
Veh (n=7)
SD-101(n=9)
SQ (n=9)
Sys (n=9)
Student's t-test **p<0.01 vs. Veh

D. B-Cells

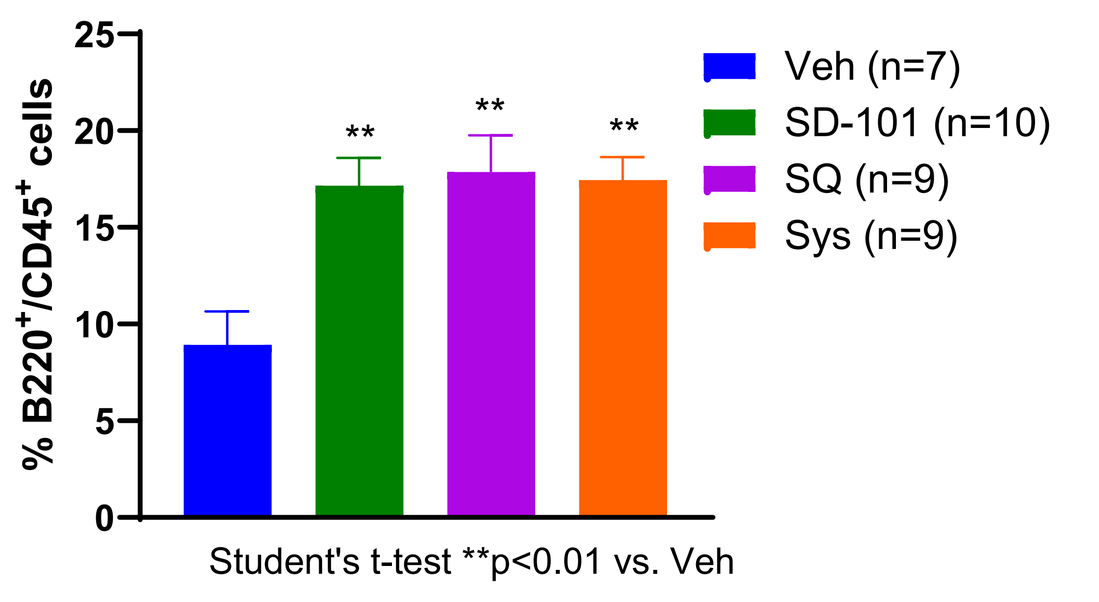

% B220⁺/CD45⁺ cells
Veh (n=7)
SD-101 (n=10)
SQ (n=9)
Sys (n=9)
Student's t-test **p<0.01 vs. Veh

E. CD11c

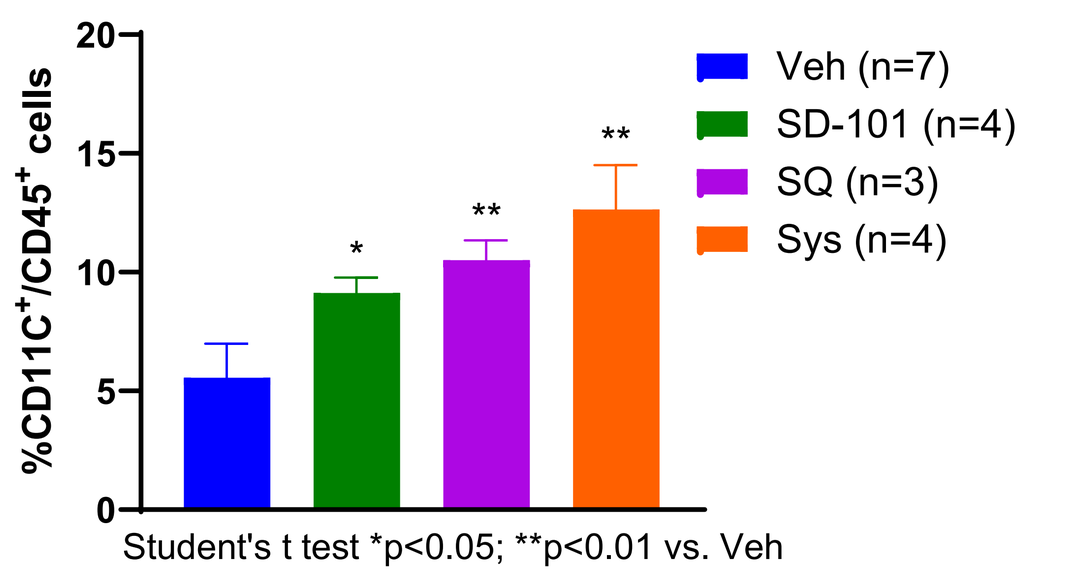

%CD11C⁺/CD45⁺ cells
Veh (n=7)
SD-101 (n=4)
SQ (n=3)
Sys (n=4)
Student's t test *p<0.05; **p<0.01 vs. Veh

F. T-Cells

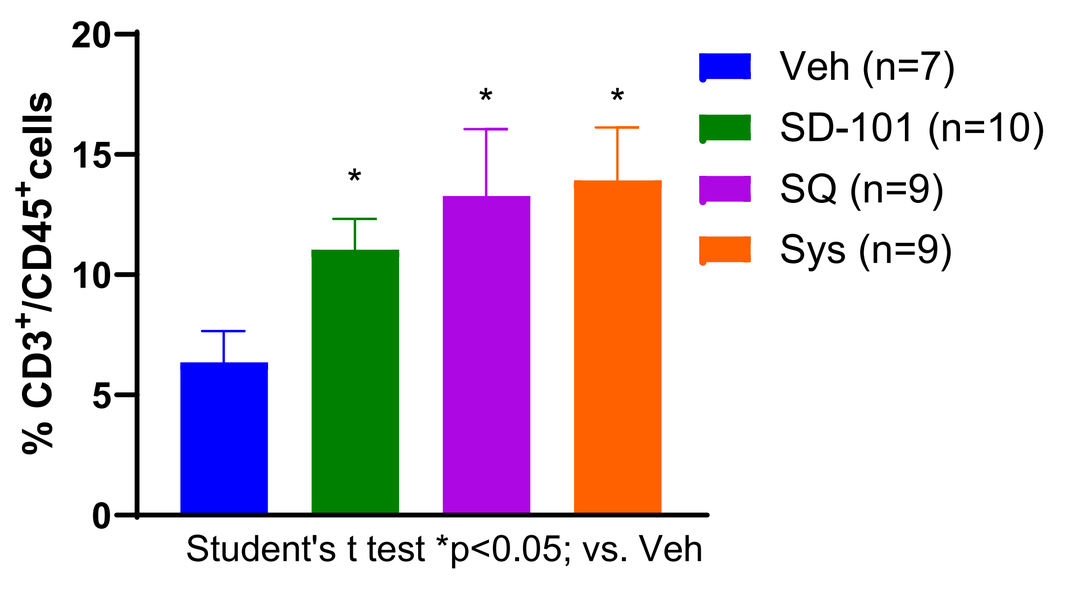

% CD3⁺/CD45⁺cells
Veh (n=7)
SD-101 (n=10)
SQ (n=9)
Sys (n=9)
Student's t test *p<0.05; vs. Veh

