Clinical activity of SD-101 with immune checkpoint inhibition (ICI) in metastatic uveal melanoma liver metastasis (MUM-LM) from the PERIO-01 Phase 1 trial

[1]Sapna P. Patel*, [2]Richard Carvajal, [3]Kamaneh Montazeri, [4]Jose Lutzky, [5]Bartosz Chmielowski, [6]Sunil Reddy, [7]Shailander Bhatia, [2]Shaheer Khan, [8]Theresa Medina, [1]Cara Haymaker, [1]Rahul Sheth, [1]Joshua D. Kuban, [4]Lindsay Thornton, [3]Eric Wehrenberg-Klee, [9]Paula Novelli, [1]Anthony Lucci, [1]Vanessa Sarli, [1]Salyna Meas, [10]Bryan Cox, [10]Jason LaPorte, [10]Prajna Guha, [10]Chandra Ghosh, [10]Ann-Marie Hulstine, [10]Robert Knight, [10]Ashley Moody, [9]David Geller, [11]Marlana Orloff, [10,12]Steven C. Katz, [9]Diwakar Davar

[1]MD Anderson Cancer Center, Houston; [2]Northwell Health Cancer Institute, New Hyde Park, NY; [3]Massachusetts General Hospital, Boston, MA; [4]University of Miami, Miami, FL; [5]University of California, Los Angeles, CA; [6]Stanford Cancer Center, Stanford, CA; [7]University of Washington, Seattle, WA; [8]University of Colorado, Denver, CO; [9]University of Pittsburgh Medical Center, Pittsburgh, PA, TX; [10]TriSalus Life Sciences, Westminster, CO; [11]Thomas Jefferson University, Philadelphia, PA; [12]Department of Surgery, Brown University, Providence, RI

* Presenting Author

Contact: SPPatel@mdanderson.org

Abstract #1534

Background

- MUM-LM are resistant to ICIs for several reasons including the prevalence of myeloid-derived suppressor cells (MDSCs)
- PFS has been limited, even with approved therapies such as tebentafusp (median 3.3 months) with grade 3/4 AE rates typically >30%
- TLR9 agonists are capable of MDSC polarization but drug delivery has historically been limited using an intra-tumoral approach
- Pressure-enabled drug delivery (PEDD™) of SD-101, a TLR9 agonist, has the potential to overcome these barriers to improve outcomes

Methods

- PERIO-01 is a phase 1 trial of hepatic arterial infusion of SD-101 via PEDD™ in MUM-LM (NCT04935229), with dose-escalation cohorts as monotherapy (Cohort A), with nivolumab (Cohort B), or nivolumab + ipilimumab (Cohort C)
- SD-101 is delivered over 2 outpatient cycles, with 3 weekly doses/cycle.
- Plasma was analyzed for SD-101 levels by LC-MS and cytokine levels by Luminex
- NanoString was used to analyze gene expression levels within PBMCs and LM. The nSolver advanced analysis was performed for pathway scoring.
- FFPE tissue sections were analyzed by multiplex IF using the Akoya Bioscience PhenoCycler-Fusion system staining for DAPI, CD14, HLA-DR, CD163, CD68, CD11b, CD8, CD15, CD56, CD3e, CD4, CD20, FOXP3, gp100, & CD45. Quantitative cell density data was generated using QuPath 0.4.3
- Flow cytometry was performed to evaluate $CD8^+$ T cell proliferation ($CD45^+CD3^+CD8^+Ki67^+$), activation ($CD45^+CD3^+CD8^+ICOS^+$) and NK cell proliferation ($CD45^+CD3^-CD56^+Ki67^+$), using CytoFlex

Figure 1. Schema

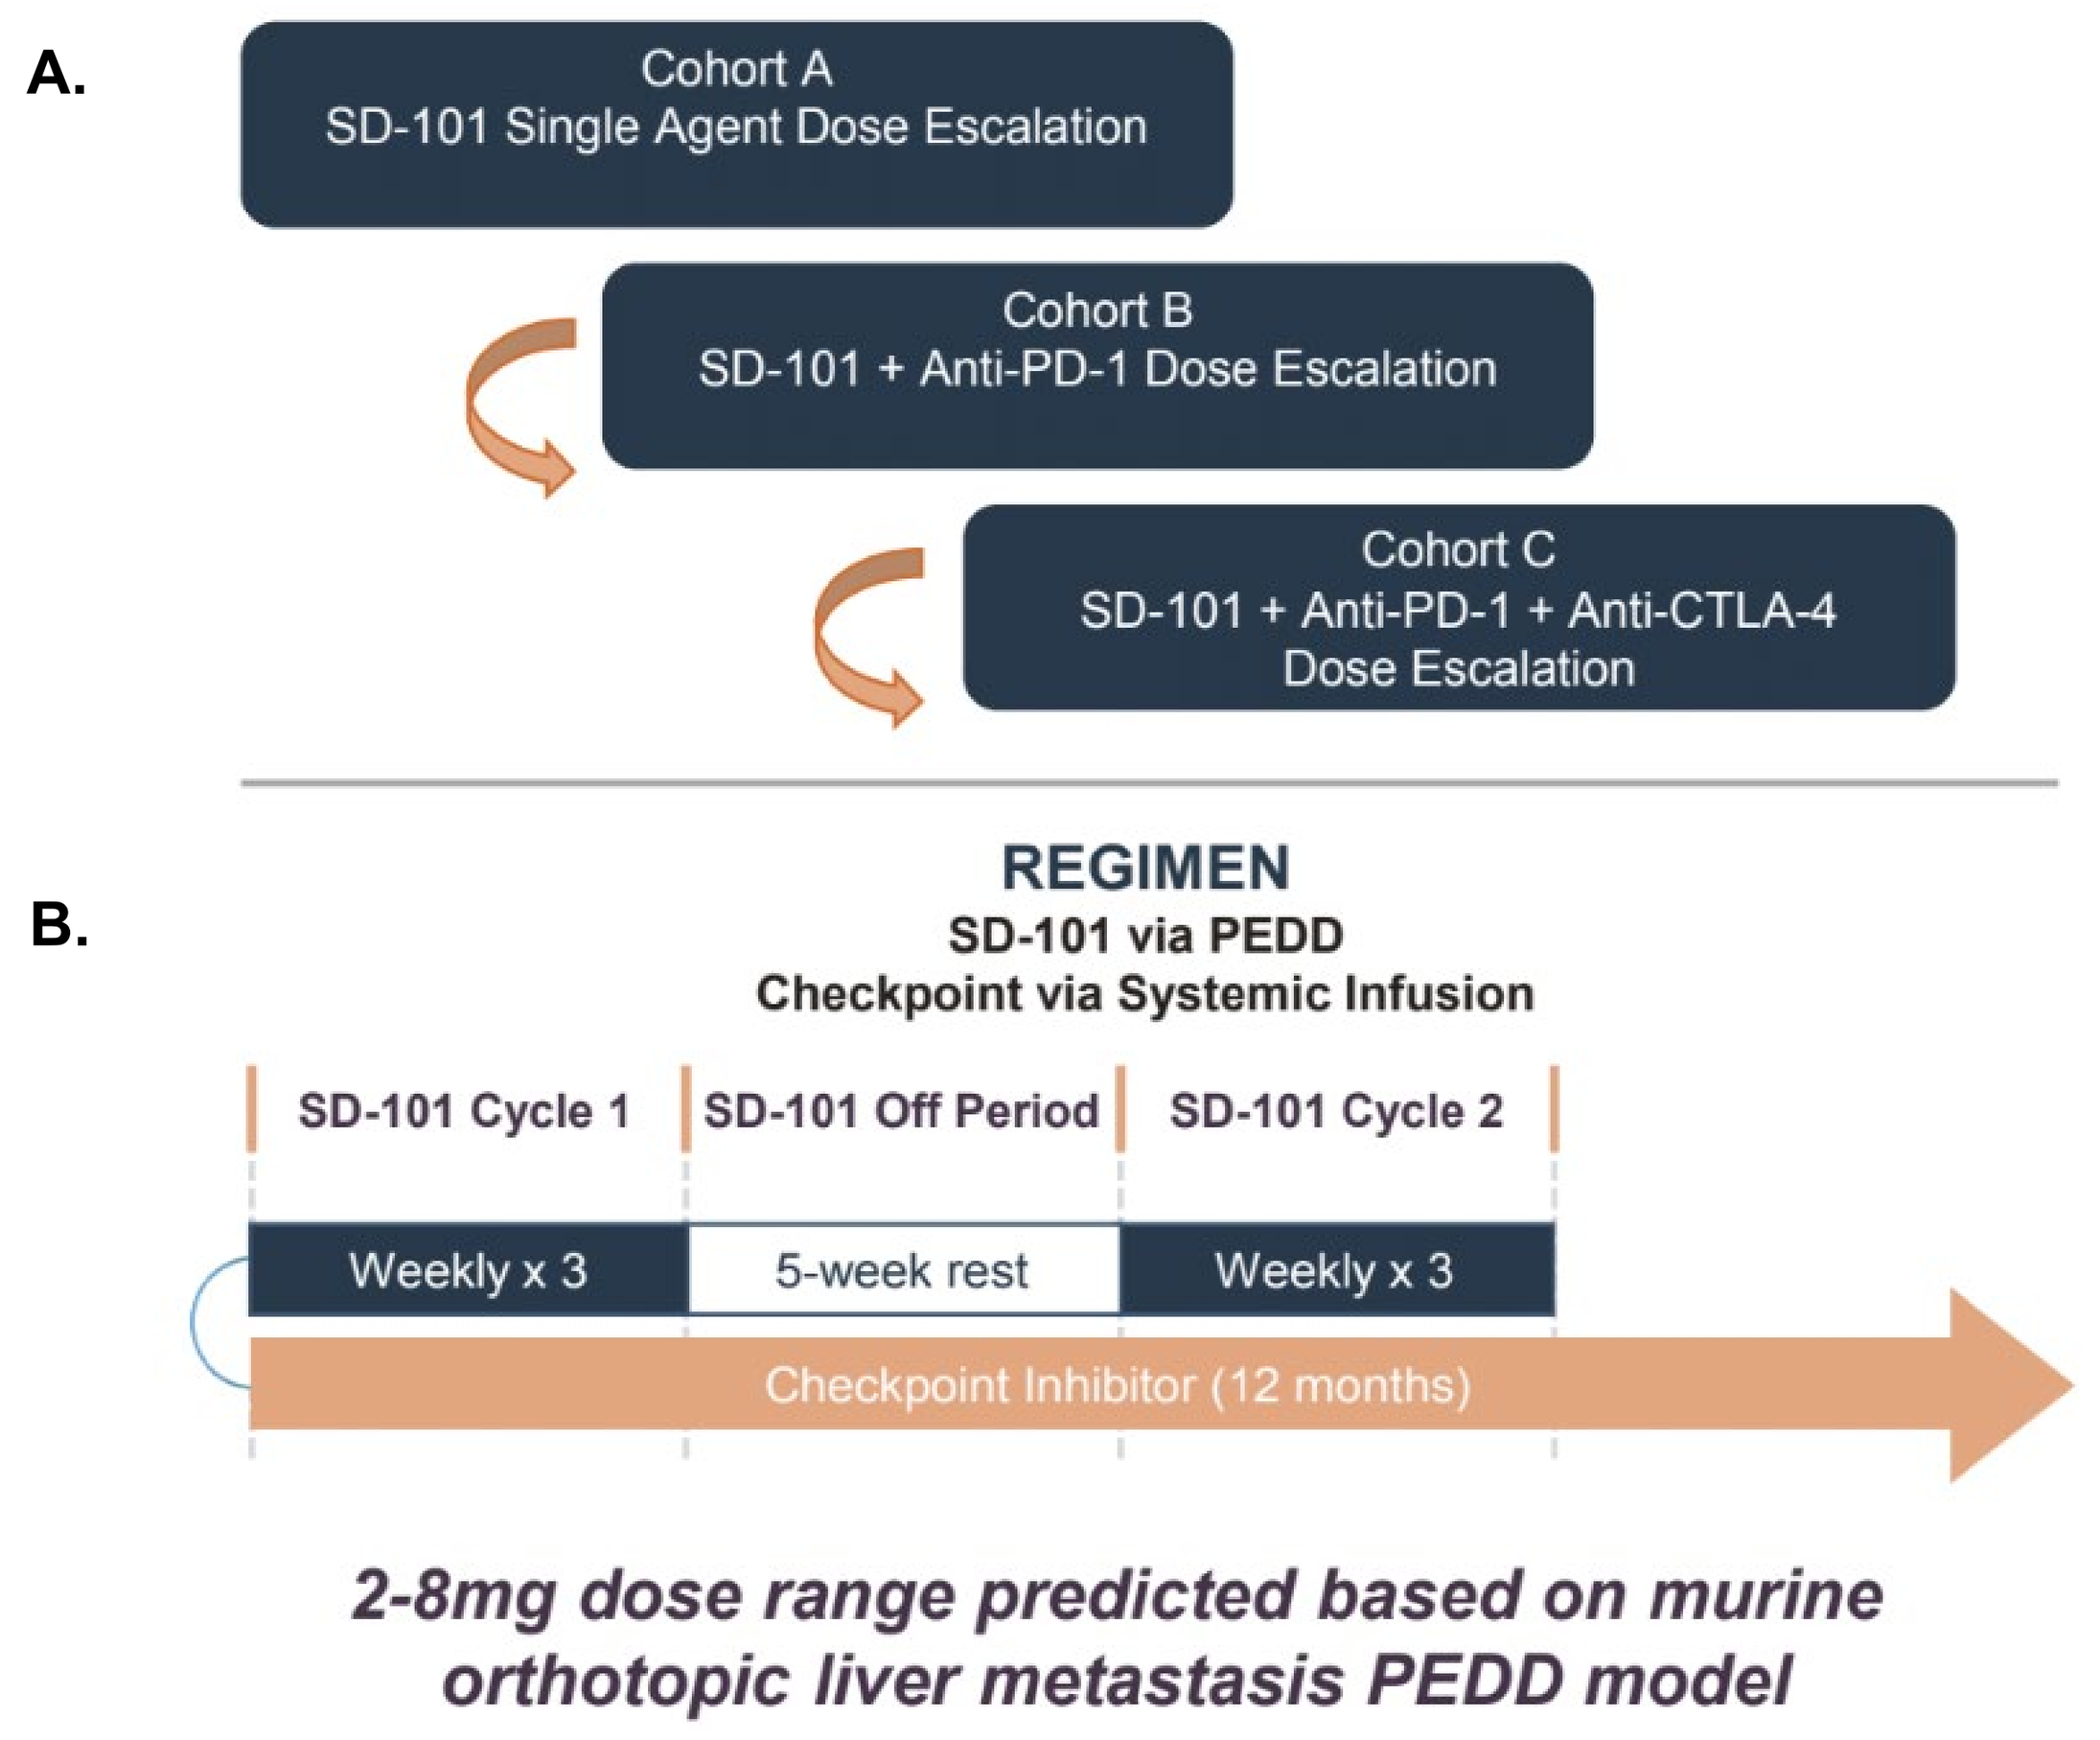
A.
Cohort A
SD-101 Single Agent Dose Escalation
Cohort B
SD-101 + Anti-PD-1 Dose Escalation
Cohort C
SD-101 + Anti-PD-1 + Anti-CTLA-4
Dose Escalation
B.
REGIMEN
SD-101 via PEDD
Checkpoint via Systemic Infusion
SD-101 Cycle 1
SD-101 Off Period
SD-101 Cycle 2
Weekly x 3
5-week rest
Weekly x 3
Checkpoint Inhibitor (12 months)
2-8mg dose range predicted based on murine orthotopic liver metastasis PEDD model