G. M1-Macrophages

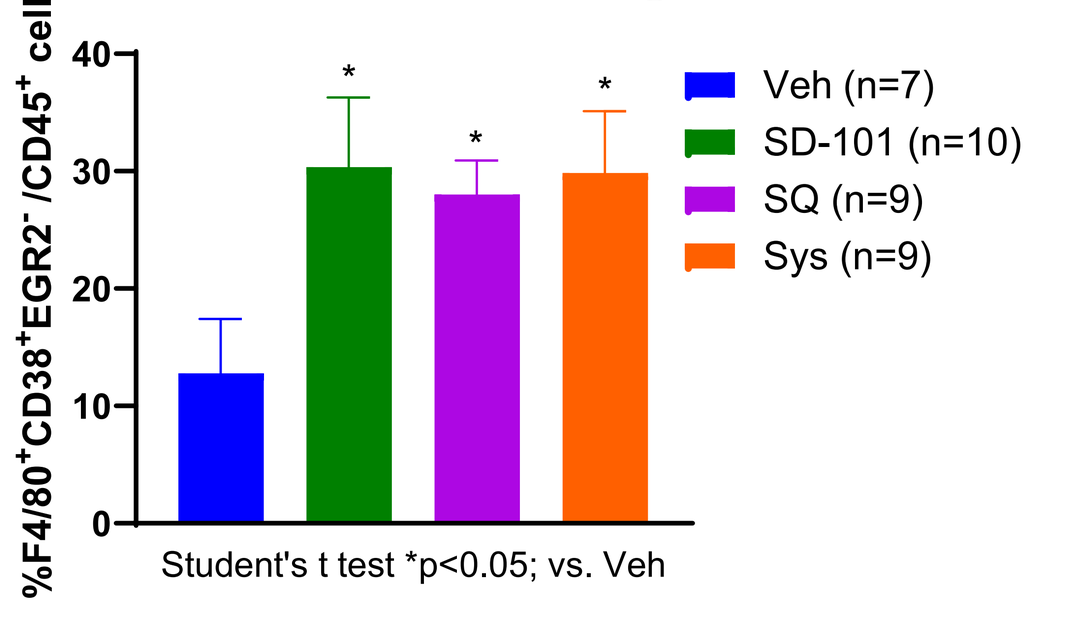

%F4/80⁺CD38⁺EGR2⁻/CD45⁺ cells
Veh (n=7)
SD-101 (n=10)
SQ (n=9)
Sys (n=9)
Student's t test *p<0.05; vs. Veh

Figure 2: Liver of tumor-bearing mice were harvested 10 days post-treatment. CD45+ cells were isolated from non-parenchymal cells (NPCs). A. MDSC cell population (CD11b+Gr1+), B. monocytic MDSCs (M-MDSC; CD11b+Ly6C+/hiLy6G-/lo), C. CD11c+ cells, D. B (B220+) cells, E. T (CD3+) and F. M1 (F/4/80+CD38+EGR2-) cell were quantified by flow cytometry. Data is presented as mean ± SEM. Student's t-test was performed for group-wise comparison and are described in each graph.

Figure 3: Peripheral immunostimulatory effects of PEDD-SD-101 in combination with Sys or SQ α-PD-1

A. IFNγ

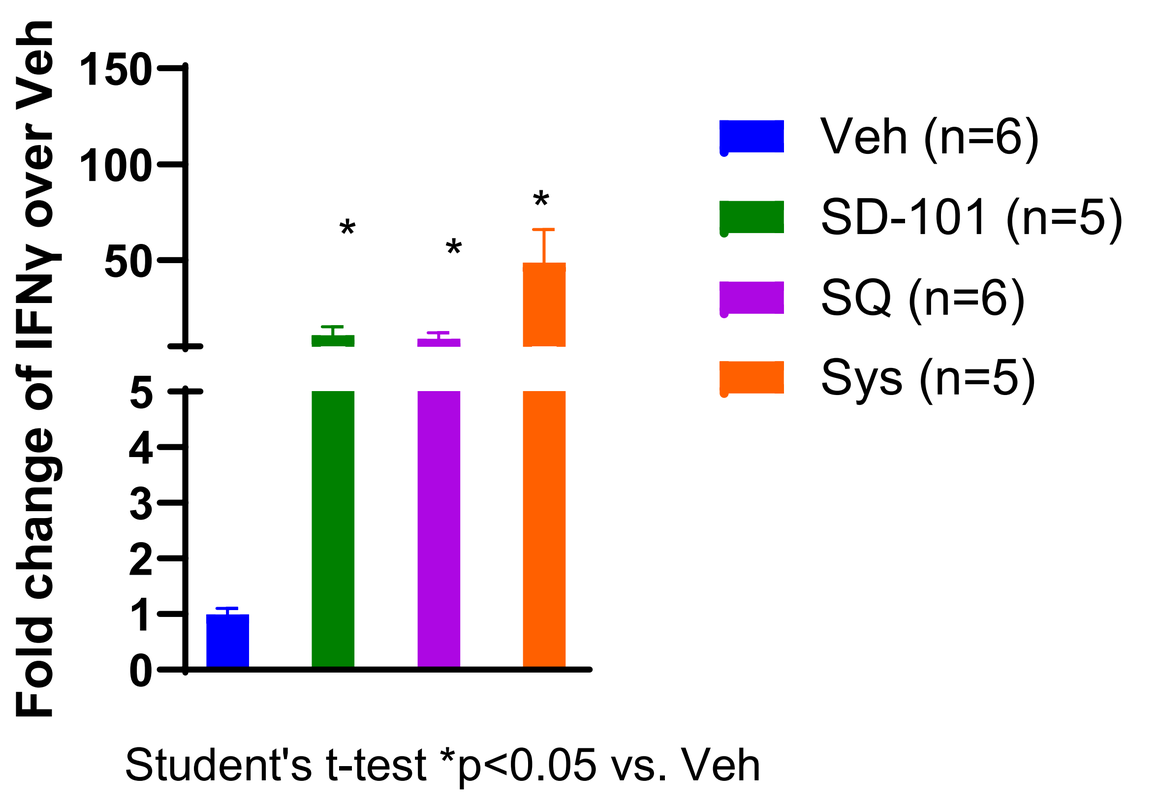

Fold change of IFNγ over Veh
Veh (n=6)
SD-101 (n=5)
SQ (n=6)
Sys (n=5)
Student's t-test *p<0.05 vs. Veh

B. IP-10

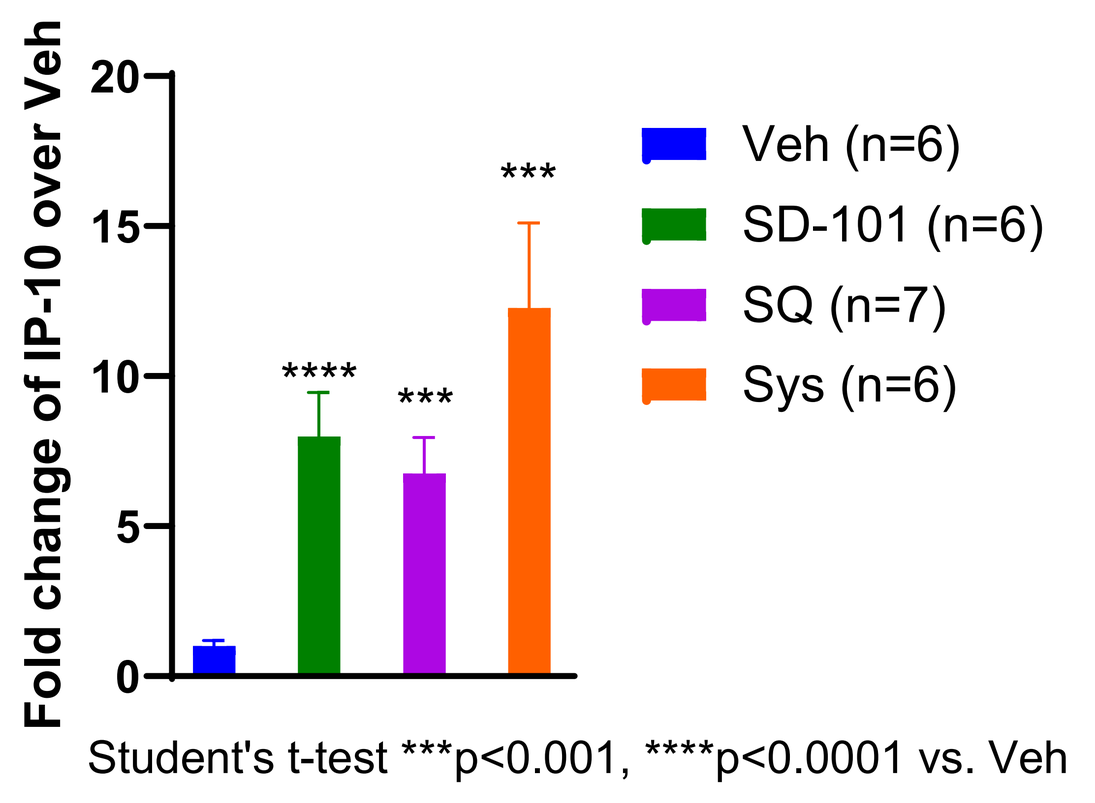

Fold change of IP-10 over Veh
Veh (n=6)
SD-101 (n=6)
SQ (n=7)
Sys (n=6)
Student's t-test ***p<0.001, ****p<0.0001 vs. Veh

Figure 3: The serum collected on D3 was analyzed for IFNγ **(A)** and IP10 **(B)** were measured by Luminex and reported as fold change compared to Veh. Data presented as mean ± SEM. Student's t-test was performed for group-wise comparison and are described in each graph.

Figure 4: PEDD-SD-101 in combination with Sys or SQ α-PD-1 promoted transcriptomic changes consistent with enhancement of anti-tumor immunity in the liver TME

A. Venn Diagram of SQ/Sys/SD-101 vs. Veh

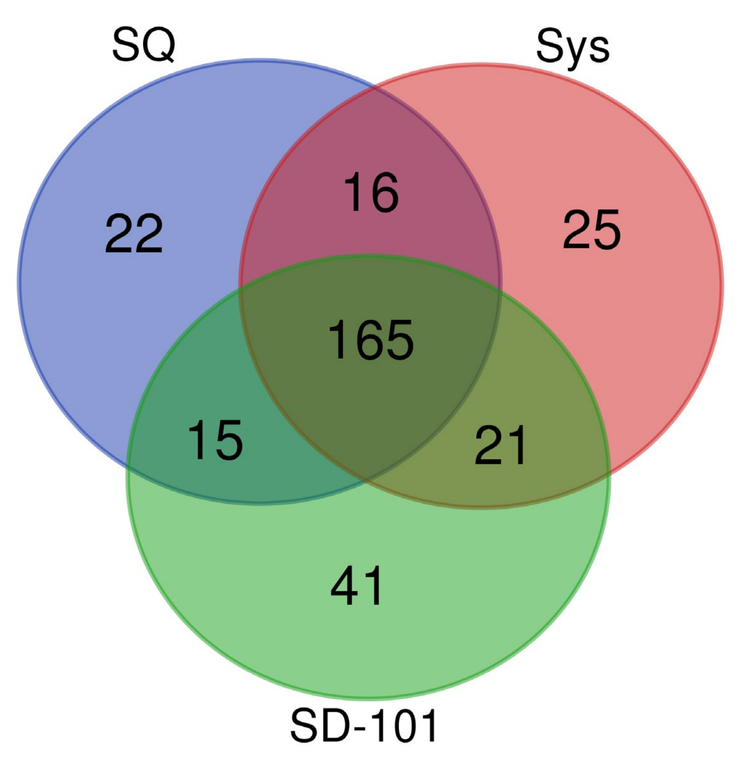

SQ
Sys
SD-101
22
16
25
165
15
21
41

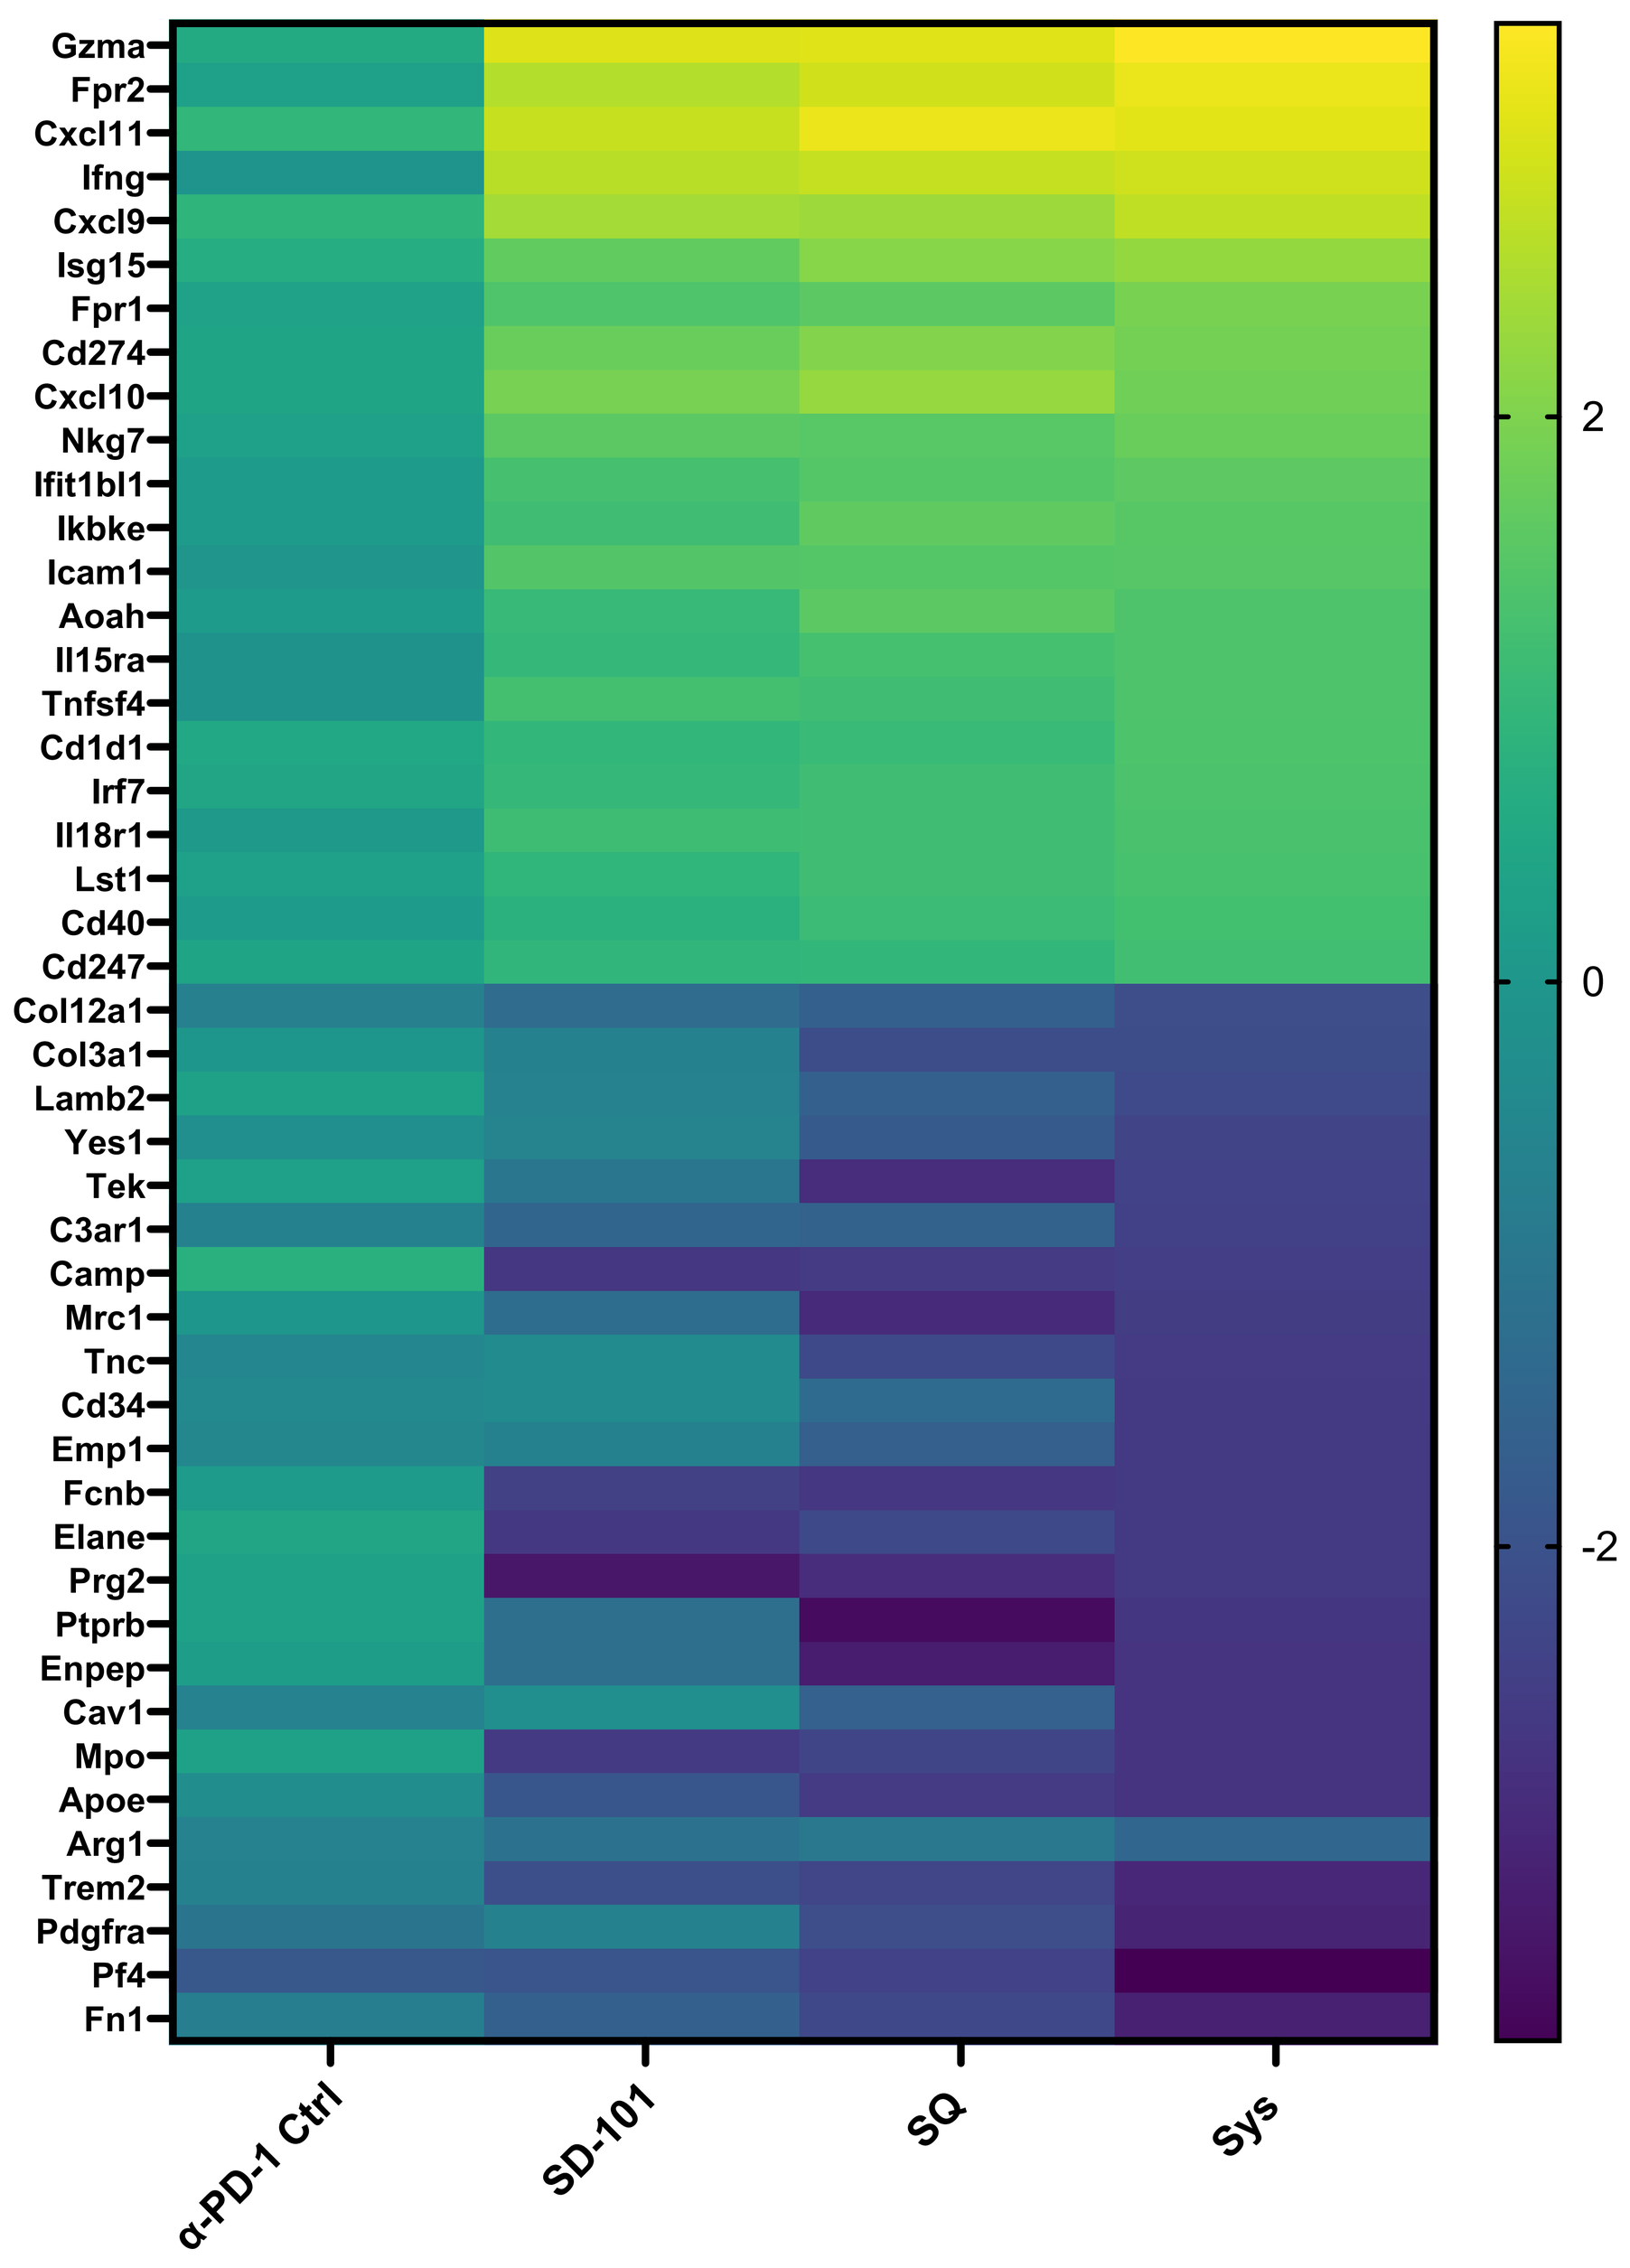

Gzma
Fpr2
Cxcl11
Ifng
Cxcl9
Isg15
Fpr1
Cd274
Cxcl10
Nkg7
Ifit1bl1
Ikbke
Icam1
Aoah
Il15ra
Tnfsf4
Cd1d1
Irf7
Il18r1
Lst1
Cd40
Cd247
Col12a1
Col3a1
Lamb2
Yes1
Tek
C3ar1
Camp
Mrc1
Tnc
Cd34
Emp1
Fcnb
Elane
Prg2
Ptprb
Enpep
Cav1
Mpo
Apoe
Arg1
Trem2
Pdgfra
Pf4
Fn1
α-PD-1 Ctrl
SD-101
SQ
Sys

Figure 4: A. Venn Diagram comparing genes that were significantly modulated by PEDD-SD-101/SQ/Sys compared to Veh **B.** Heat Map of genes that were significantly up-/down-regulated followed by α-PD-1 Ctrl, SD-101, Sys and SQ treatment compared to Veh control