Figure 1. (**A**) Overall study design and (**B**) Treatment regimen.

Table 1. Patient Characteristics

Patient Characteristics	N=56 (%)
Age (median, range)	64 (35-86)
Gender	
Female	26 (46)
Male	30 (54)
LDH	
>ULN	27 (48)
Normal	27 (48)
N/A	2 (4)
Performance Status	
0	45 (80)
1	11 (20)
# prior lines therapy	
0	16 (29)
1	17 (30)
2	12 (21)
≥3	8 (14)
N/A	3 (5)
Prior tebentafusp	9 (16)

Results

Figure 2. PEDD of SD-101 via HAI reduced ctDNA

Specified time points (A.): 27% ctDNA clearance and 65% response rate
Best response on treatment (B.): 59% ctDNA clearance and 86% response rate

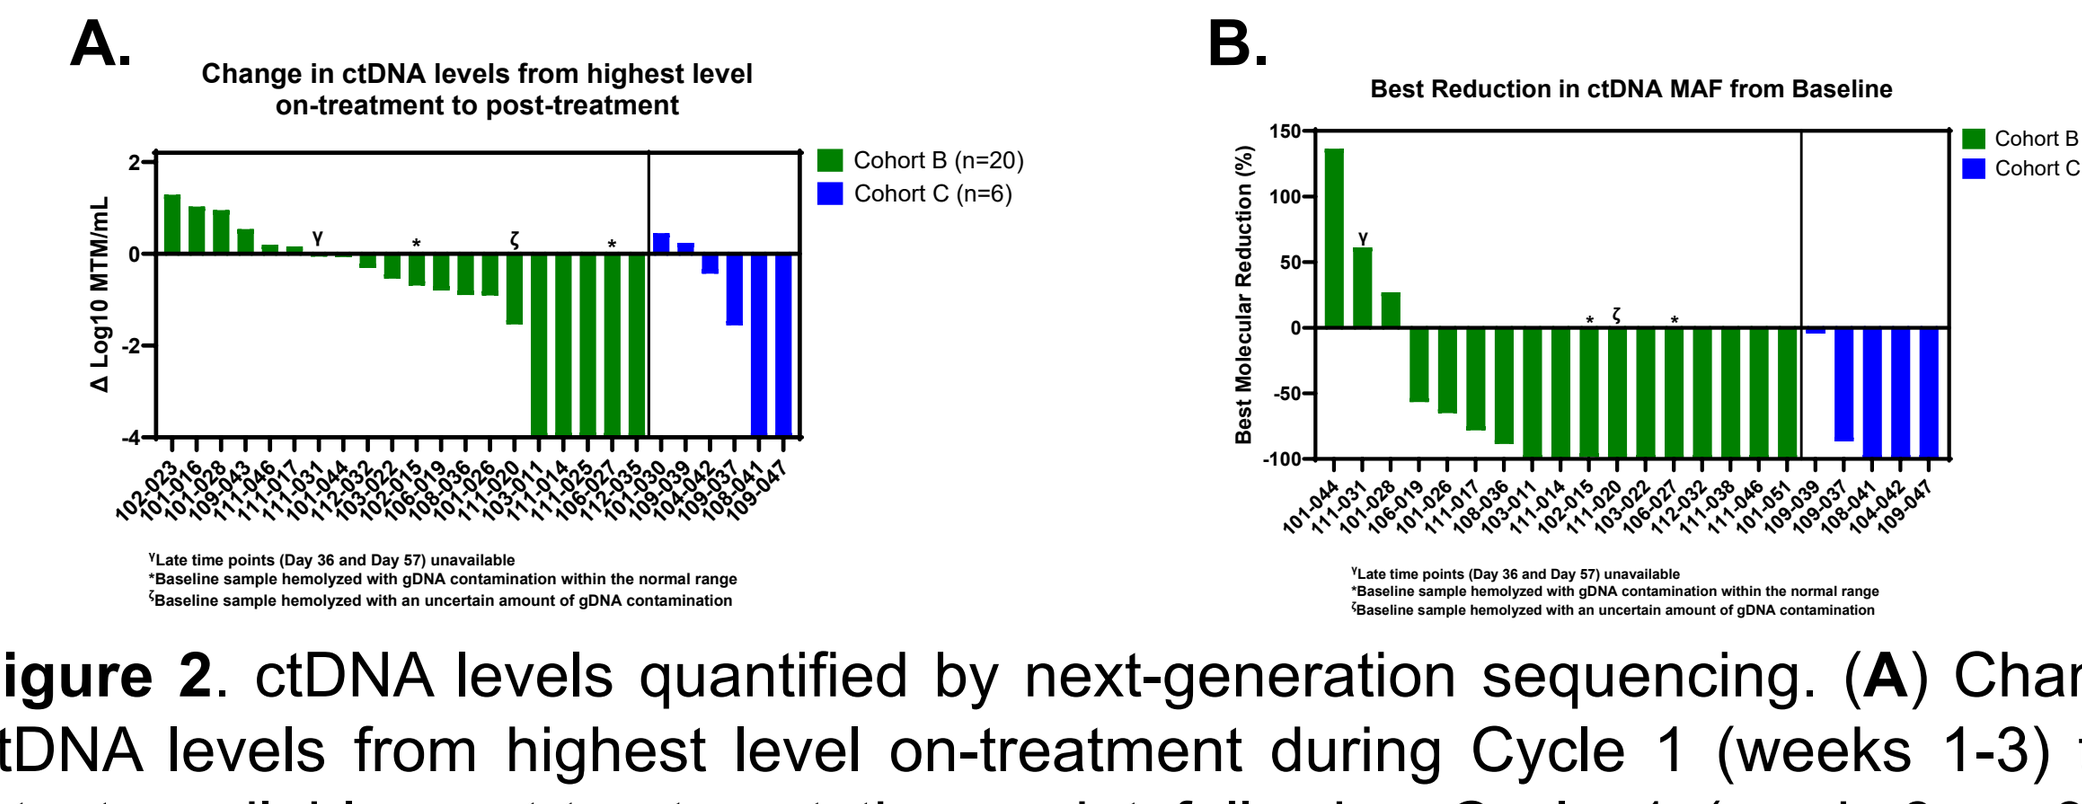
A.
Change in ctDNA levels from highest level on-treatment to post-treatment
Cohort B (n=20)
Cohort C (n=6)
Δ Log10 MTM/mL
B.
Best Reduction in ctDNA MAF from Baseline
Cohort B (n=17)
Cohort C (n=5)
Best Molecular Reduction (%)

Figure 2. ctDNA levels quantified by next-generation sequencing. (**A**) Change in ctDNA levels from highest level on-treatment during Cycle 1 (weeks 1-3) to the latest available post-treatment time point following Cycle 1 (week 6 or 9). (**B**) Change in ctDNA levels from baseline to any time point with the lowest ctDNA level.

Figure 3. Overall Survival Kaplan-Meier Curves

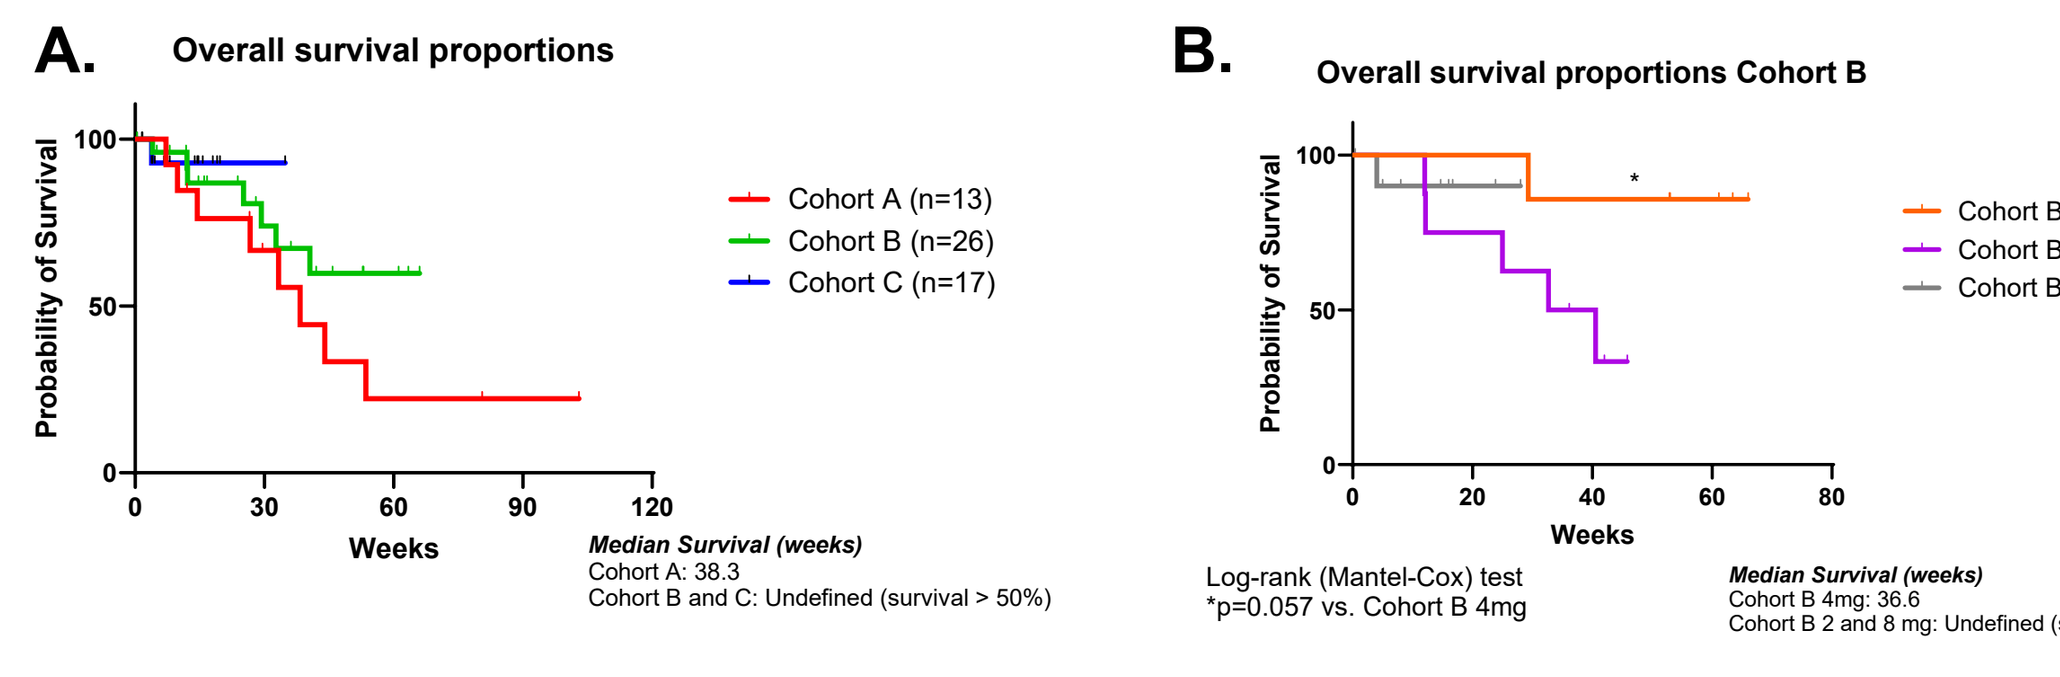
A.
Overall survival proportions
Cohort A (n=13)
Cohort B (n=26)
Cohort C (n=17)
Probability of Survival
Weeks
Median Survival (weeks)
Cohort A: 38.3
Cohort B and C: Undefined (survival > 50%)
B.
Overall survival proportions Cohort B
Cohort B 2mg (n=7)
Cohort B 4mg (n=8)
Cohort B 8mg (n=11)
Log-rank (Mantel-Cox) test
*p=0.057 vs. Cohort B 4mg
Median Survival (weeks)
Cohort B 4mg: 36.6
Cohort B 2 and 8 mg: Undefined (survival > 50%)

Figure 3. Kaplan–Meier curves showing overall survival for patients in different Cohorts (**A**) and in Cohort B (**B**).

Summary

- Regional delivery of SD-101 by PEDD results in high drug levels within the liver and transient serum exposure
- Median PFS of 11.7 months in patients receiving 2 mg + nivolumab is promising (median OS not reached)
- Increased $CD8^+$ T cell, NK cell and M1 macrophage infiltration in the tumors
- Decreased immunosuppressive Tregs, M-MDSCs and M2 macrophages in liver metastases
- Gene expression analysis in LM and in PBMCs revealed increased signals related to $CD8^+$ cytotoxic T lymphocyte activity, Th1 activation, cytokine and chemokine signaling
- Systemic immune activation observed by an increase in pro-inflammatory cytokines
- Systemic increase in NK cell proliferation and $CD8^+$ T cell activation

Conclusions

Delivery of SD-101 by PEDD plus systemic ICI in MUM-LM patients results in clinical activity with median PFS of 11.7 months, ctDNA molecular responses, MDSC re-programming, and evidence of peripheral and intra-tumoral immune activation.

Table 2. Safety Summary

*All listed events are related to SD-101, ipi, and/or nivo	Cohort A Summary n (%) N=13	Cohort A 2mg SD-101 n (%) N=4	Cohort A 4mg SD-101 n (%) N=5	Cohort A 8mg SD-101 n (%) N=4	Cohort B Summary n (%) N=26	Cohort B 2mg SD-101 + Nivo n (%) N=7	Cohort B 4mg SD-101 + Nivo n (%) N=8	Cohort B 8mg SD-101 + Nivo n (%) N=11	Cohort C Summary n (%) N=17	Cohort C 2mg SD-101 + Ipi/Nivo n (%) N=10	Cohort C 4mg SD-101 + Ipi/Nivo n (%) N=7	All cohorts n (%) N=56
Treatment-related AEs (any grade)	5 (38)	0	3 (60)	2 (50)	19 (73)	5 (71)	6 (75)	8 (73)	15 (88)	10 (100)	5 (71)	39 (70)
DLTs	0	0	0	0	2 (8)	1 (14)	0	1 (9)	0	0	0	2 (4)
SAEs	1 (8)	0	0	1 (25)	1 (4)	0	1 (13)	0	6 (35)	4 (40)	2 (29)	8 (14)
≥G3 events (%)	1 (8)	0	1 (20)	0	5 (19)	1 (14)	2 (25)	2 (18)	8 (47)	5 (50)	3 (43)	14 (25)
Most common AEs												
-Gastrointestinal Disorders	3 (23)	0	2 (40)	1 (25)	8 (31)	2 (29)	3 (38)	3 (27)	12 (71)	8 (80)	4 (57)	23 (41)
-Fatigue	2 (15)	0	2 (40)	0	5 (19)	2 (29)	2 (25)	1 (9)	10 (59)	7 (70)	3 (43)	17 (30)
-Skin Disorders	0	0	0	0	7 (27)	3 (43)	1 (13)	3 (27)	8 (47)	6 (60)	2 (29)	15 (27)
-Fever	2 (15)	0	1 (20)	1 (25)	1 (4)	0	0	1 (9)	6 (35)	3 (30)	3 (43)	9 (16)
-Administration Site Conditions	1 (8)	0	0	1 (25)	4 (15)	2 (29)	0	2 (18)	4 (24)	3 (30)	1 (14)	9 (16)
-Increased ALT	0	0	0	0	4 (15)	1 (14)	2 (25)	1 (9)	4 (24)	2 (20)	2 (29)	8 (14)
-Increased AST	0	0	0	0	4 (15)	1 (14)	2 (25)	1 (9)	4 (24)	2 (20)	2 (29)	8 (14)

Figure 4. Swimmer Plot of patients

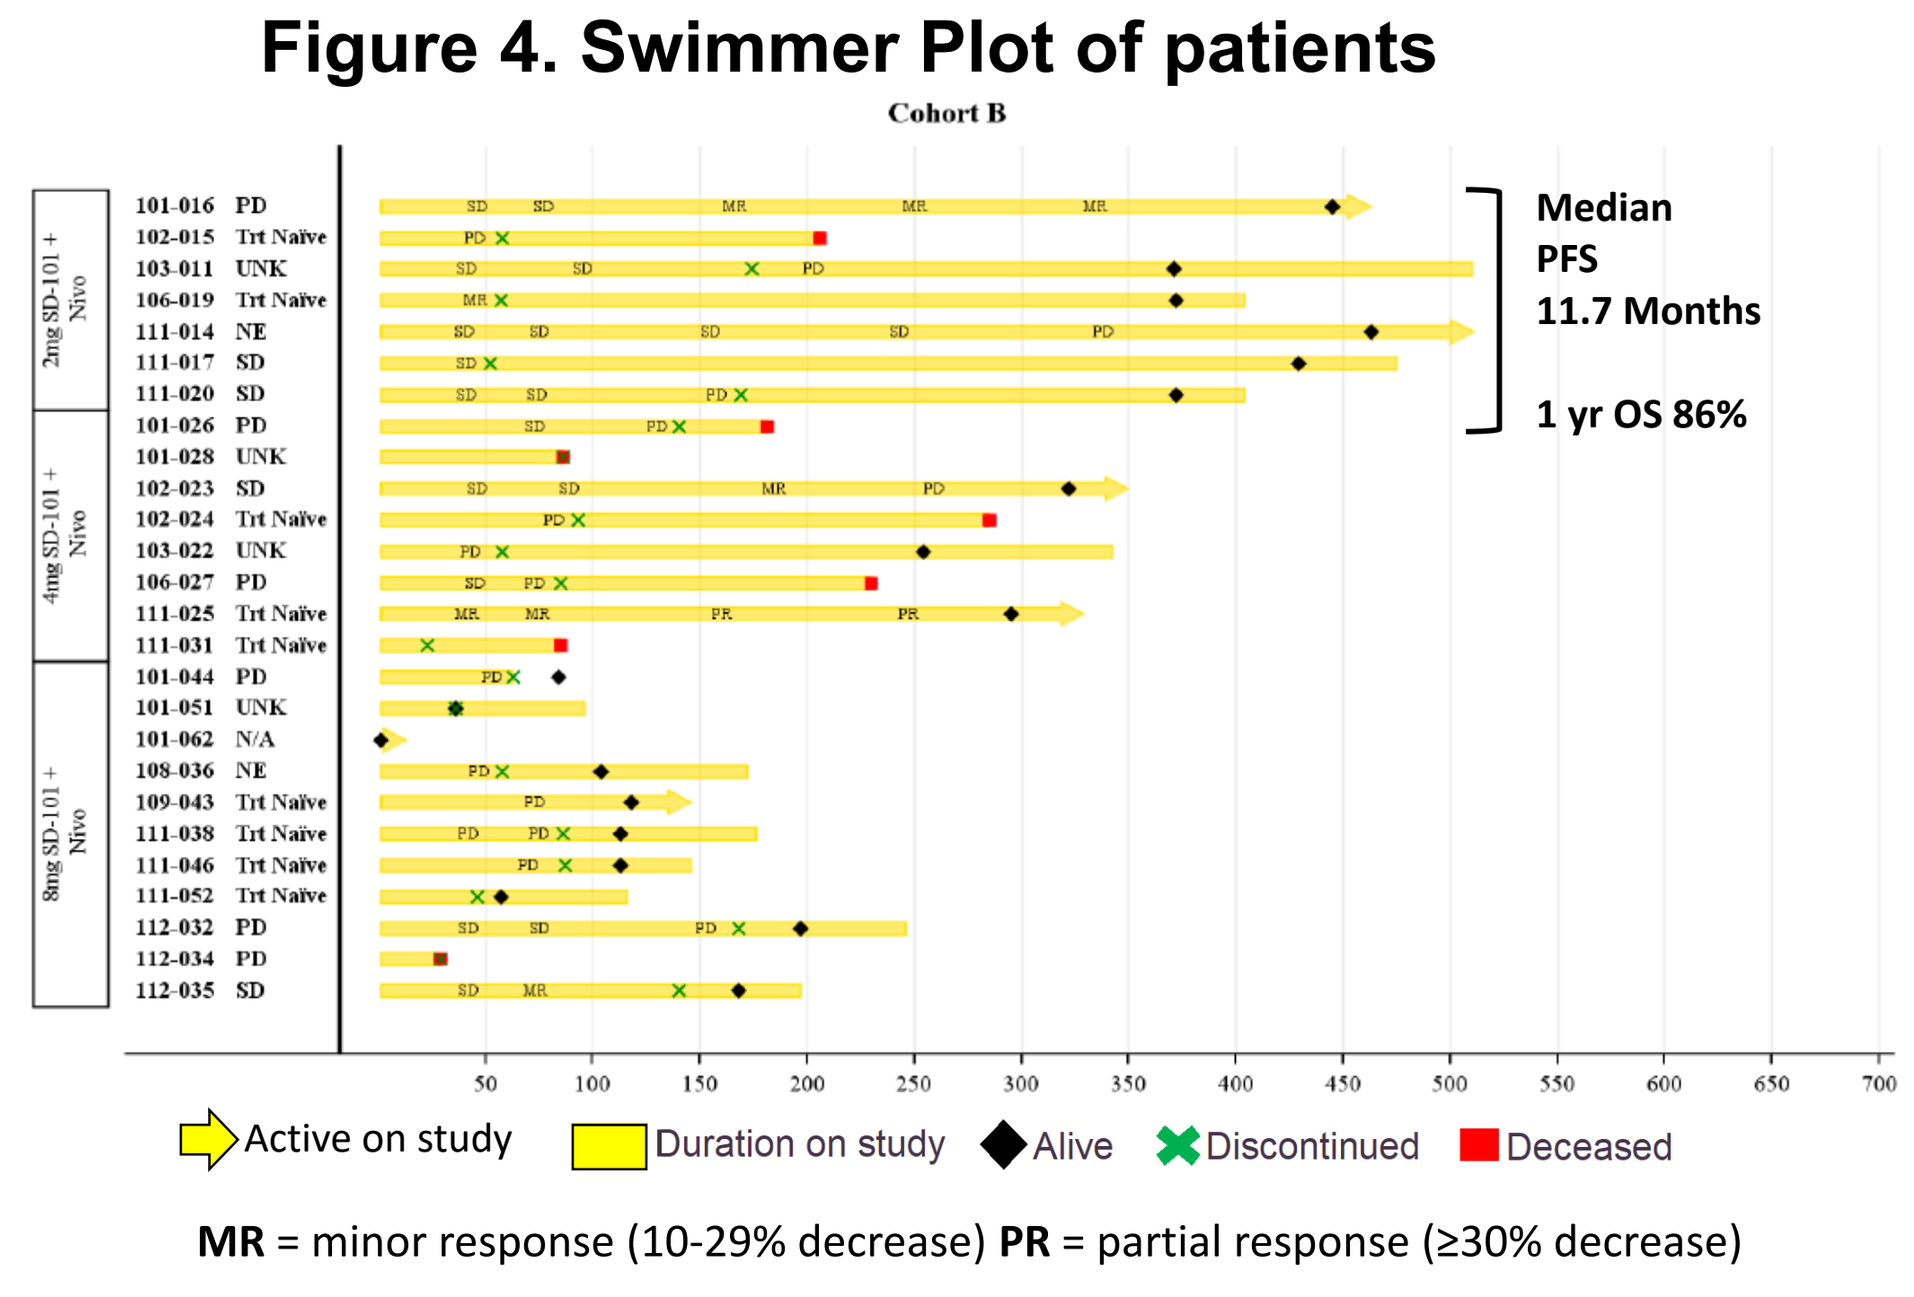
Cohort B
Median PFS 11.7 Months
1 yr OS 86%
Active on study
Duration on study
Alive
Discontinued
Deceased