Figure 5. PEDD-SD-101 in combination with Sys or SQ α-PD-1 promoted transcriptomic changes consistent with enhancement of anti-tumor immunity in the liver TME

A. Pathway scoring

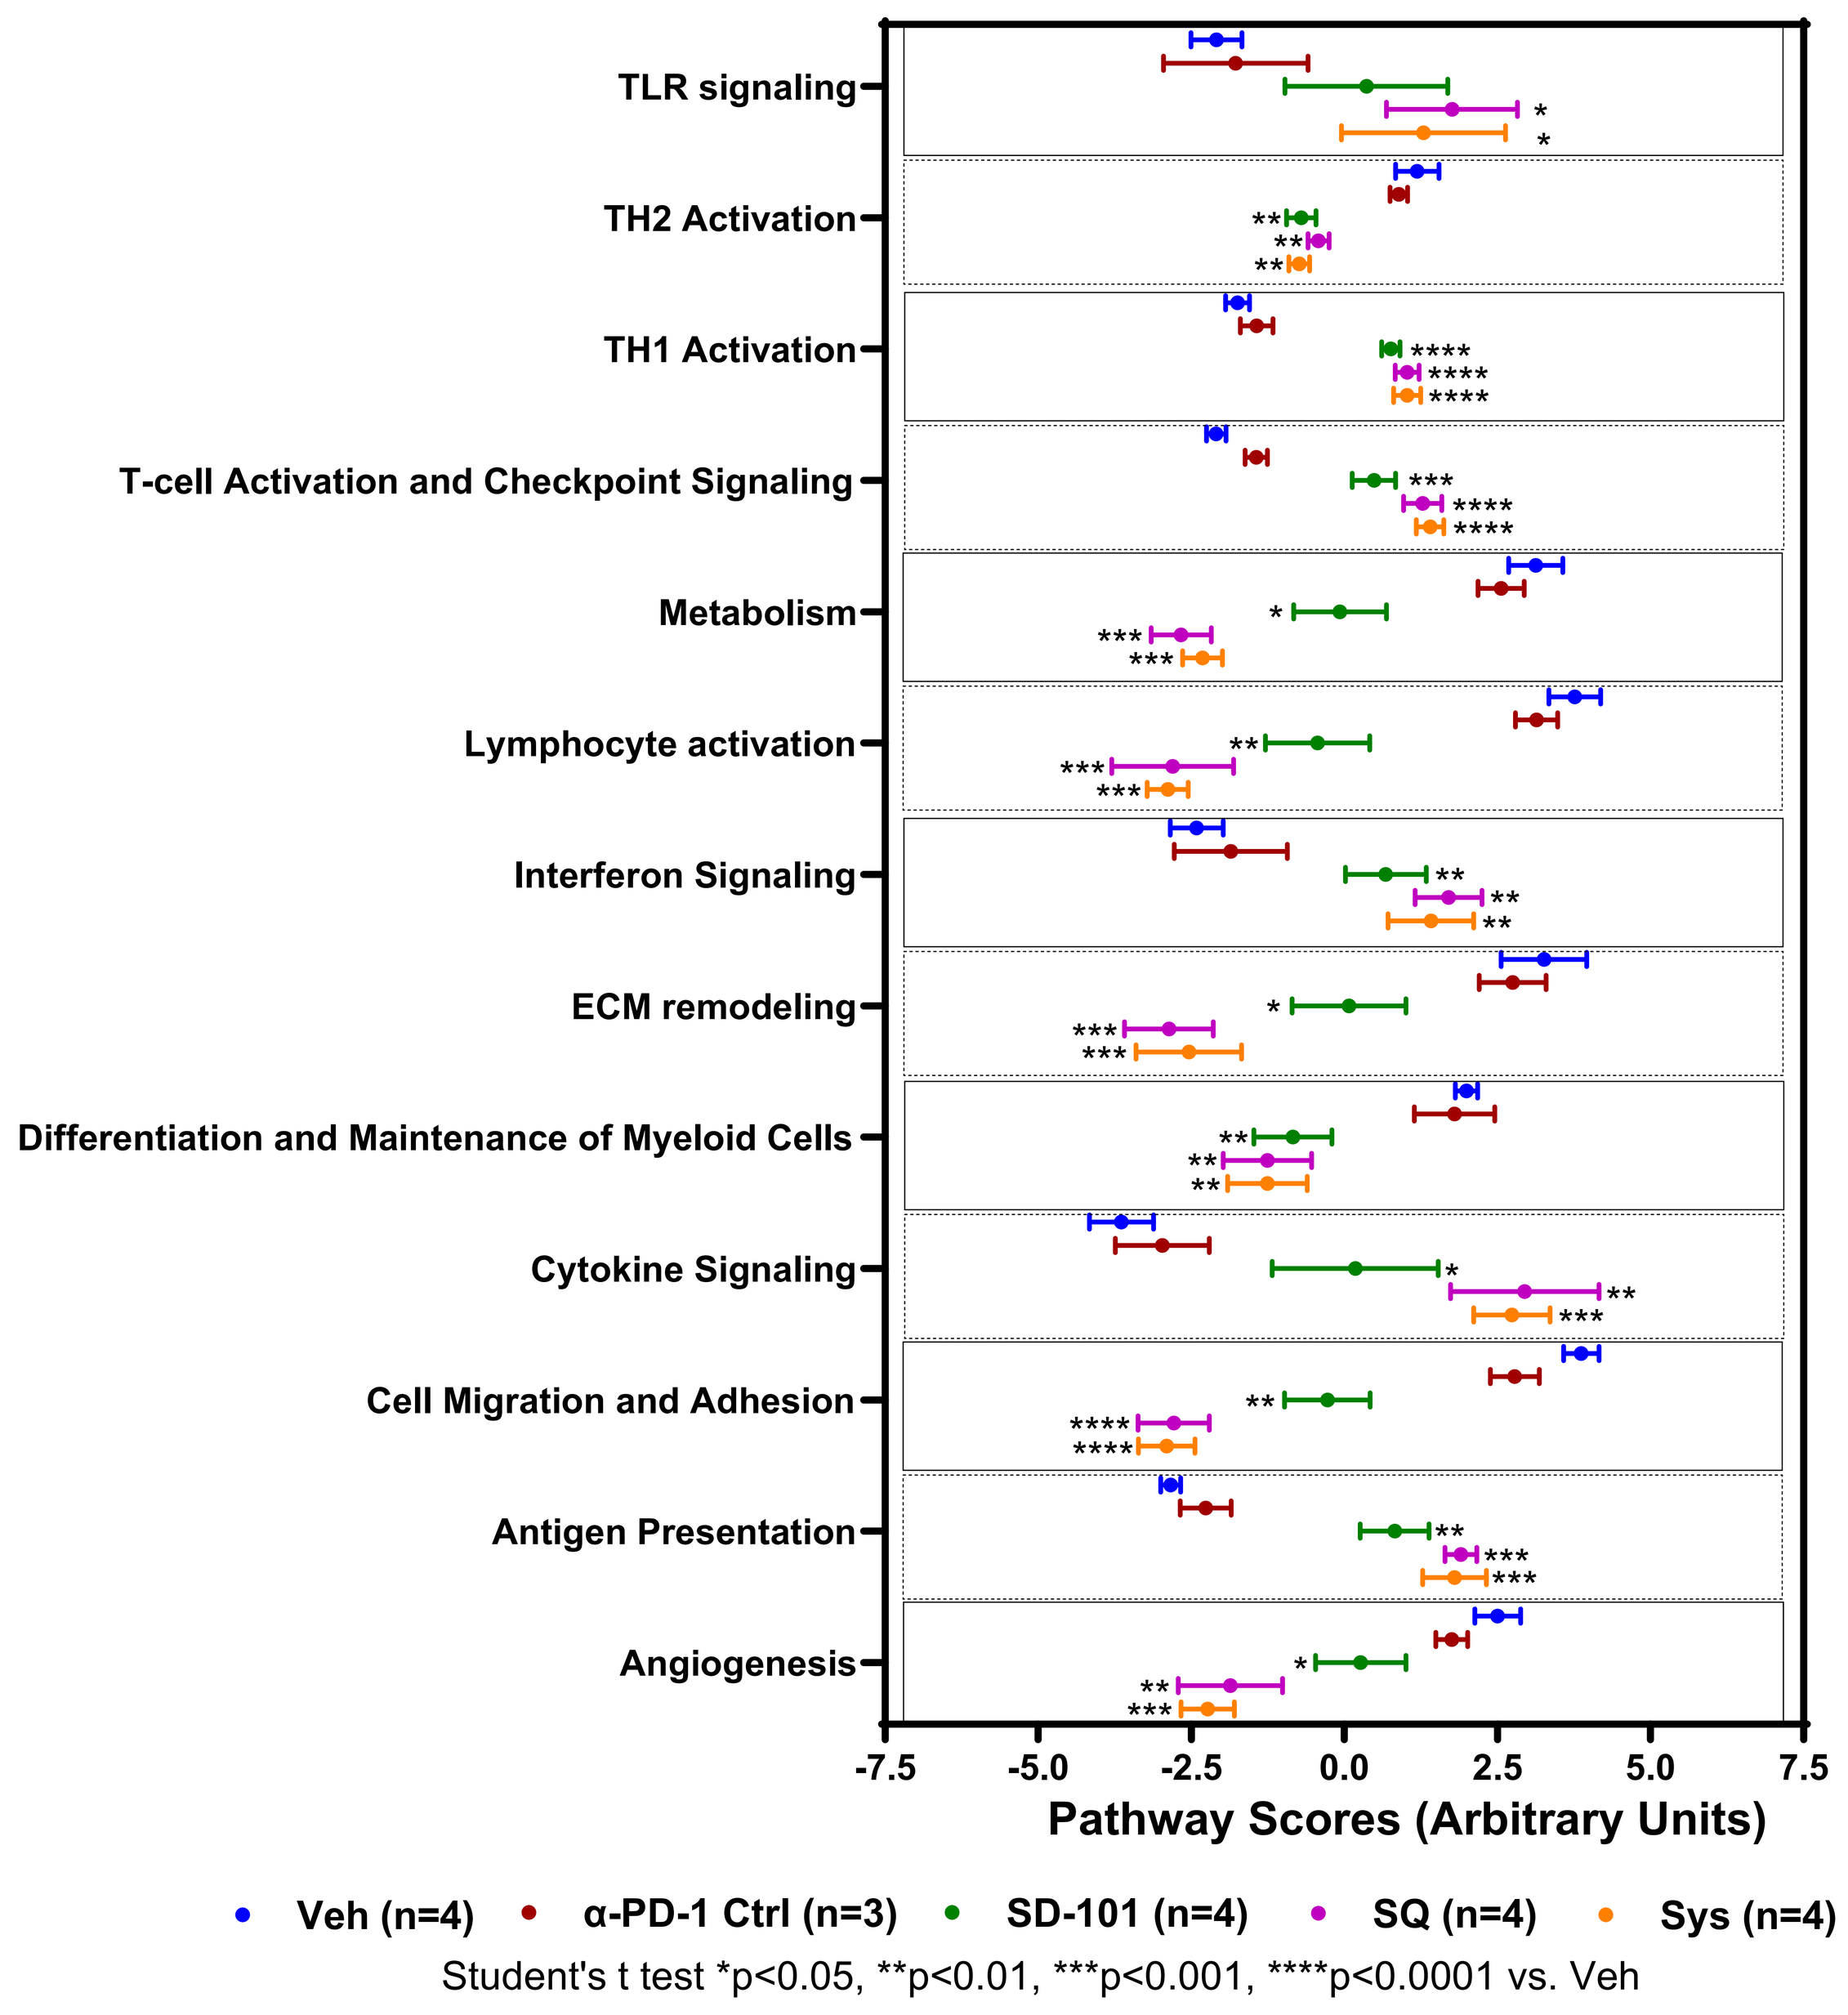

TLR signaling
TH2 Activation
TH1 Activation
T-cell Activation and Checkpoint Signaling
Metabolism
Lymphocyte activation
Interferon Signaling
ECM remodeling
Differentiation and Maintenance of Myeloid Cells
Cytokine Signaling