MR = minor response (10-29% decrease) **PR** = partial response (≥30% decrease)

Objective RECIST 1.1 response rates

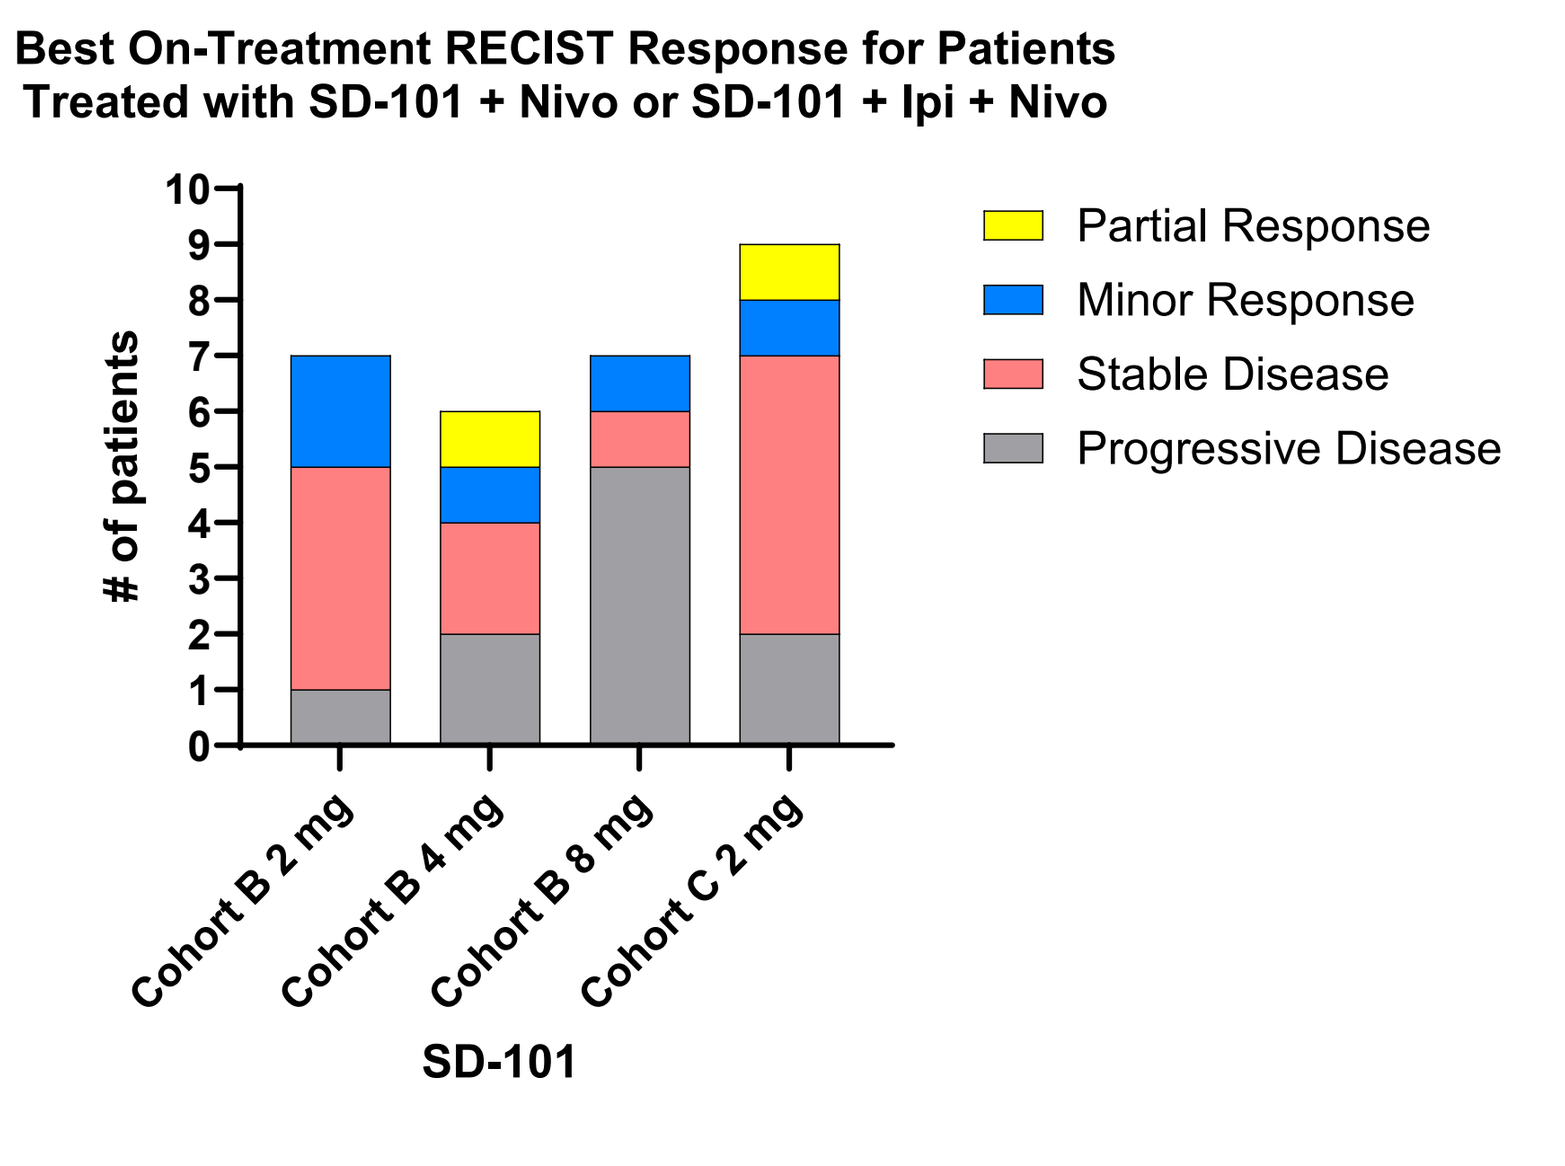
Best On-Treatment RECIST Response for Patients Treated with SD-101 + Nivo or SD-101 + Ipi + Nivo
Partial Response
Minor Response
Stable Disease
Progressive Disease
of patients
Cohort B 2 mg
Cohort B 4 mg
Cohort B 8 mg
Cohort C 2 mg
SD-101