Cell Migration and Adhesion
Antigen Presentation
Angiogenesis
Pathway Scores (Arbitrary Units)
Veh (n=4)
α-PD-1 Ctrl (n=3)
SD-101 (n=4)
SQ (n=4)
Sys (n=4)
Student's t test *p<0.05, **p<0.01, ***p<0.001, ****p<0.0001 vs. Veh

B Expression of Ifnγ (i) and Granzyme (ii)

i. Ifnγ

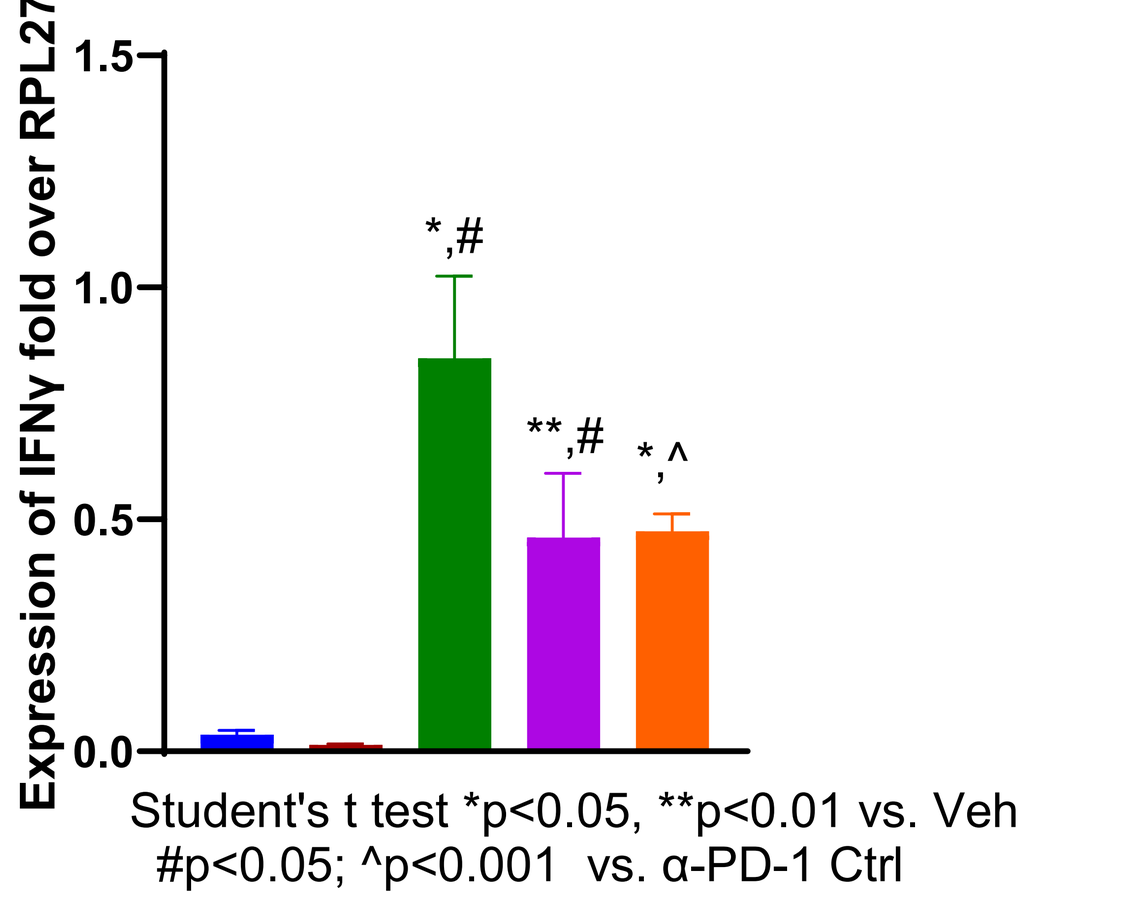

Expression of IFNγ fold over RPL27
Student's t test *p<0.05, **p<0.01 vs. Veh
#p<0.05; ^p<0.001 vs. α-PD-1 Ctrl

ii. Granzyme

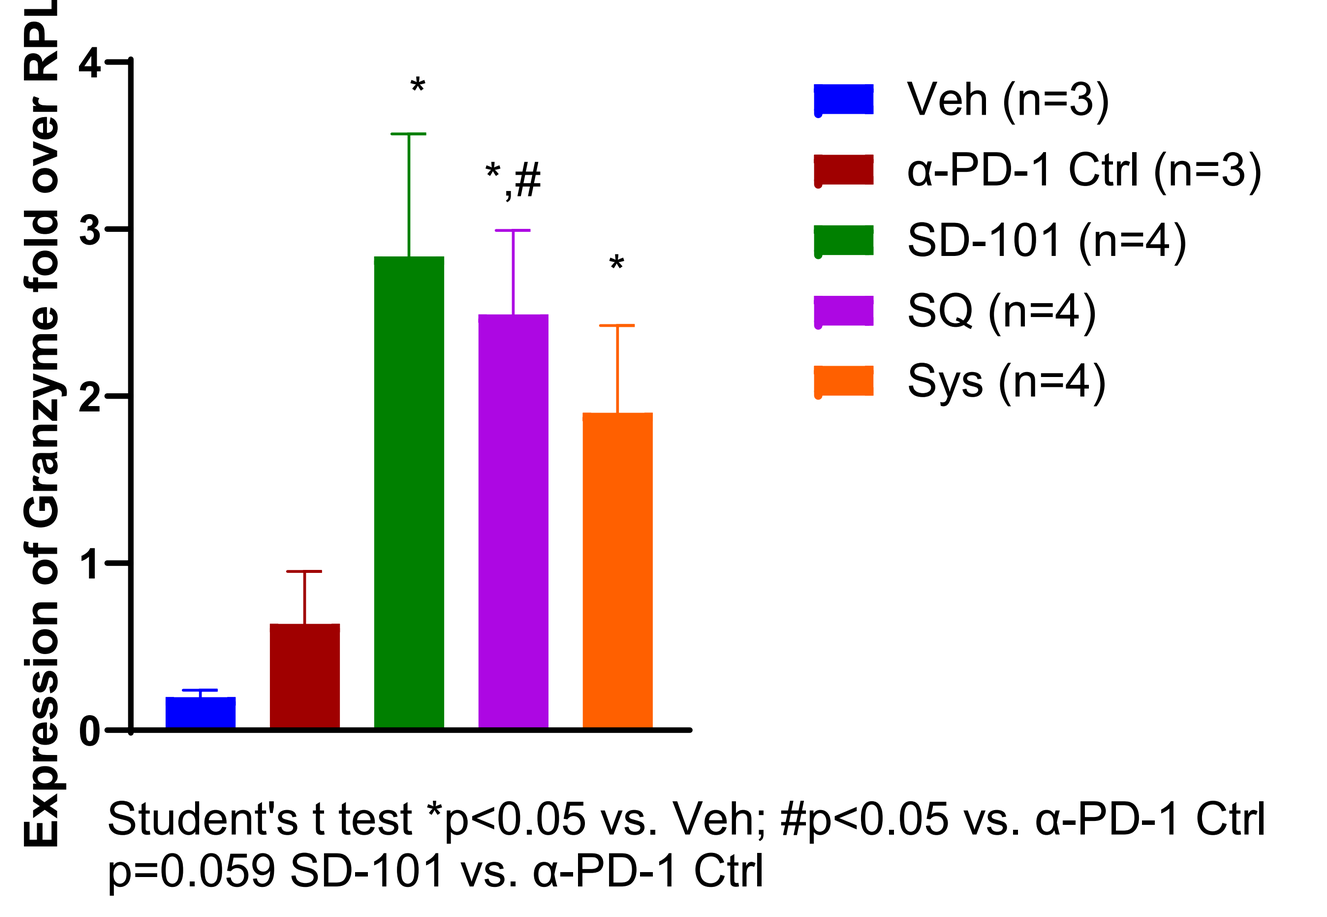

Expression of Granzyme fold over RPL27
Veh (n=3)
α-PD-1 Ctrl (n=3)
SD-101 (n=4)
SQ (n=4)
Sys (n=4)
Student's t test *p<0.05 vs. Veh; #p<0.05 vs. α-PD-1 Ctrl
p=0.059 SD-101 vs. α-PD-1 Ctrl

Figure 5: A. Total RNA was isolated from the CD45+ cells of LM and Nanostring analysis was performed using myeloid and innate panels. Pathways that were altered by SD-101, α-PD-1 monotherapy or combination therapy compared to Veh were evaluated by using nSolver advanced analysis module and plotted, respectively. **B.** Expression of Ifnγ (**i**) and Granzyme (**ii**) were quantified by qRT PCR. Data presented as mean ± SEM. Student's t-test was performed for group-wise comparison and are described in each graph.

Figure 6: PEDD-SD-101 in combination with α-PD-1 irrespective of the route of administration improved the overall survival