Figure 5. Sequential MRI/CT scans

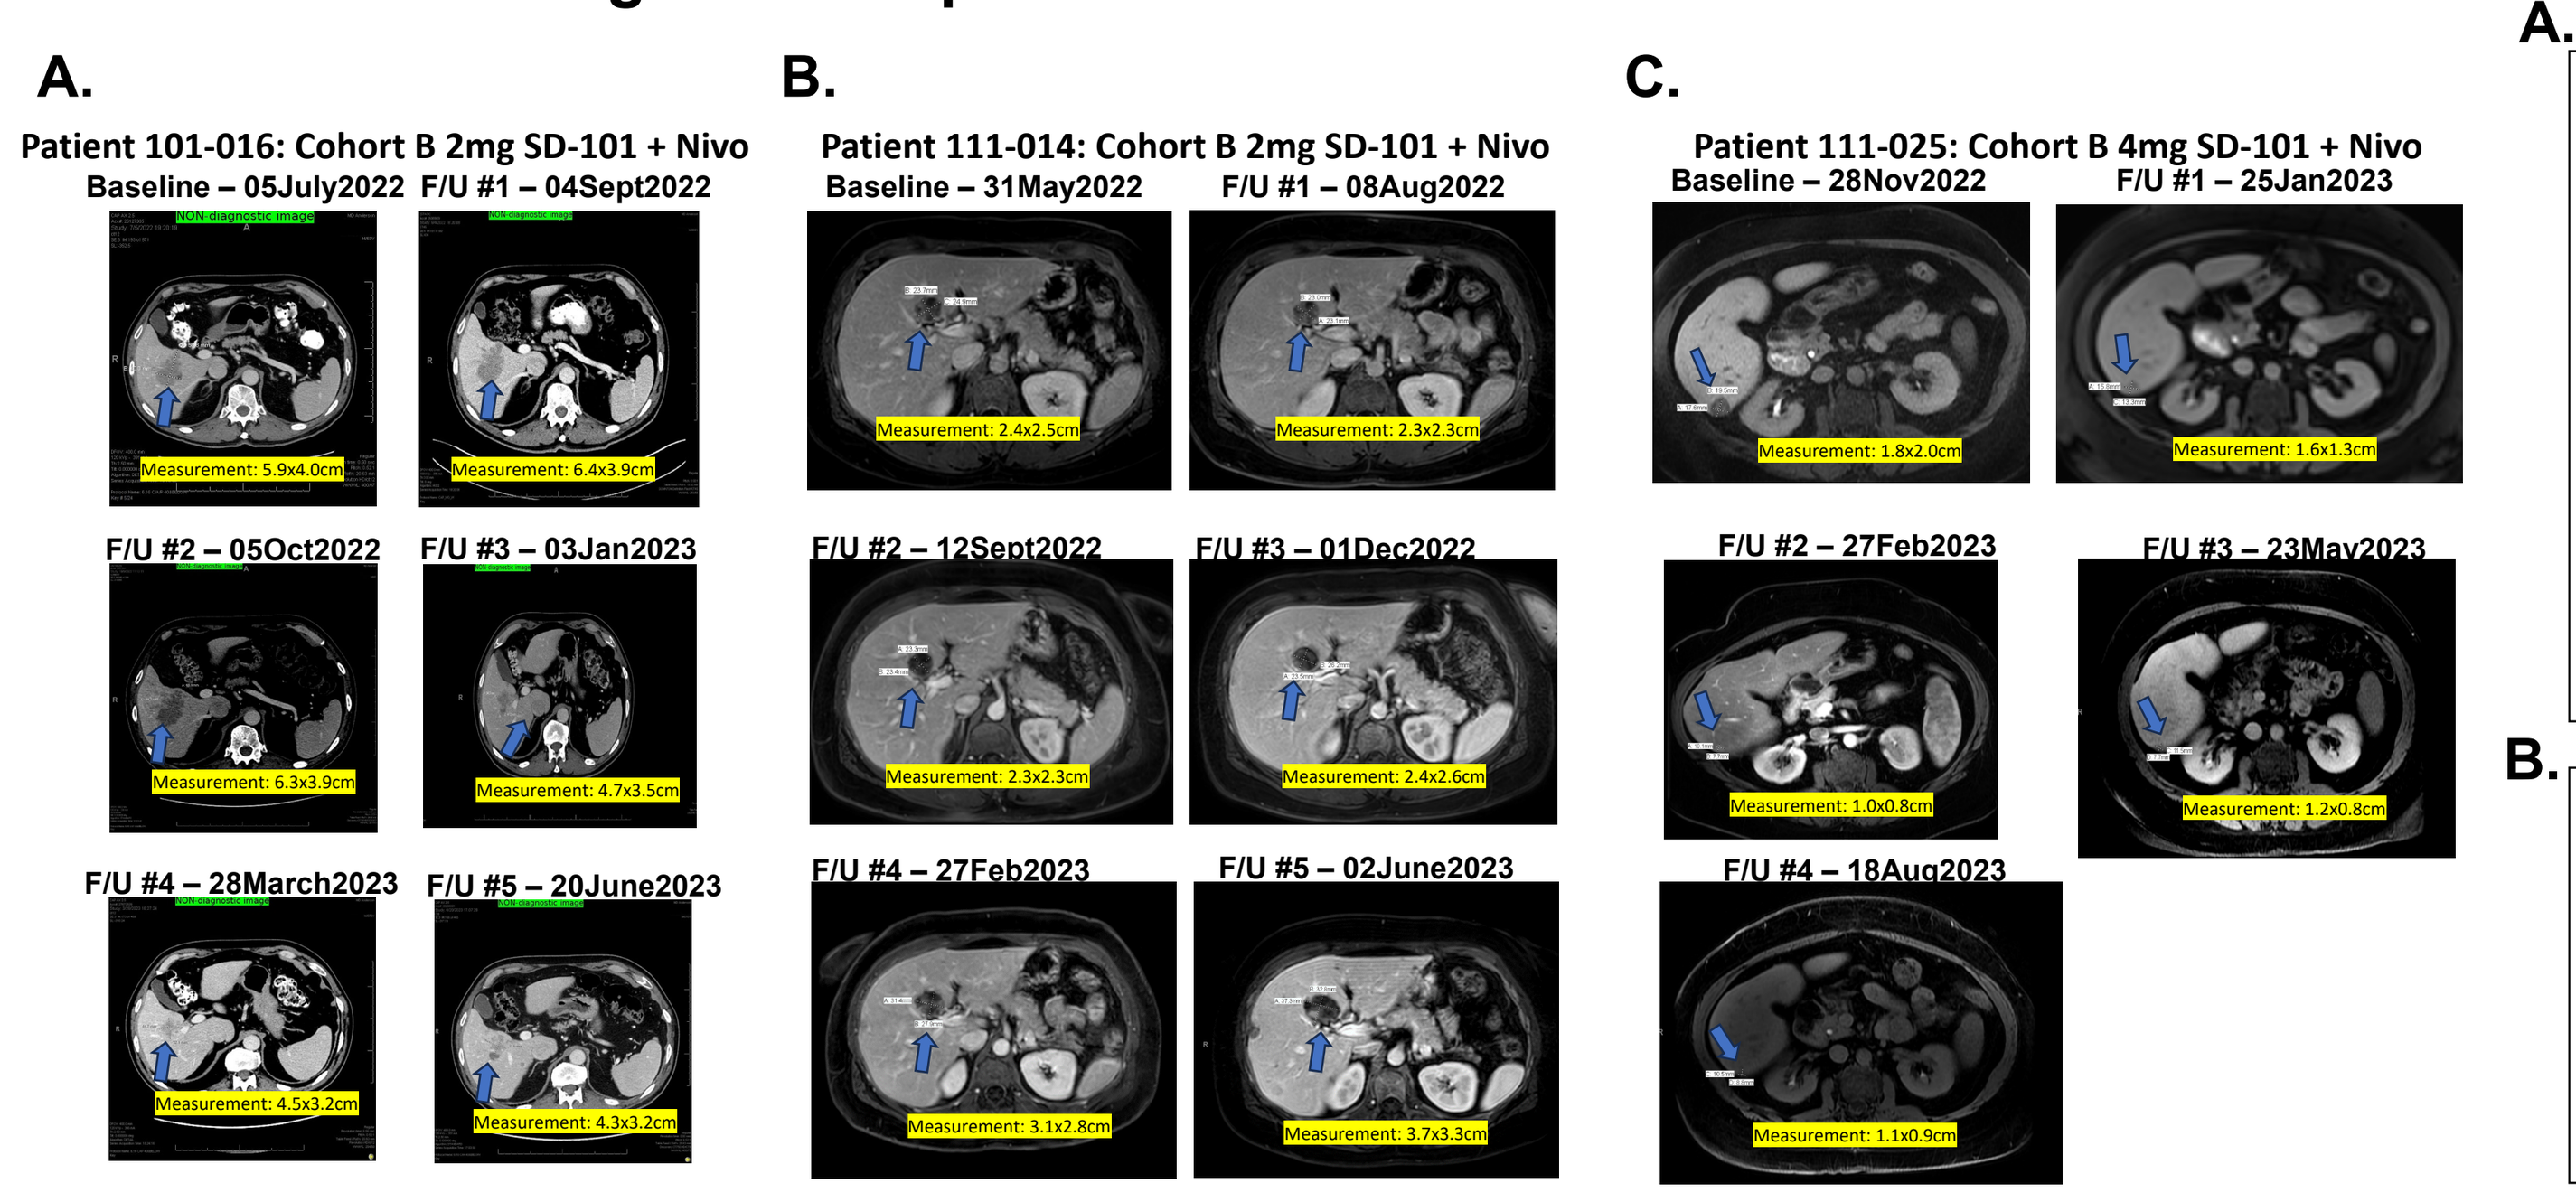
A.
Patient 101-016: Cohort B 2mg SD-101 + Nivo
Baseline – 05July2022
F/U #1 – 04Sept2022
F/U #2 – 05Oct2022
F/U #3 – 03Jan2023
F/U #4 – 28March2023
F/U #5 – 20June2023
B.
Patient 111-014: Cohort B 2mg SD-101 + Nivo
Baseline – 31May2022
F/U #1 – 08Aug2022
F/U #2 – 12Sept2022
F/U #3 – 01Dec2022
F/U #4 – 27Feb2023
F/U #5 – 02June2023
C.
Patient 111-025: Cohort B 4mg SD-101 + Nivo
Baseline – 28Nov2022
F/U #1 – 25Jan2023
F/U #2 – 27Feb2023
F/U #3 – 23May2023
F/U #4 – 18Aug2023

Figure 6. Plasma levels of SD-101 are transient and remain low following PEDD

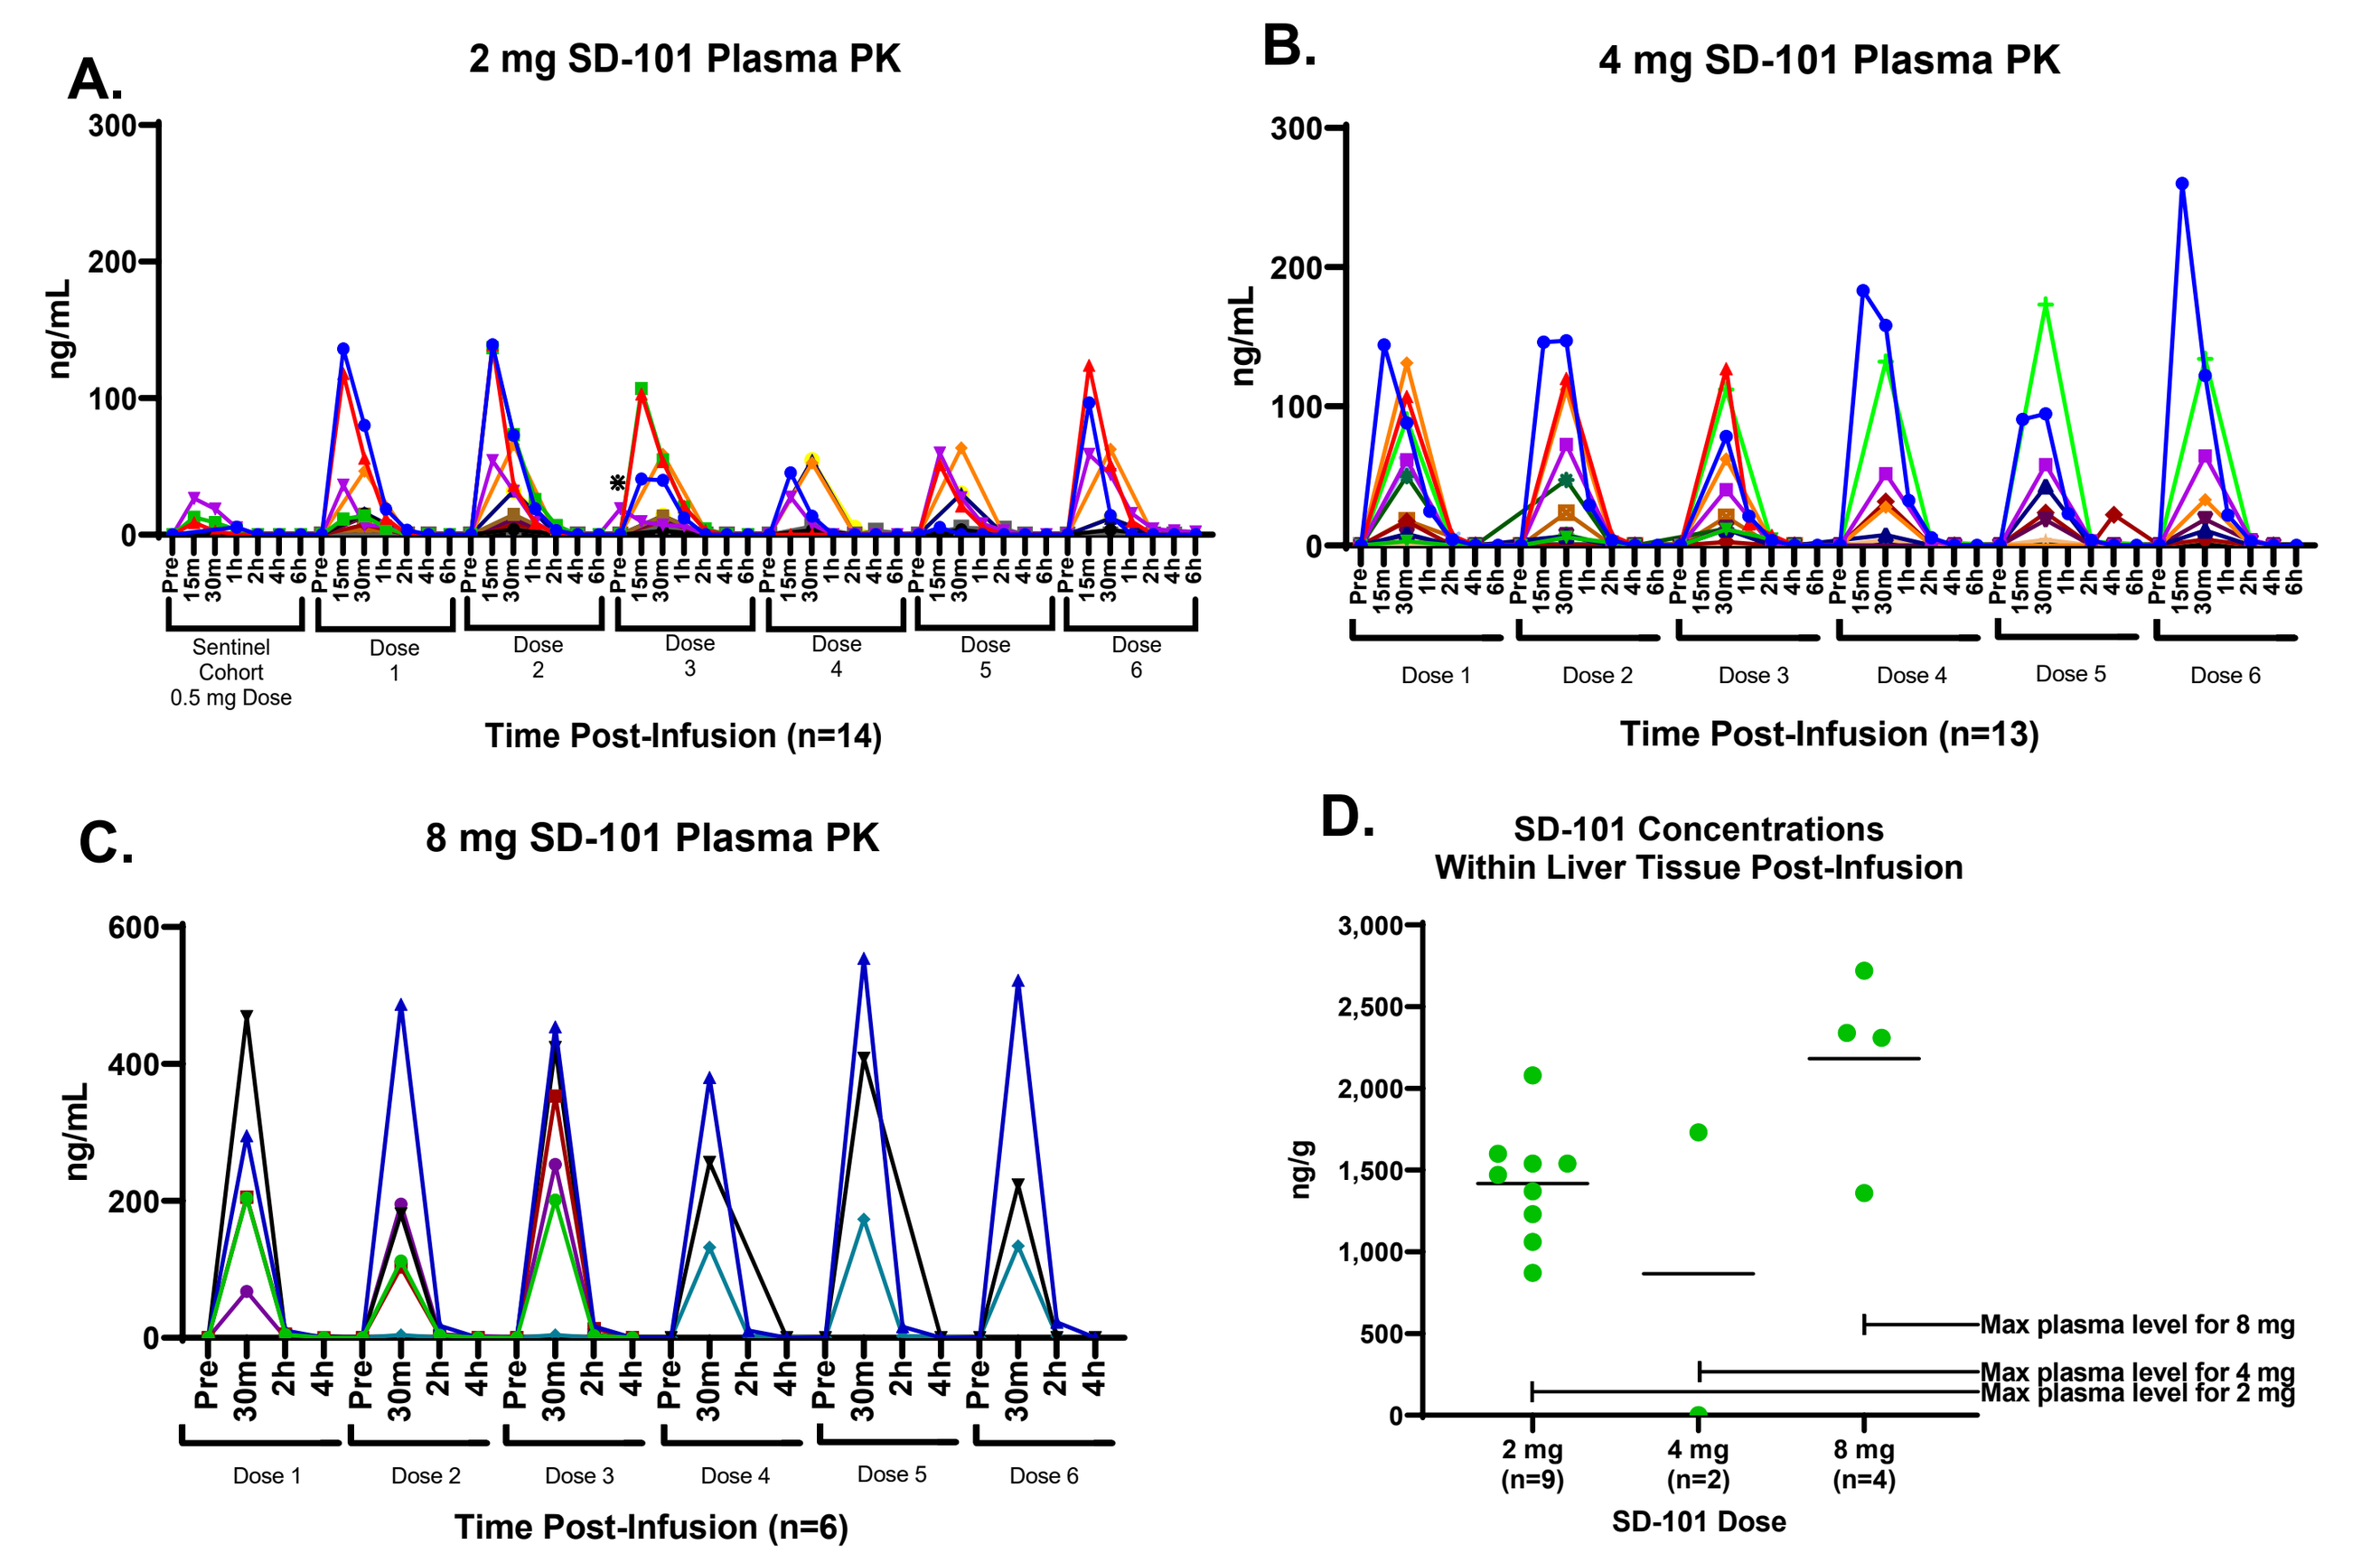
A.
2 mg SD-101 Plasma PK
Time Post-Infusion (n=14)
B.
4 mg SD-101 Plasma PK
Time Post-Infusion (n=13)
C.
8 mg SD-101 Plasma PK
Time Post-Infusion (n=6)
D.
SD-101 Concentrations Within Liver Tissue Post-Infusion
Max plasma level for 8 mg
Max plasma level for 4 mg
Max plasma level for 2 mg
SD-101 Dose

Figure 6. Plasma was collected on infusion days at the indicated time points and analyzed for SD-101 levels by LC-MS. **A.** 2mg dose **B.** 4mg dose, **C.** 8mg dose and **D.** in liver tissue post-infusion.

Figure 7. Improved immune cell infiltration in the tumor microenvironment

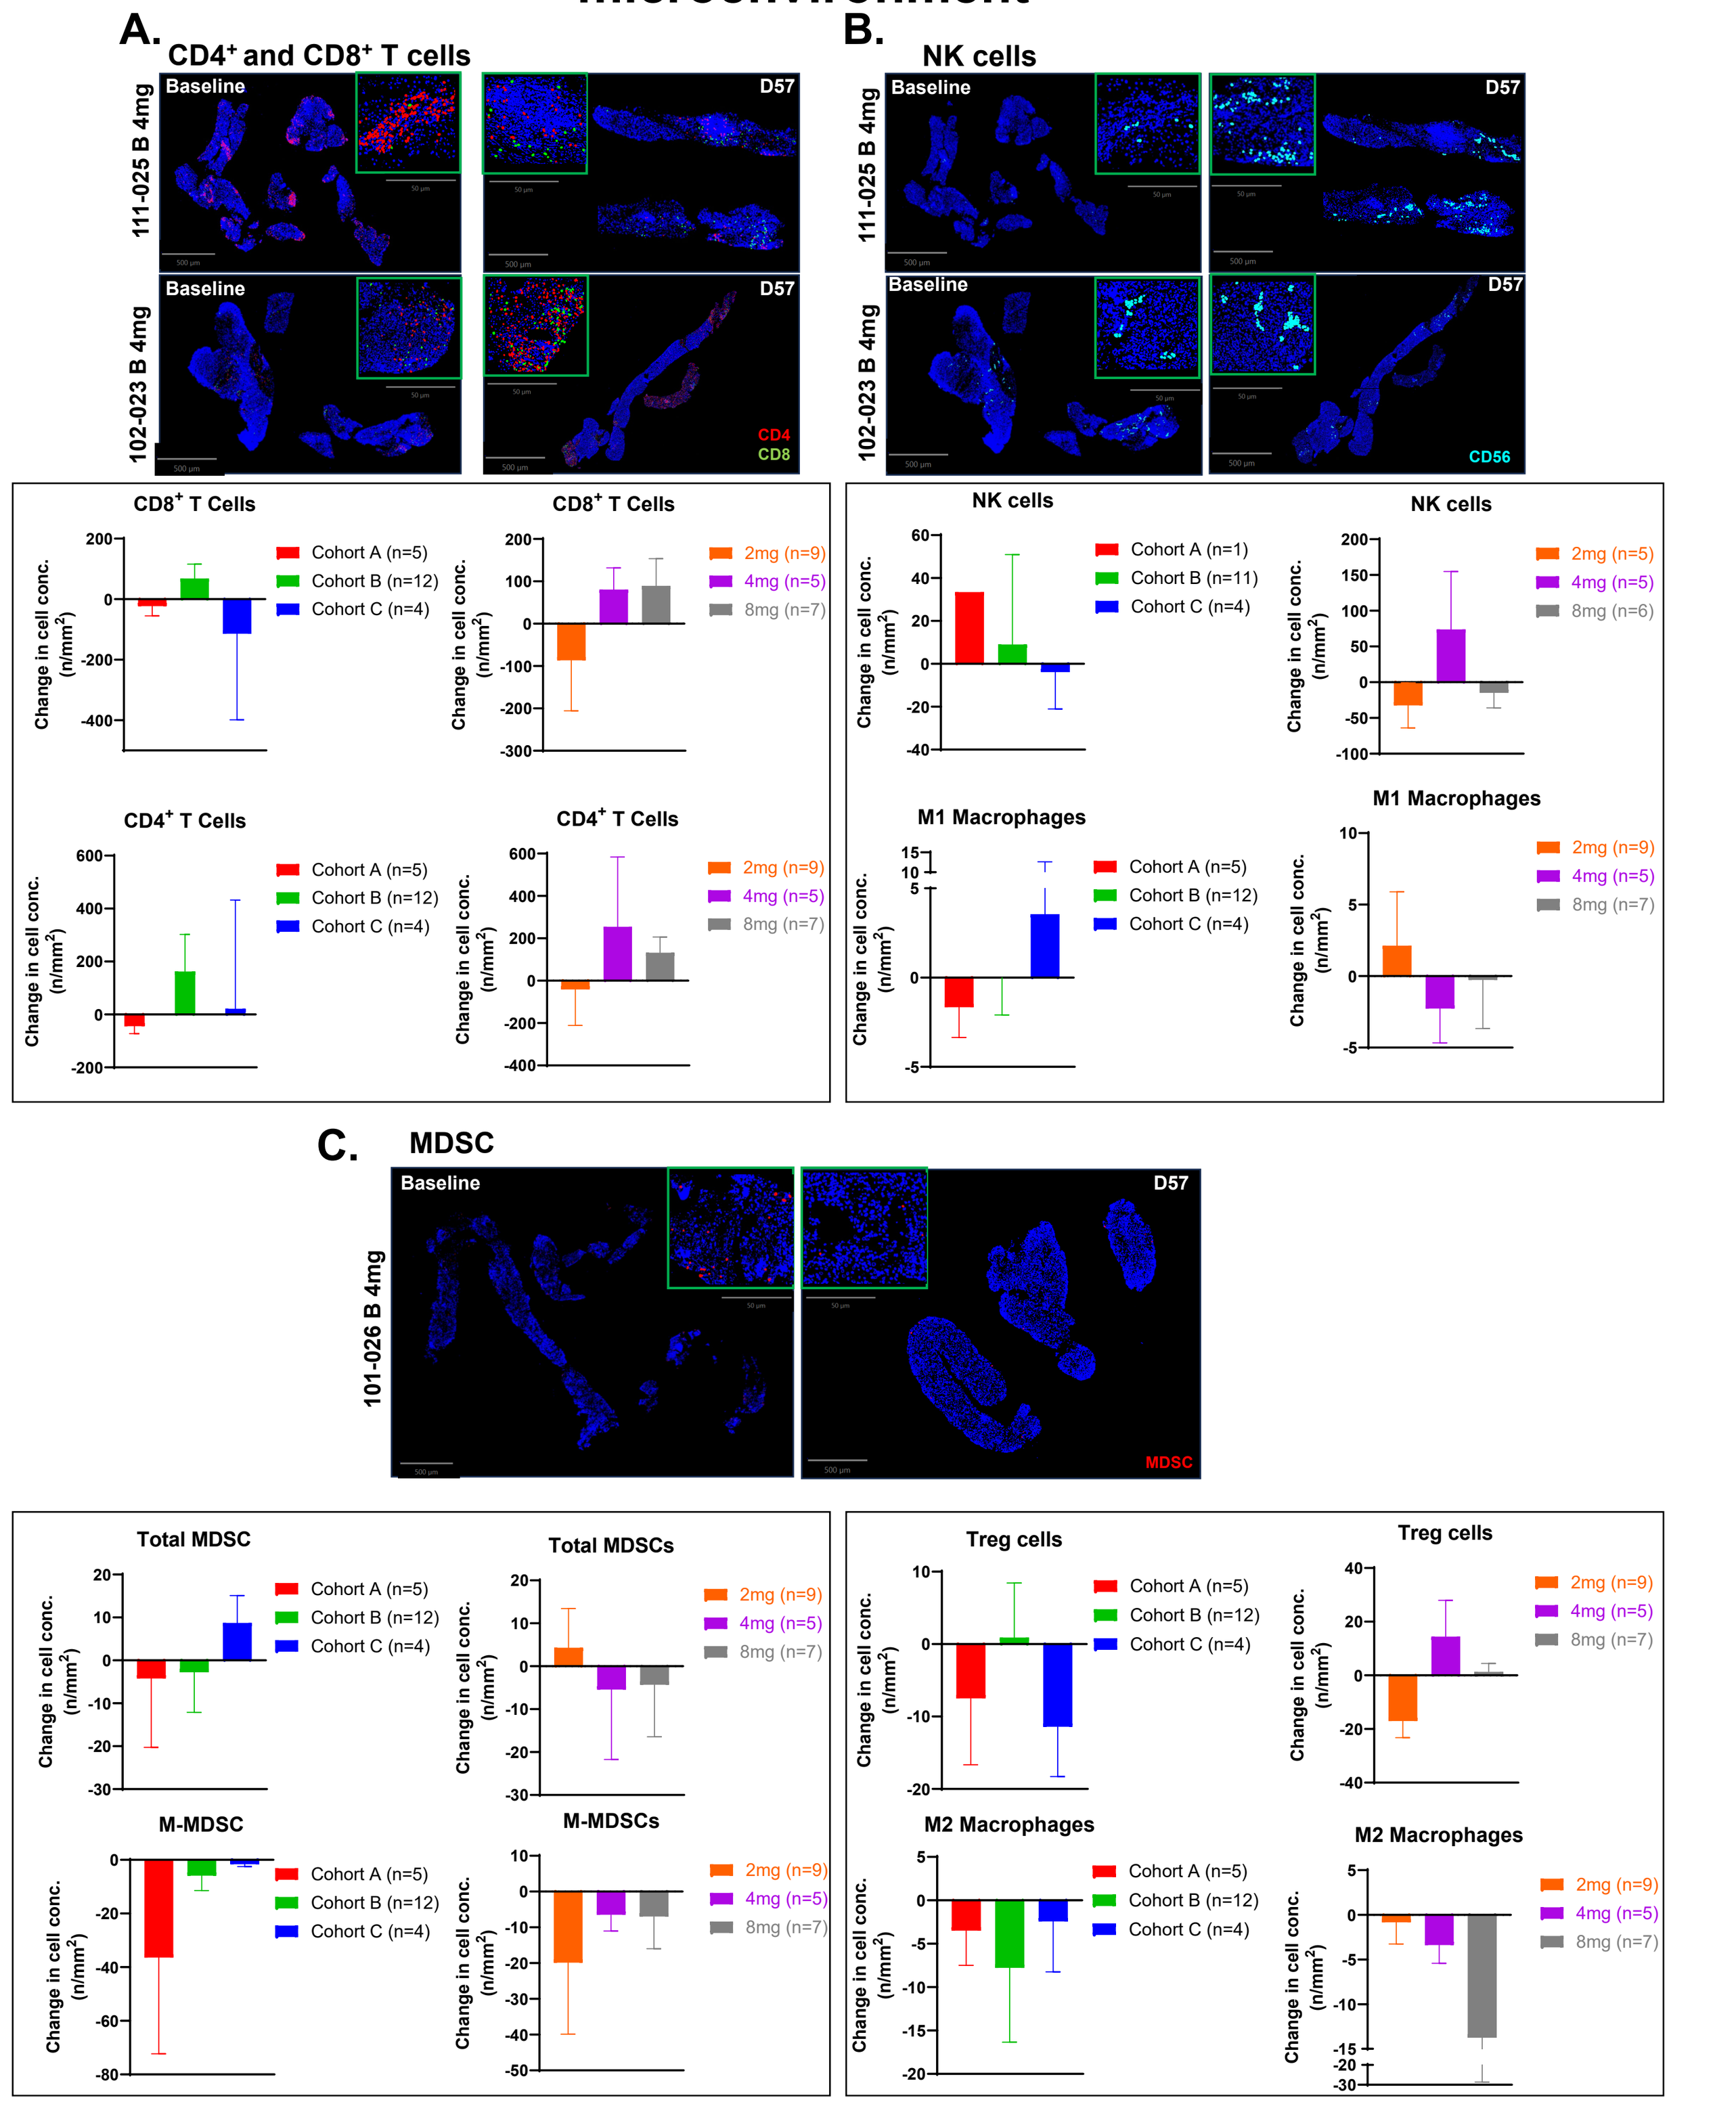
A.
CD4+ and CD8+ T cells
B.
NK cells
C.
MDSC
Baseline
D57