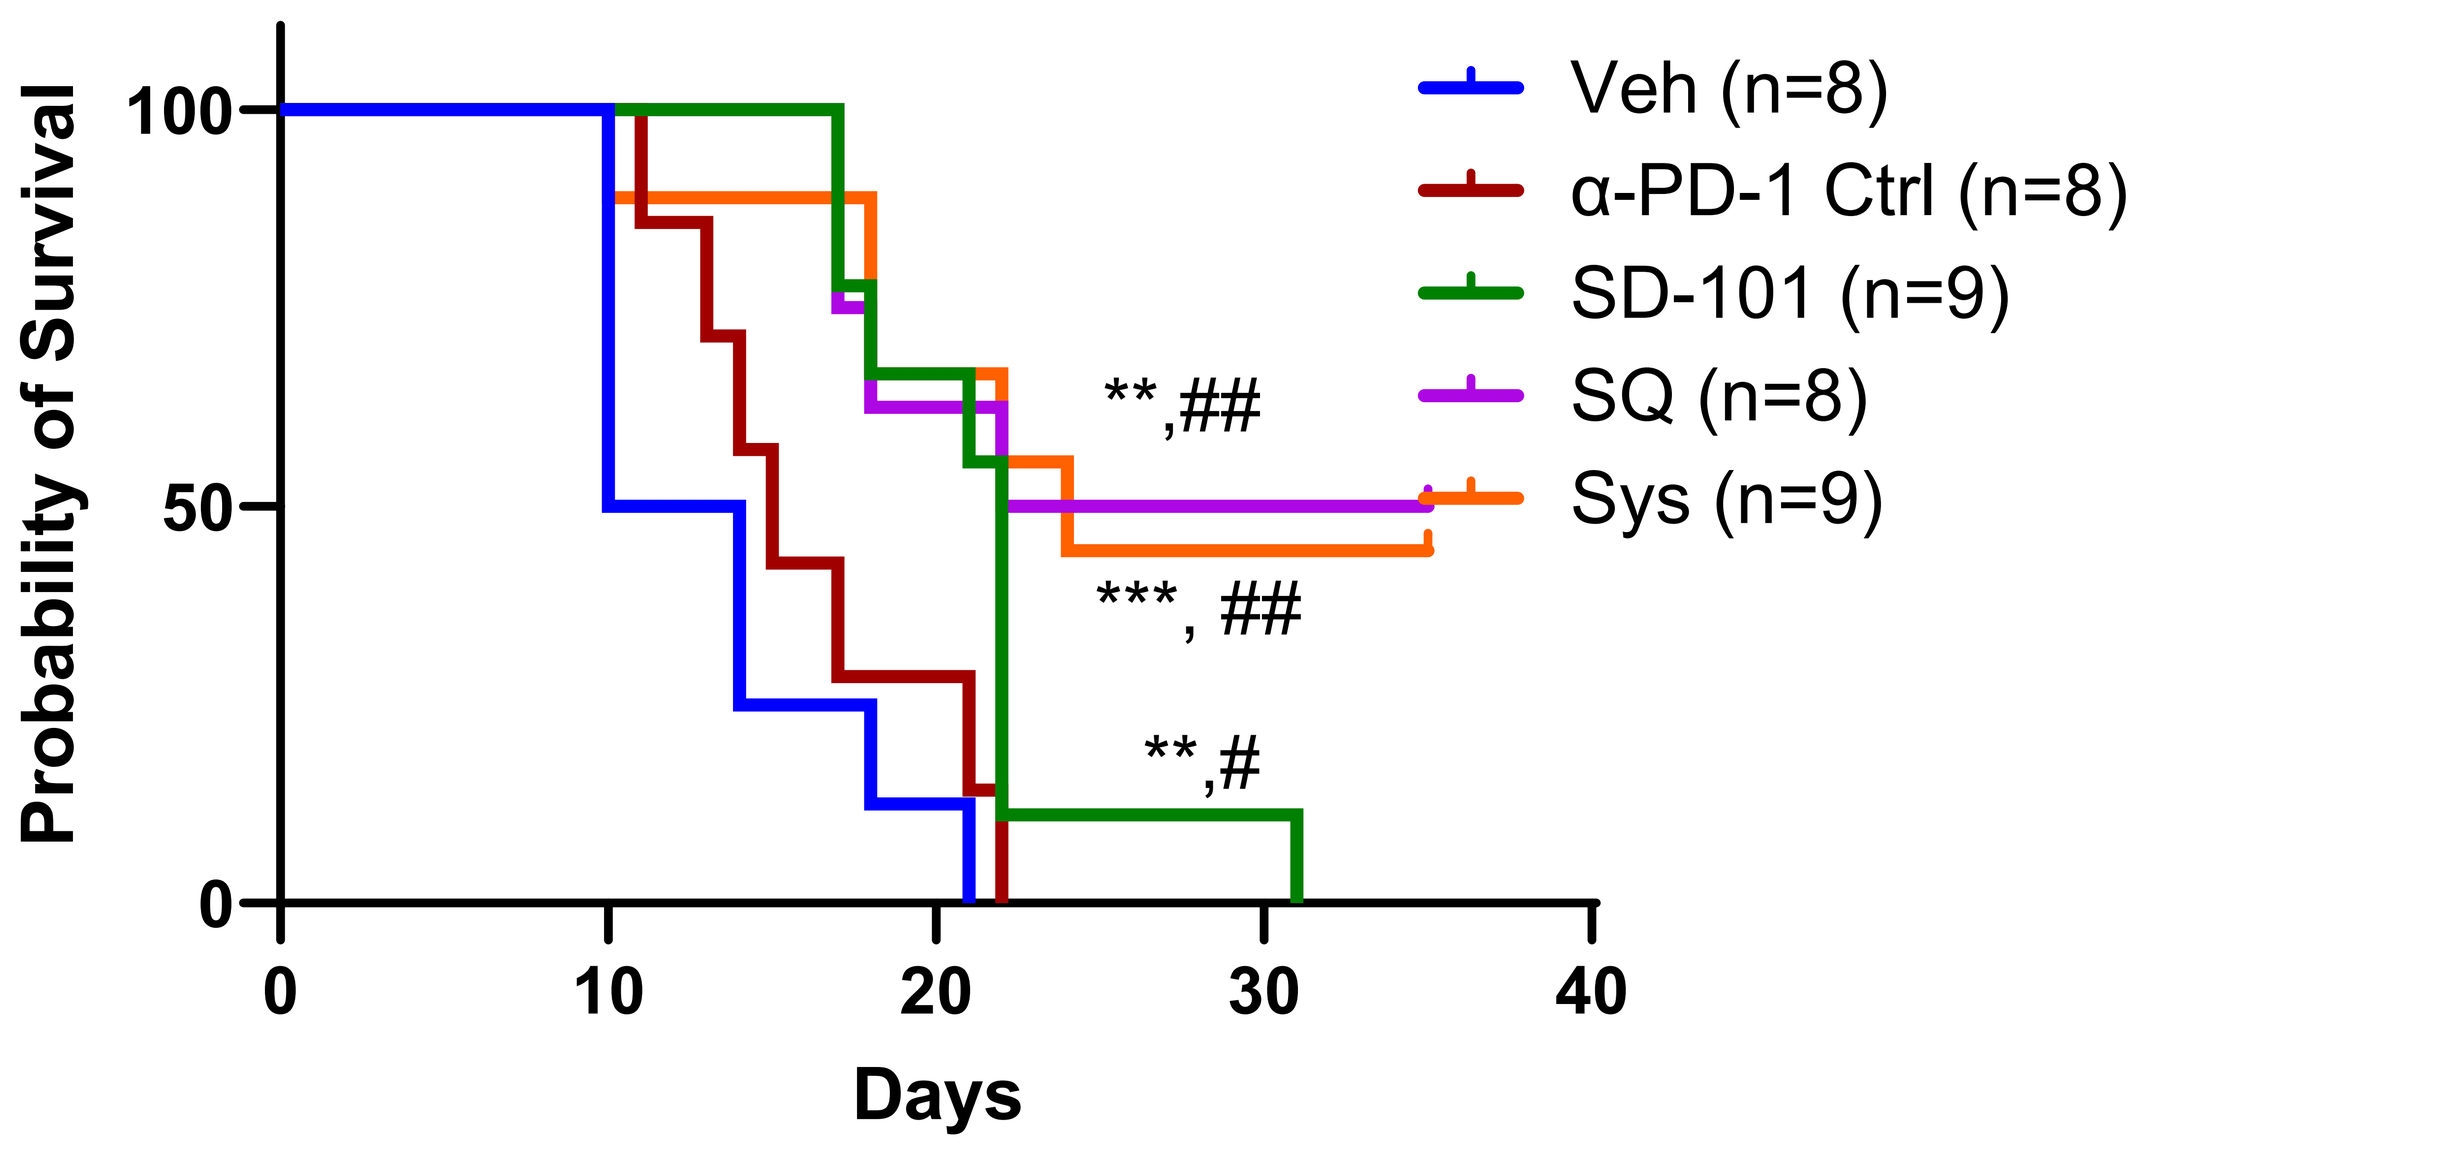

Probability of Survival
Days
Veh (n=8)
α-PD-1 Ctrl (n=8)
SD-101 (n=9)
SQ (n=8)
Sys (n=9)
**,##
***, ##
**,#

Log-Rank test **p<0.01, ***p<0.001 vs. Veh; #p<0.05, ##p<0.01 vs. α-PD-1 Ctrl
p=0.09 SD-101 vs. SQ and p=0.07 SD-101 vs. Sys

Figure 5: Kaplan-Meier survival curve for LM bearing mice treated with intravascular SD-101 with α-PD-1 administered either Sys or SQ.

Summary

- Tumor controlling effect of SD-101 delivered via PEDD was enhanced by α-PD-1 irrespective of the route of administration.
- Both the SQ and Sys resulted in superior outcomes in reducing frequencies of MDSCs, predominantly immunosuppressive M-MDSC subpopulation and enhanced B, T and dendritic cells within the liver tumor microenvironment (TME)
- SD-101 with/without α-PD-1 increased circulatory IFNγ and IP-10 leading to probable anti-tumor immunity
- In the liver TME, SD-101 monotherapy and in combination with α-PD-1 either via SQ or Sys, enhanced genes that inhibited extracellular matrix remodeling and promoted pro-inflammatory, anti-tumorigenic, anti-angiogenic effects, thereby driving pathways that promote anti-tumor immunity
- SD-101 as a monotherapy and in combination with α-PD-1 irrespective of the route of administration improved the survival of mice with aggressive LM

Reference

Ghosh CC *et al*, Regional infusion of a class C TLR9 agonist enhances liver tumor microenvironment reprogramming and MDSC reduction to improve responsiveness to systemic checkpoint inhibition. Cancer Gene Ther. 2022. PMID: 35697801.