Figure 7. Multiplex immunofluorescence images of immune cell infiltration in baseline and Day 57 LM. $CD4^+$ and $CD8^+$ T cell infiltration in LM from patients 111-025 (top panel) and 102-023 (bottom panel) (**A**), NK cells ($CD3^-CD56^+$) (**B**) and MDSC ($CD45^+CD11b^+HLA\text{-}DR^-CD68^-$) for patient 101-026 (**C**). Images were quantified with QuPath. Change in cell concentration from baseline data represented as mean ± SEM.

Figure 8. Gene expression patterns in tumors and in circulation associated with tumor regression

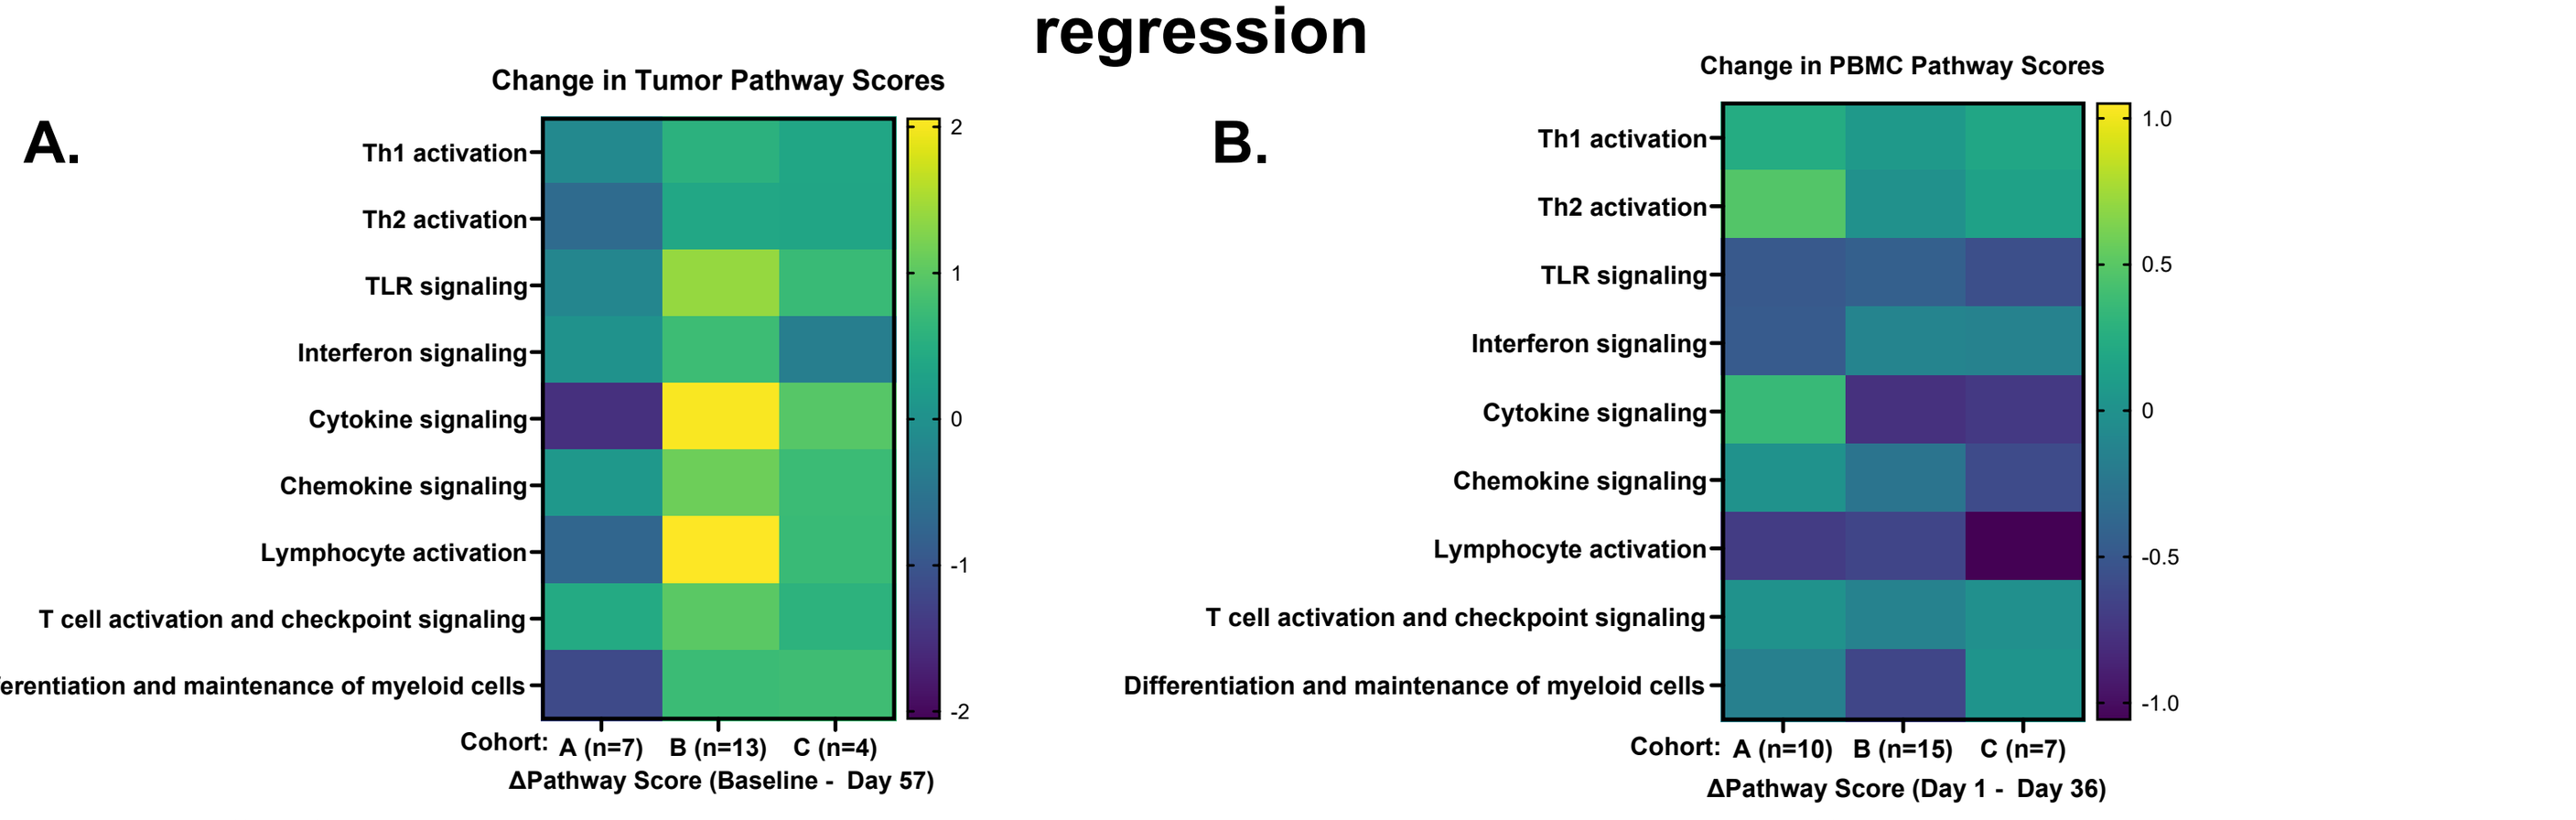
A.
Change in Tumor Pathway Scores
ΔPathway Score (Baseline - Day 57)
B.
Change in PBMC Pathway Scores
ΔPathway Score (Day 1 - Day 36)

Figure 8. Pathway scoring determined by advanced analysis of NanoString gene expression data at baseline and Day 57 for LM (**A**) and Day 36 for PBMCs (**B**).

Figure 9. Immune signatures in circulation associated with tumor regression

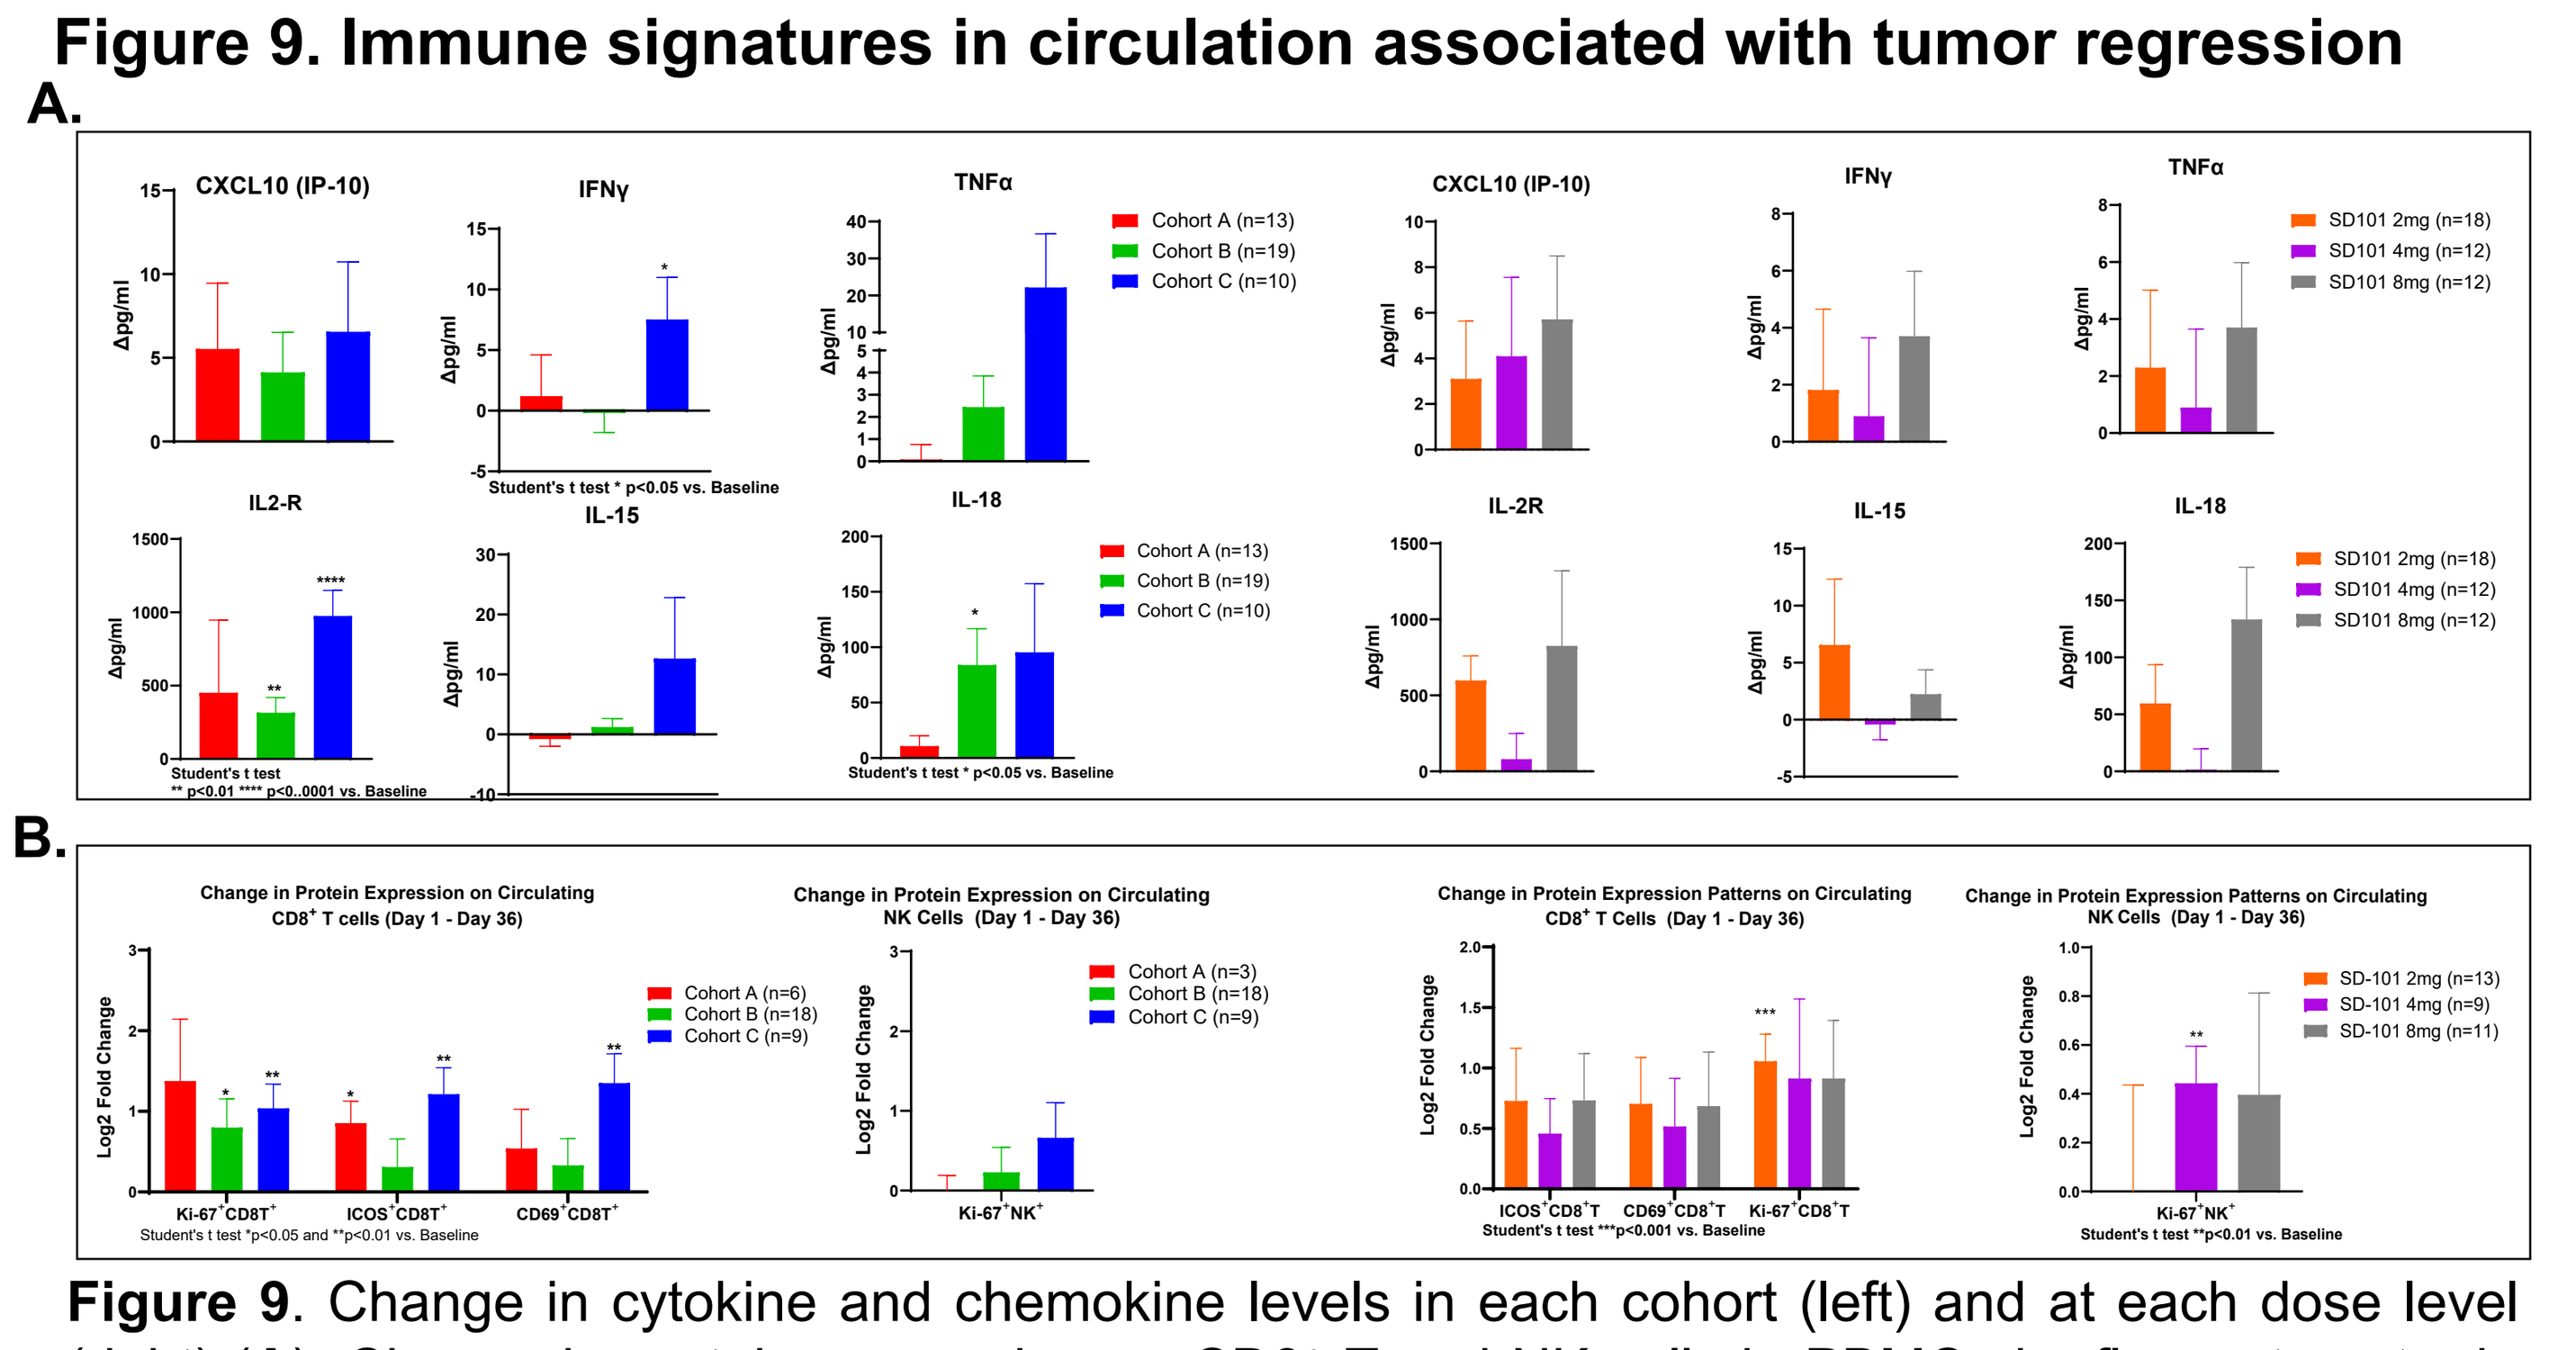

Figure 9. Change in cytokine and chemokine levels in each cohort (left) and at each dose level (right) (**A**). Change in protein expression on $CD8^+$ T and NK cells in PBMCs by flow cytometry in each cohort (left) and at each dose level (right) (**B**). Data presented as mean ± SEM.